Permanent Abstract Number 2629

The synergy between BXCL701, a DPP inhibitor, and immune checkpoint inhibitors discovered using AI and Big Data analytics

Luca Rastelli, Snigdha Gupta, Akhil Dahiya, Zeenia Jagga, Krishnan Nandabalan, Sanatan Upmanyu

BioXcel Corporation, Branford, CT

INTRODUCTION

Evolvere
BioXcel's in-house, artificial intelligence (AI) enabled platform, is designed to identify the most optimal therapeutic modalities and compounds to address target and disease biology. Evolvere was used to analyze all the known mechanisms of immune-activation and immune-evasion. The purpose was to identify the set of clinically validated compounds that could synergize with immune-checkpoint inhibitors (ICI) by remodelling the tumor micro-environment (ref.1).

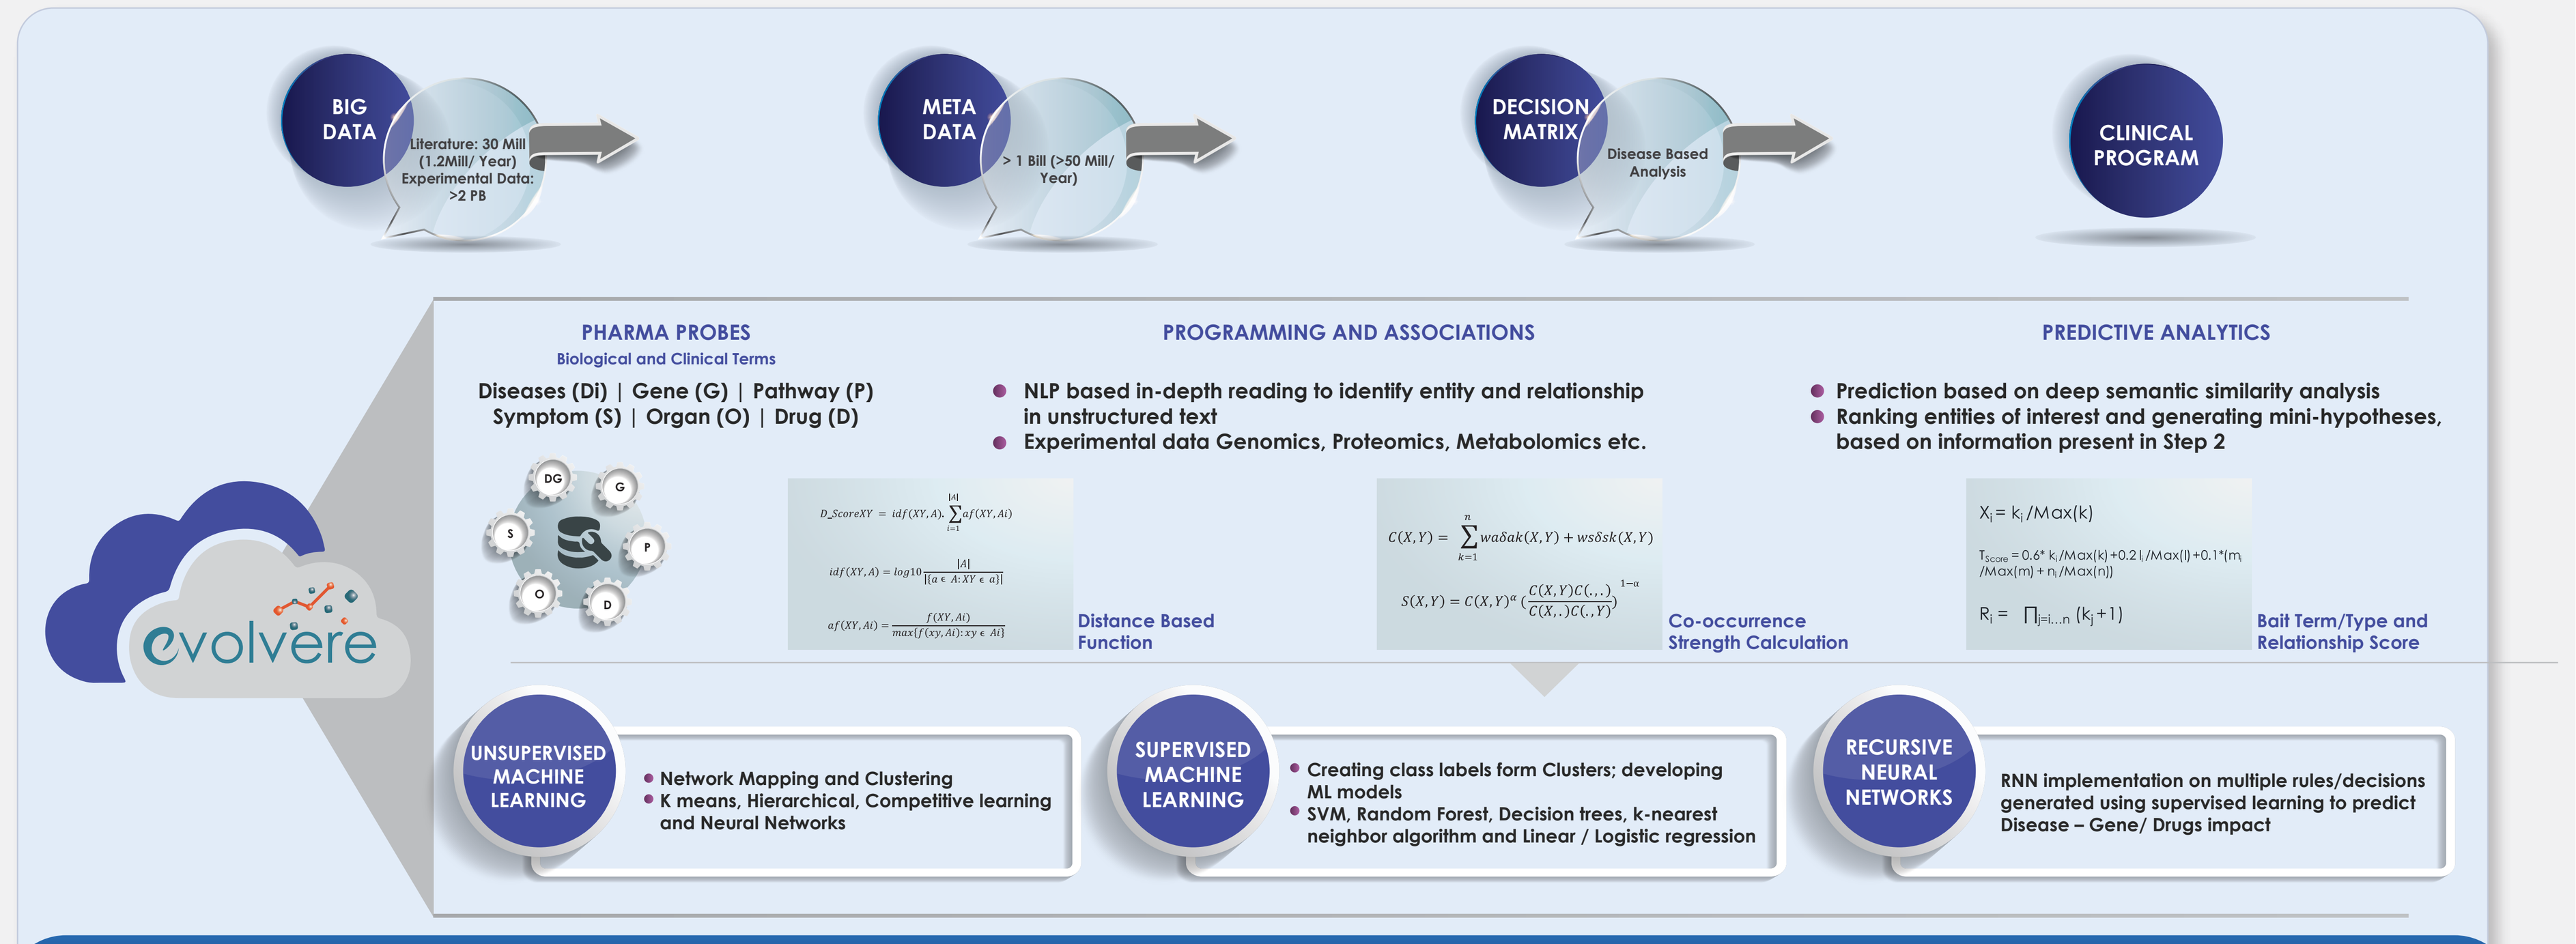

BIG DATA
Literature: 30 Mill (1.2Mill/ Year) Experimental Data: >2 PB
META DATA
> 1 Bill (>50 Mill/ Year)
DECISION MATRIX
Disease Based Analysis
CLINICAL PROGRAM
PHARMA PROBES
Biological and Clinical Terms
Diseases (Di) | Gene (G) | Pathway (P) Symptom (S) | Organ (O) | Drug (D)
PROGRAMMING AND ASSOCIATIONS
NLP based in-depth reading to identify entity and relationship in unstructured text
Experimental data Genomics, Proteomics, Metabolomics etc.
PREDICTIVE ANALYTICS
Prediction based on deep semantic similarity analysis
Ranking entities of interest and generating mini-hypotheses, based on information present in Step 2
Distance Based Function
Co-occurrence Strength Calculation
Bait Term/Type and Relationship Score
UNSUPERVISED MACHINE LEARNING
Network Mapping and Clustering
K means, Hierarchical, Competitive learning and Neural Networks
SUPERVISED MACHINE LEARNING
Creating class labels form Clusters; developing ML models
SVM, Random Forest, Decision trees, k-nearest neighbor algorithm and Linear / Logistic regression
RECURSIVE NEURAL NETWORKS
RNN implementation on multiple rules/decisions generated using supervised learning to predict Disease – Gene/ Drugs impact
evolvere

Figure 1: Evolvere - BioXcel's in-house artificial intelligence enabled platform

BXCL701 (Talabostat/PT-100) targeting FAP, DPP8 and DPP9, meets all the search criteria:
Inhibition of DPP8/9 results in the activation of multiple pro-inflammatory immune cells (Neutrophil, T-cell, NK) and decrease the immuno-suppressive activity of MDSCs by inducing a granulocytic differentiation. FAP inhibition blocks the activation of immuno-suppressive fibroblasts and it increases immune cell extravasation into the tumor tissue. Further immunogenomics analysis revealed that BXCL701 targets are co-overexpressed with PDL1 in a subset of castration-resistant prostate cancer making this segment attractive for clinical trial with this combination.

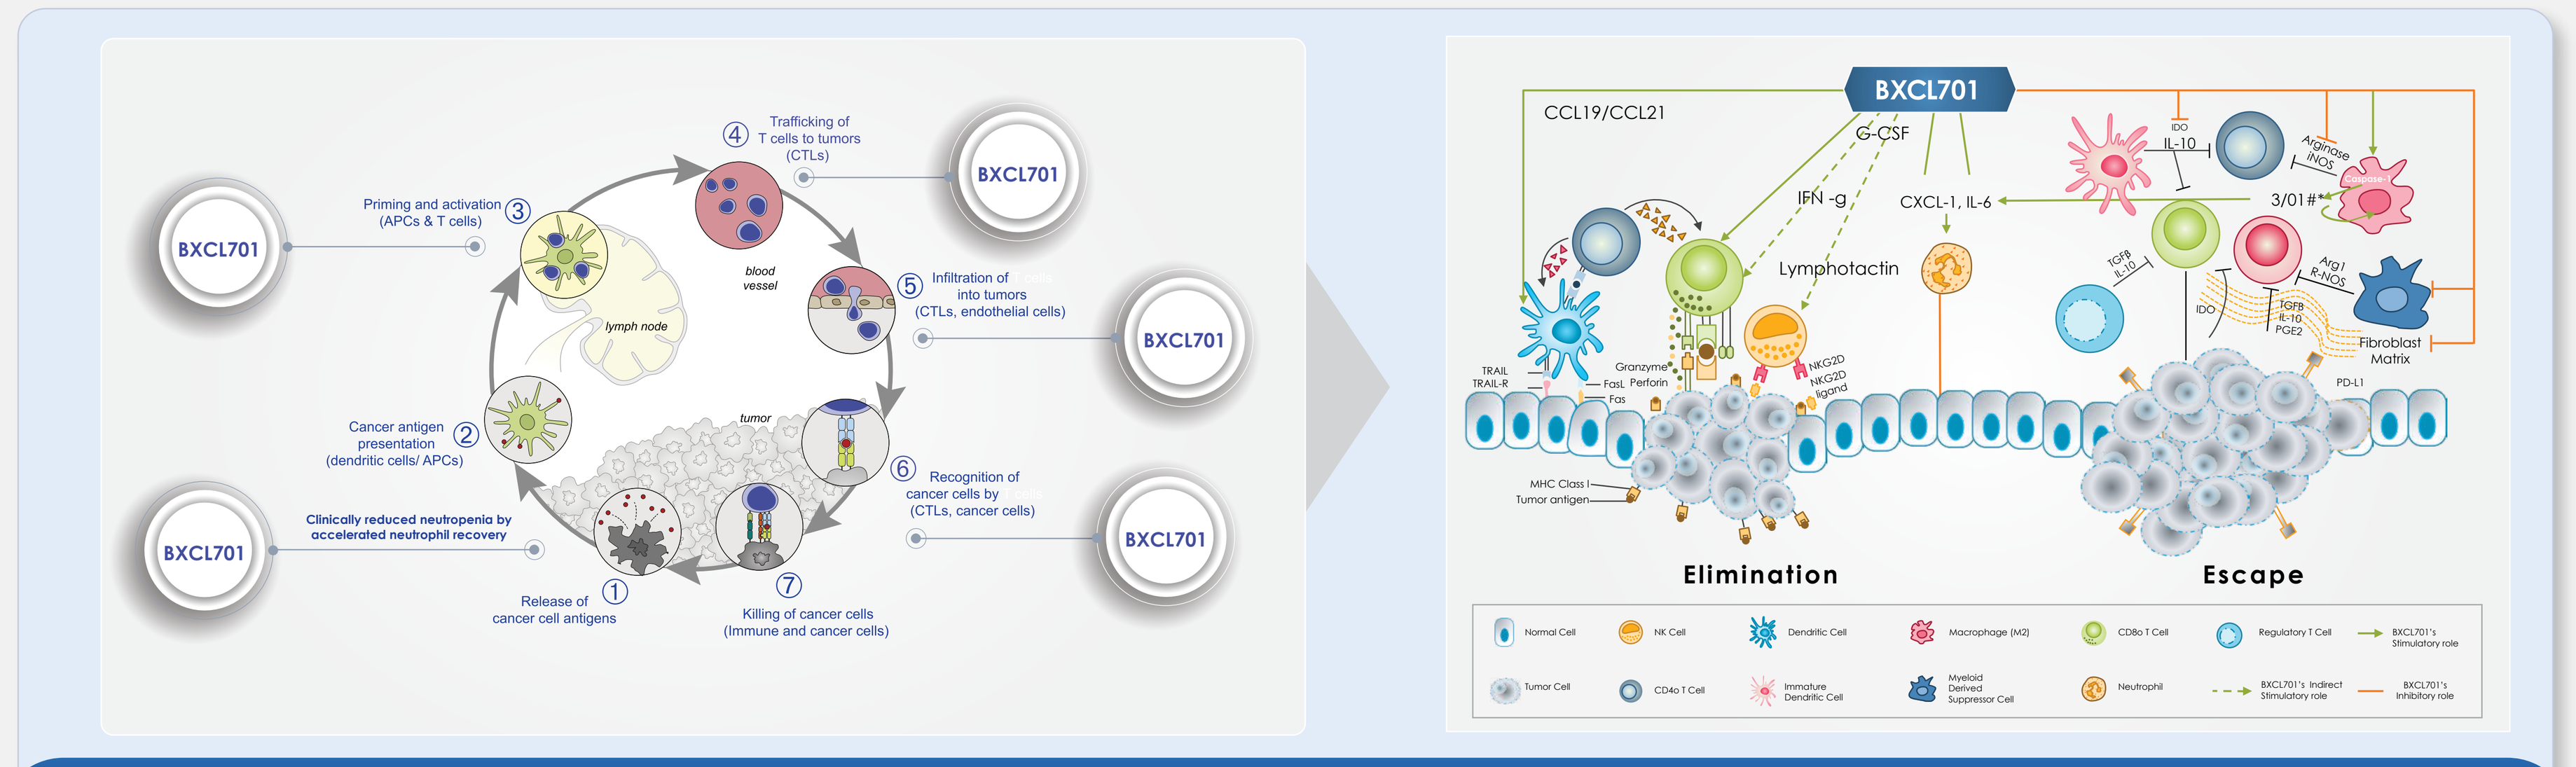

BXCL701
Trafficking of T cells to tumors (CTLs)
Priming and activation (APCs & T cells)
Infiltration of into tumors (CTLs, endothelial cells)
Cancer antigen presentation (dendritic cells/ APCs)
Recognition of cancer cells by (CTLs, cancer cells)
Clinically reduced neutropenia by accelerated neutrophil recovery
Release of cancer cell antigens
Killing of cancer cells (Immune and cancer cells)
CCL19/CCL21
G-CSF
IFN -g
CXCL-1, IL-6
Lymphotactin
Fibroblast Matrix
Elimination
Escape

Figure 2: BXCL701 influencing cancer-immune cycle and its dual role in immuno-oncology

OBJECTIVES

- To identify in-silico hypotheses for synergistic combinations in ICI therapy based on public domain literature and experimental dataset using Evolvere
- To validate experimentally the Evolvere-prioritized compounds by testing their antitumor activity in combination with anti-PD1 in syngeneic mouse models and define the biochemical and cellular mechanism for synergy
- To use immune-genomics approaches to prioritize tumors for BXCL701 and anti-PD1 combination therapy

KEY FINDINGS ...

A. Evolvere IO test case: BXCL701 as a potential combination agent for ICI therapy

Evolvere leveraged techniques like natural language processing, co-occurrence scores and relationships mapping to unravel novel connections between compounds, genes, immune modulation and cancer types and generate combination hypothesis[A1].
These hypotheses were prioritized based on a proprietary matrix scoring the biological relevance of the mechanism and the level of clinical validation, safety concerns and opportunity for biomarker use [A2]. Using SysBiomics, a protein interaction map of BXCL701 targets with PDL1 identfied 5 transcriptional modulators with link to prostate cancer –GATA2 (prostate cancer aggressiveness {ref.2}); C/EBP (proliferative inflammatory atrophy of the prostate {ref.3}); STAT3 (target and biomarker in advanced prostate cancer {ref.4}); EGR1(gene expression modulation in CRPC {ref.5}) and CTCF (mutations associated with prostate cancer {ref.6}) (A3).

A1: Evolvere Methodology to generate combination hypotheses in Immuno Oncology Space

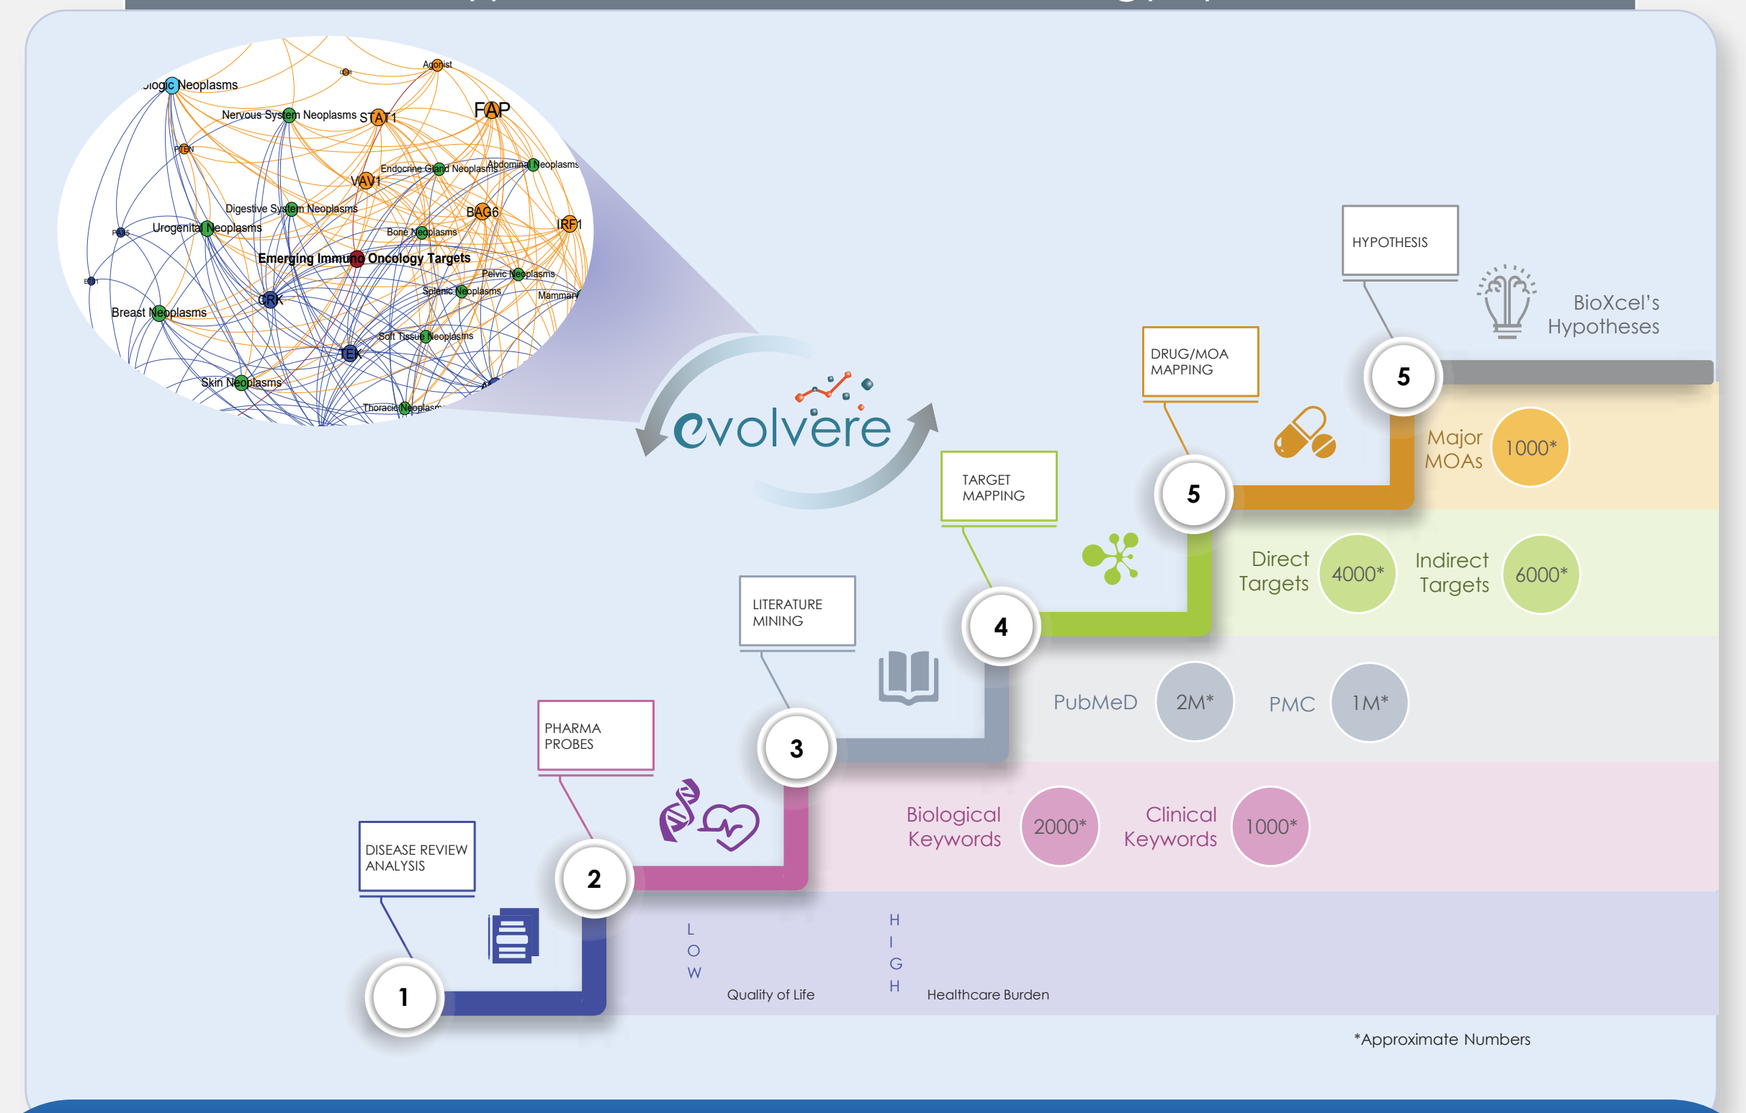

evolvere
DISEASE REVIEW ANALYSIS
PHARMA PROBES
LITERATURE MINING
TARGET MAPPING
DRUG/MOA MAPPING
HYPOTHESIS
BioXcel's Hypotheses
Major MOAs 1000*
Direct Targets 4000*
Indirect Targets 6000*
PubMeD 2M*
PMC 1M*
Biological Keywords 2000*
Clinical Keywords 1000*
Quality of Life
Healthcare Burden
*Approximate Numbers

Figure 3: Evolever's methodology to identify potential repurposing opportunities in immuno-oncology space

A2: BXCL701 emerged as potential repurposing opportunity

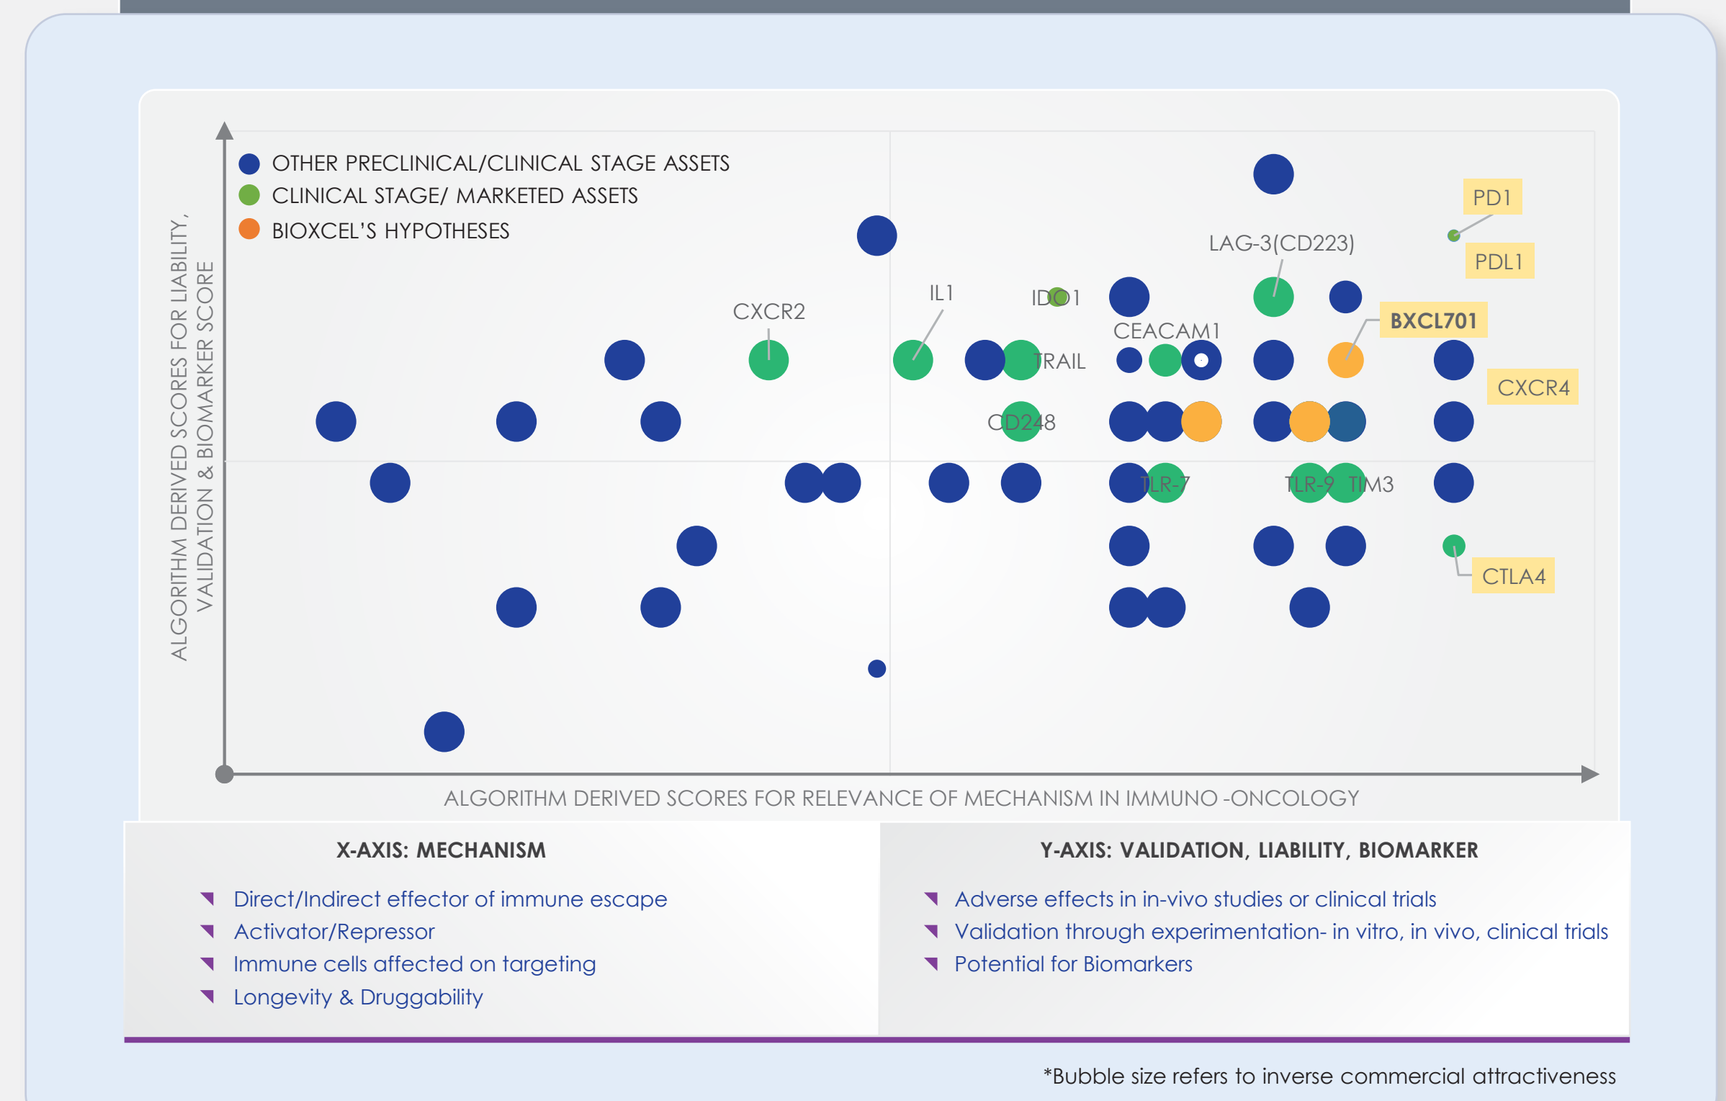

OTHER PRECLINICAL/CLINICAL STAGE ASSETS
CLINICAL STAGE/ MARKETED ASSETS
BIOXCEL'S HYPOTHESES
ALGORITHM DERIVED SCORES FOR LIABILITY, VALIDATION & BIOMARKER SCORE
ALGORITHM DERIVED SCORES FOR RELEVANCE OF MECHANISM IN IMMUNO -ONCOLOGY
PD1
PDL1
LAG-3(CD223)
BXCL701
CXCR4
CXCR2
CEACAM1
CTLA4
X-AXIS: MECHANISM
Direct/Indirect effector of immune escape
Activator/Repressor
Immune cells affected on targeting
Longevity & Druggability
Y-AXIS: VALIDATION, LIABILITY, BIOMARKER
Adverse effects in in-vivo studies or clinical trials
Validation through experimentation- in vitro, in vivo, clinical trials
Potential for Biomarkers
*Bubble size refers to inverse commercial attractiveness

Figure 4: Hypotheses prioritization based on mechanism, liability, target-validation, biomarkers potential, and commercial attractiveness.

A3: Interactome for BXCL701 targets and PDL1

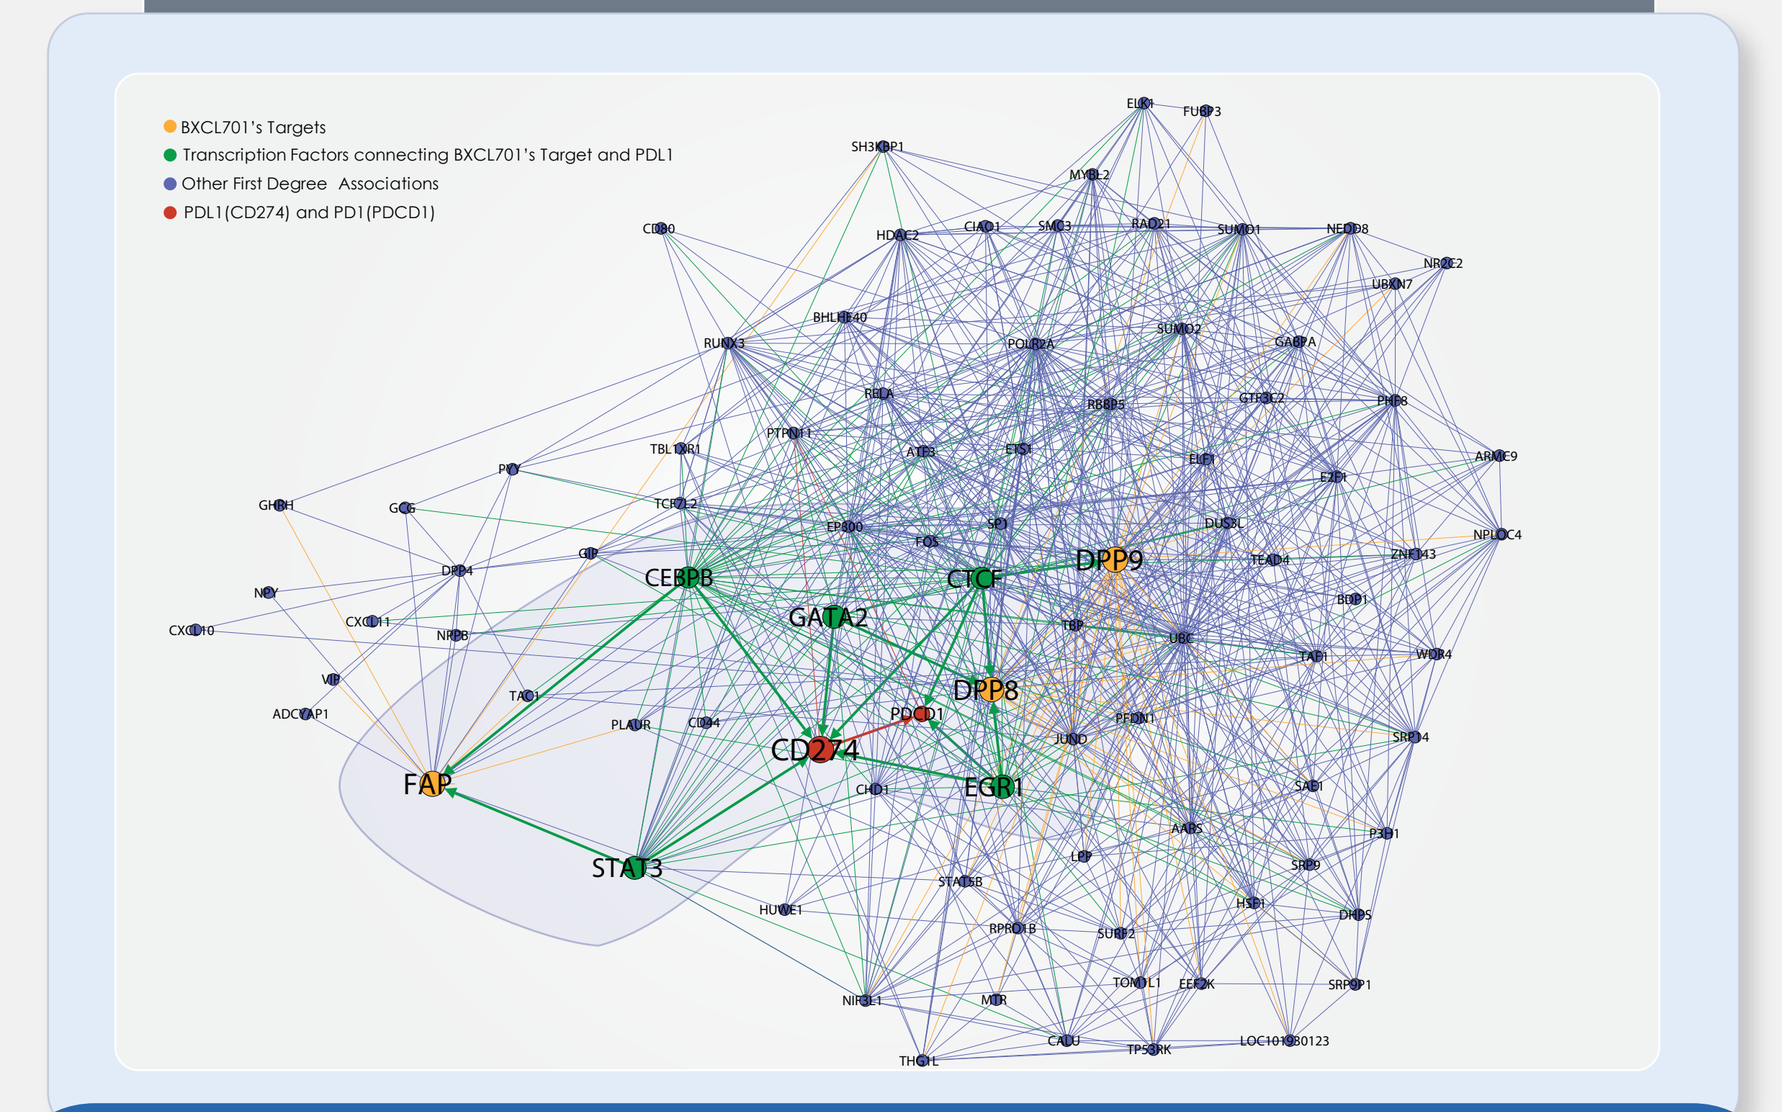

BXCL701's Targets
Transcription Factors connecting BXCL701's Target and PDL1
Other First Degree Associations
PDL1(CD274) and PD1(PDCD1)
CEBPB
GATA2
CTCF
DPP9
DPP8
CD274
EGR1
FAP
STAT3

Fig 5. Interaction Network of BXCL701 targets (as orange nodes) with PDL1/CD274 (as green nodes) through 5 transcription factors (as green nodes) using Cytoscape v3.3.0 by Bisogenet v.3.0 plugin.

B. Experimental Validation- for BXCL701 with anti-PD1 in M38 syngeneic mouse model

MC38 mouse colon cancer cell line (1.0x106 MC-38 cells/ 0.1mL Hanks Balanced Salt Solution) were implanted in the subcutaneous space of the flank of mice of female C57BL/6 mice. In the first experiment [B1a] tumors grow slower and all treatment groups had statistically higher anti-tumor activity compared to controls and BXCL701 and anti-PD1 antibody combination had higher efficacy than the single agent treatments. In the second experiment [B1b], tumor grew much faster. None of the single agent treatment had statistically higher anti-tumor activity compared to controls but BXCL701 and anti-PD1 antibody combination had significant antitumor activity and 10 µg BID and 20 µg QD showed comparable efficacy, Upregulation of pro-inflammatory cytokines (IL-2, IL-6, IL-12p40), immunostimulatory chemokines (GM-CSF, G-CSF) and cytokines predicting memory T-cell response (IL-15 and IL-7) were observed post-treatment [B2]. Immunophenotyping revealed that the combination of BXCL701 with anti-PD1 increased the percentage of the cytotoxic NK cells and macrophages in the tumor and blood respectively while decreased in the immunosuppressive T regulatory cells [B3].

B1: Multiple repurposing hypotheses were generated by Evolvere for ICI combination

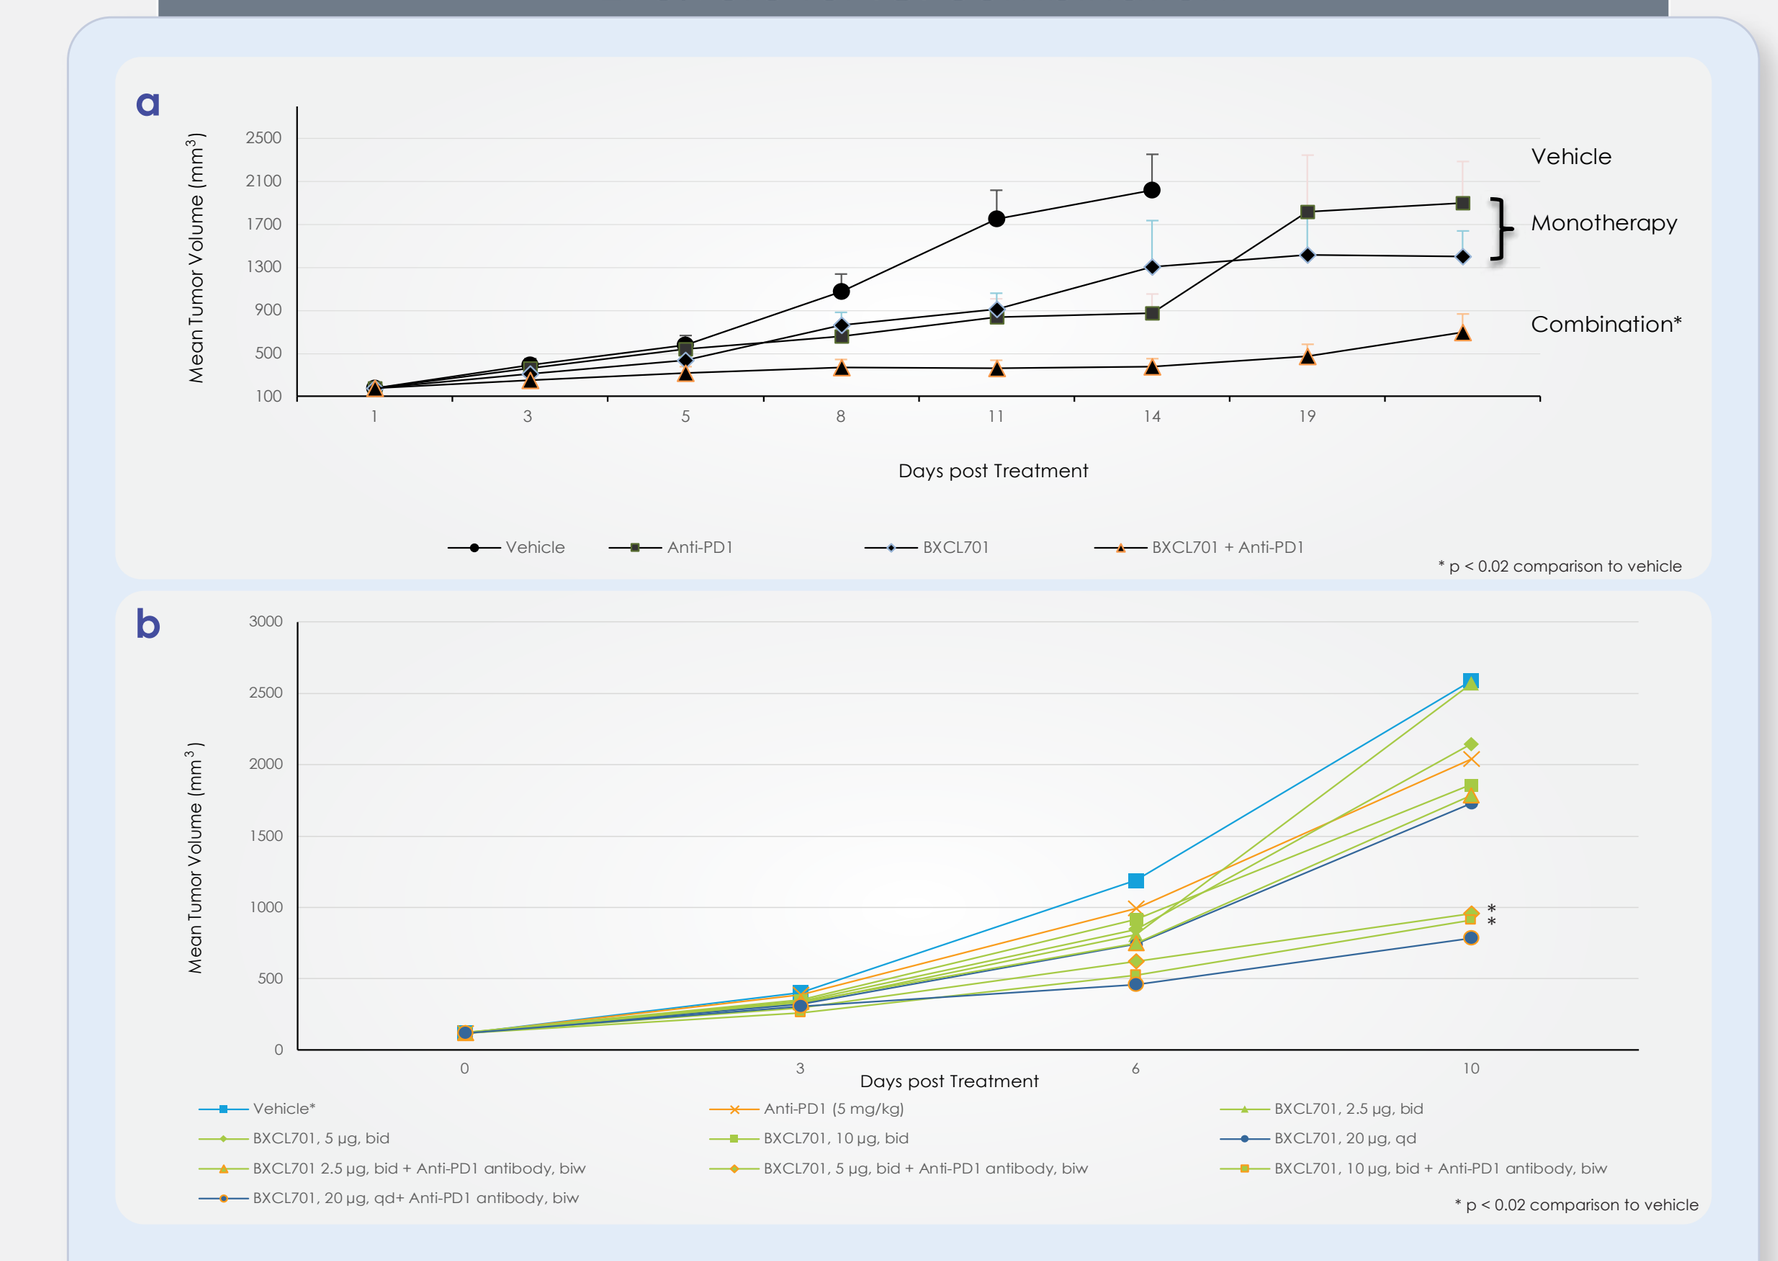

a
Mean Tumor Volume (mm³)
Vehicle
Monotherapy
Combination*
Days post Treatment
Vehicle
Anti-PD1
BXCL701
BXCL701 + Anti-PD1
* p < 0.02 comparison to vehicle
b
Mean Tumor Volume (mm³)
Days post Treatment
* p < 0.02 comparison to vehicle

Figure 6: Comparing treatment effect of BXCL701 with anti-PD1 (combination), BXCL701 (single agent), anti-PD1(control), vehicle (control) on mean tumor volume in syngenic mouse model - MC38 in C57BL/6 mice

B2: BXCL701 emerged as potential repurposing opportunity

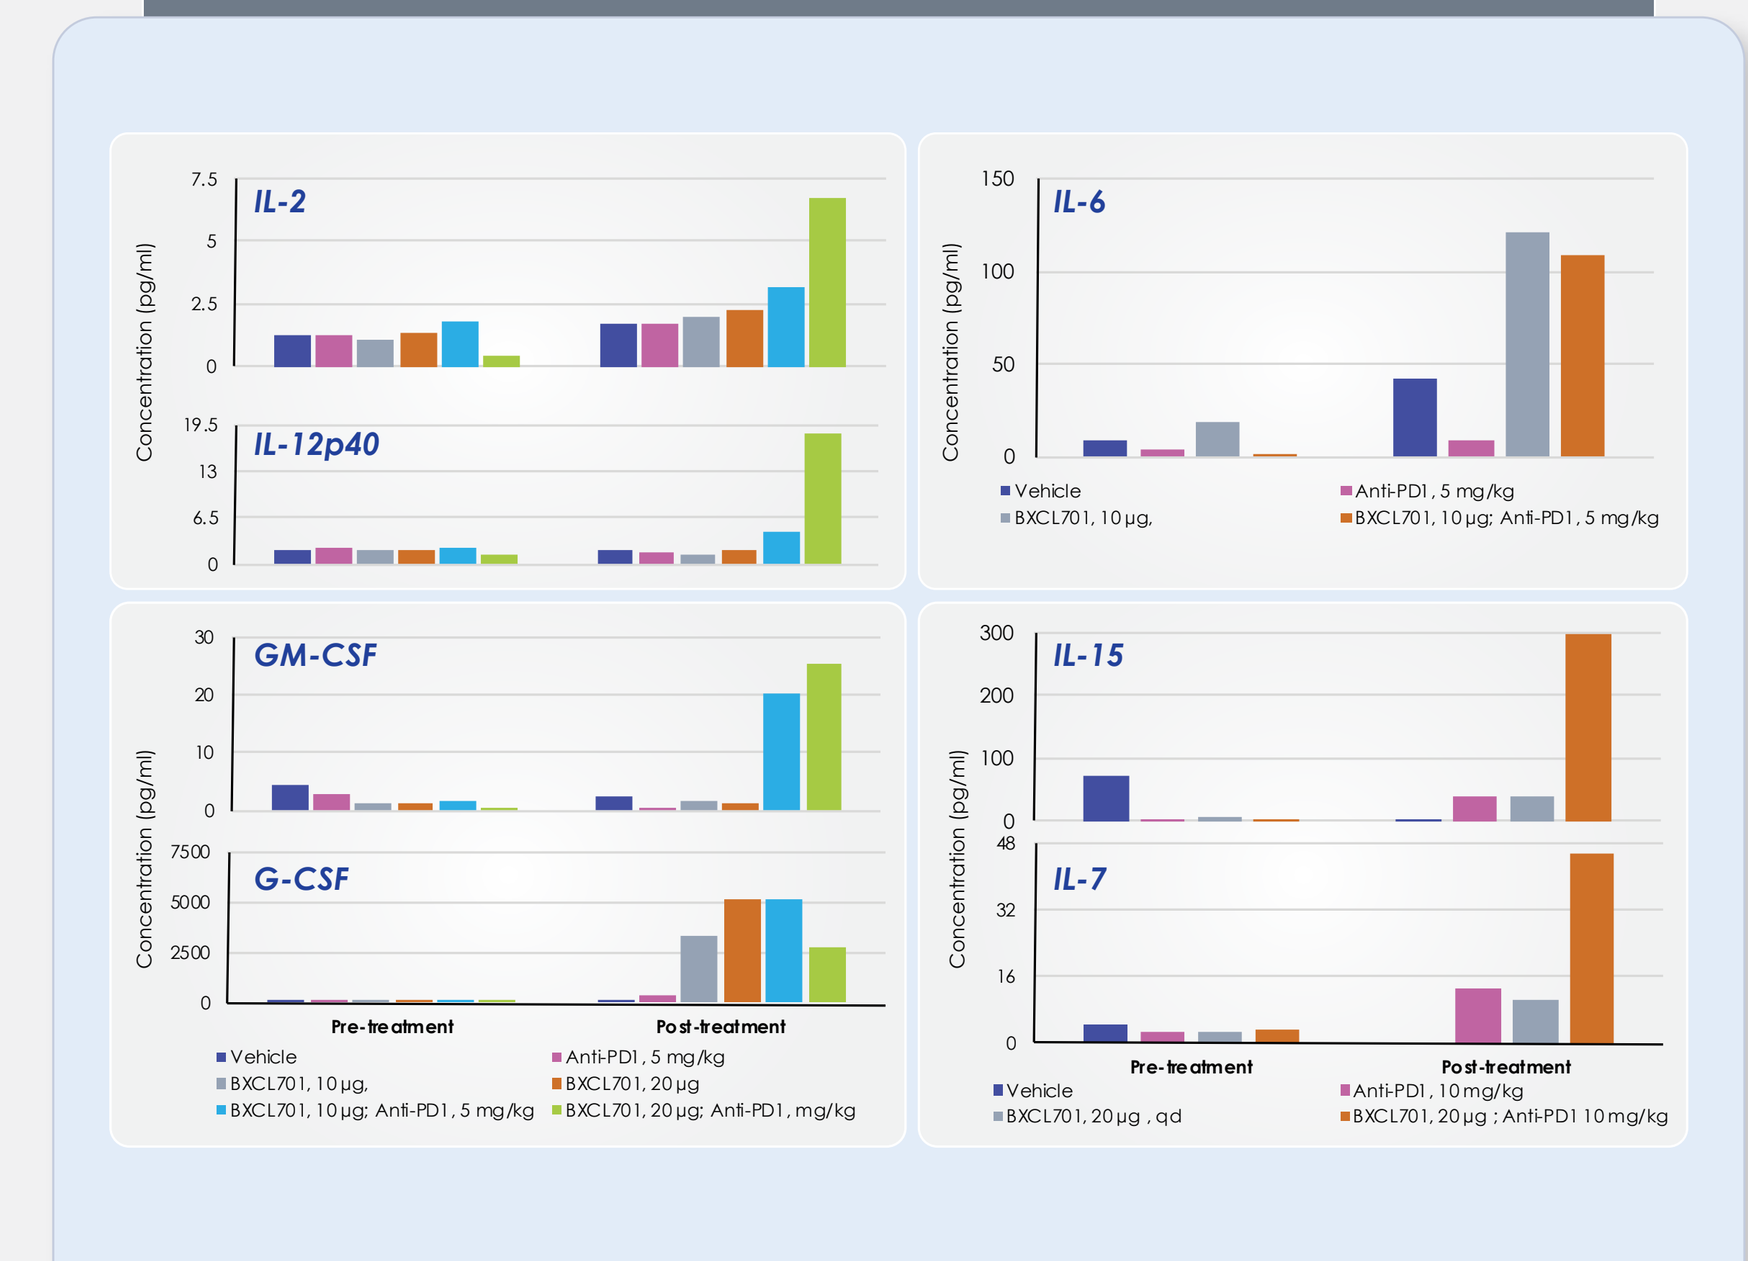

IL-2
IL-12p40
IL-6
GM-CSF
G-CSF
IL-15
IL-7
Concentration (pg/ml)
Pre-treatment
Post-treatment
Vehicle
Anti-PD1, 5 mg/kg
BXCL701, 10µg
BXCL701, 20µg
BXCL701, 10µg: Anti-PD1, 5 mg/kg
BXCL701, 20µg: Anti-PD1, mg/kg

Figure 7: Comparing immuomodulatory effects by Luminex assay for (a) pro-inflammatory cytokines (b) chemokines and (c) associated memory T-cells by dose administration at QD, BID as single agent and in combination with anti-PD1

B3: Colorectal cancer was selected for BXCL701 for experimental validation

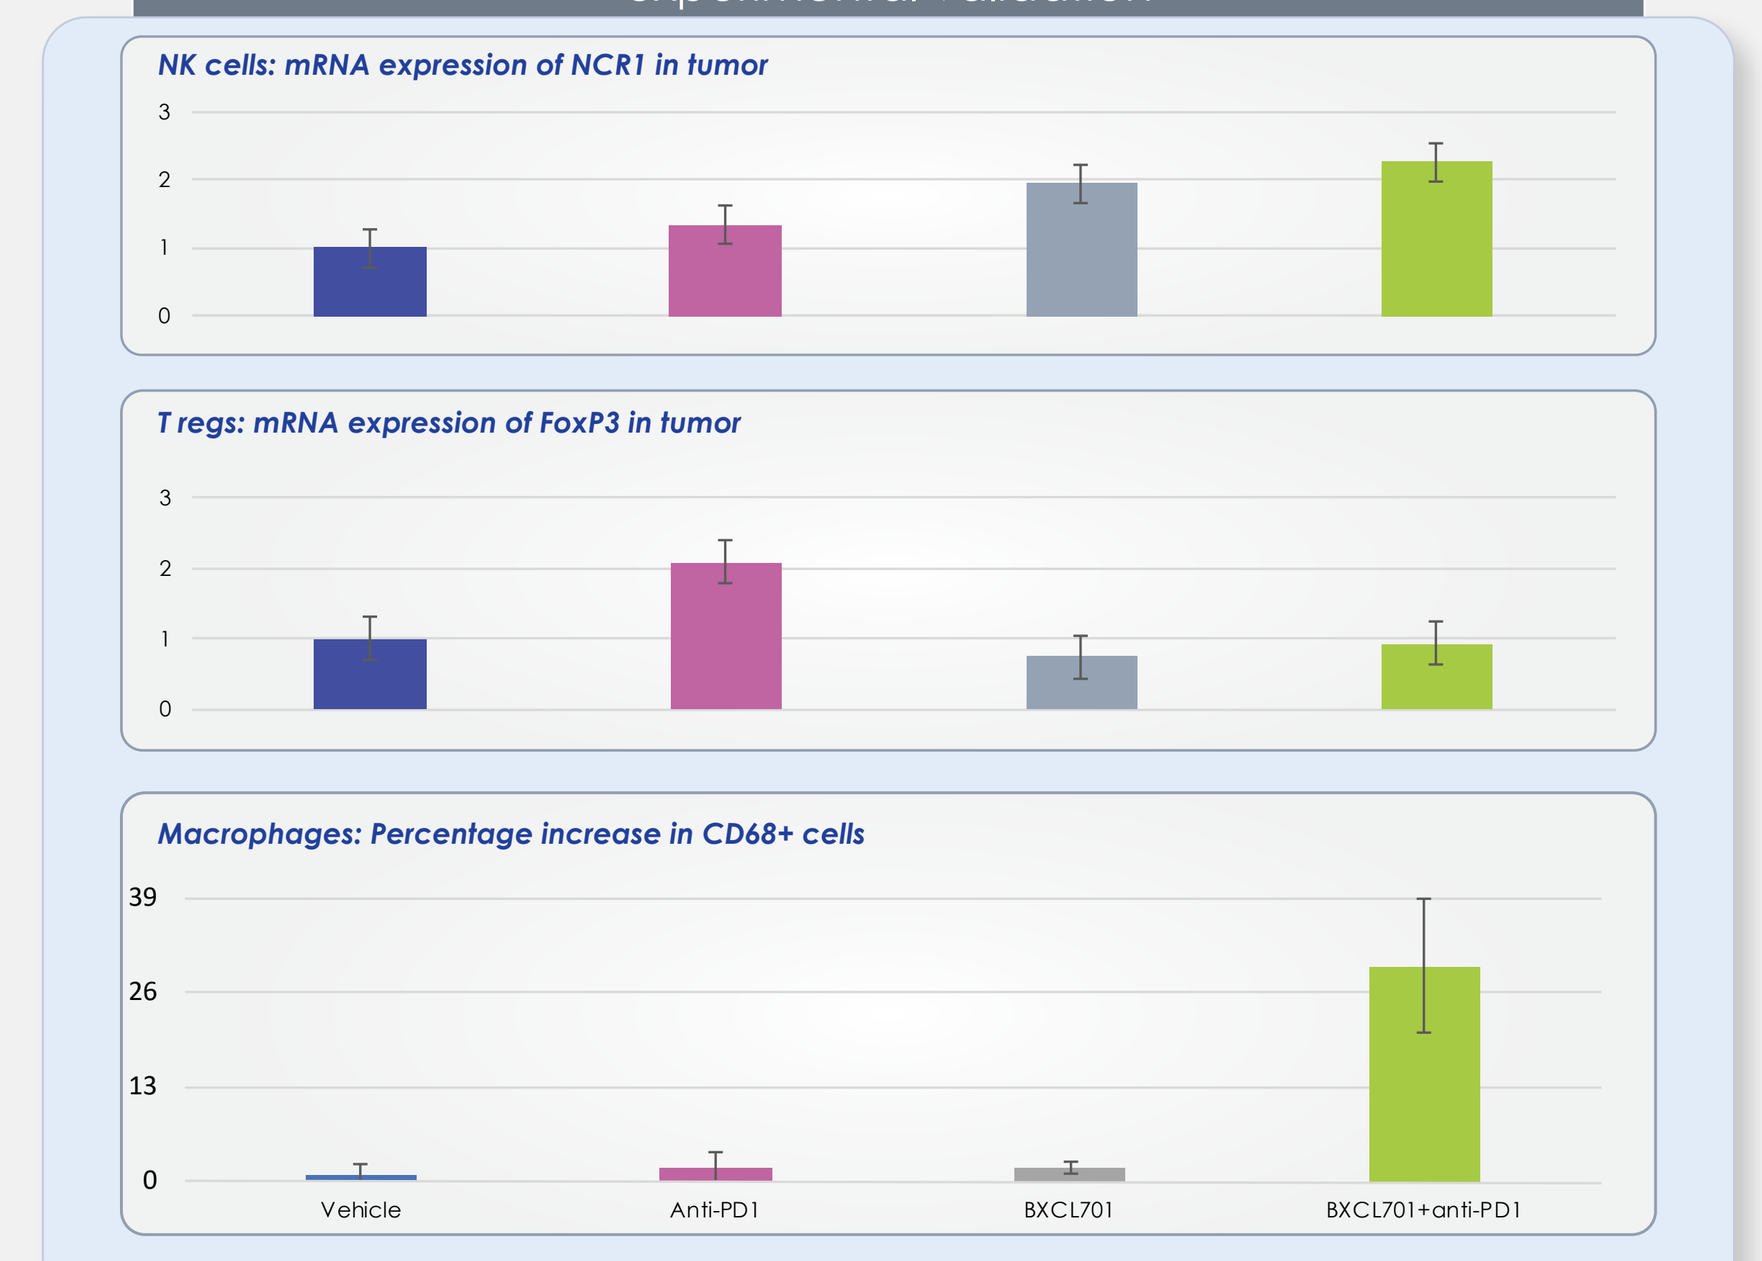

NK cells: mRNA expression of NCR1 in tumor
T regs: mRNA expression of FoxP3 in tumor
Macrophages: Percentage increase in CD68+ cells
Vehicle
Anti-PD1
BXCL701
BXCL701+anti-PD1

Figure 8: Comparing percentage of NK cells, macrophages and T-regulatory cells in tumor and blood samples by immunophenotyping for anti-PD1, BXCL701 and BXCL701 + anti-PD1 combination

C. Analysis of genomic alterations in FAP, DPP8 and DPP9 across cancer genomic profiles

Genomic alterations in FAP, DPP8 and DPP9 were analyzed across 33172 patients from 150 different cancer studies included dataset from TCGA at cBioPortal identified castration-resistant prostate cancer patients pretreated with Abiterone and Enzulatimide (ref.7-9) as an outlier. High level amplification of DPP9 (14%) and overexpression of DPP8 in 50% of CRPC suggests that this patient population could be uniquely sensitive to this monotherapy treatment. In addition the almost complete overlap of the overexpression of DPP8 and PDL1 suggests could be sensitive to combination treatment.

KEY FINDINGS ...

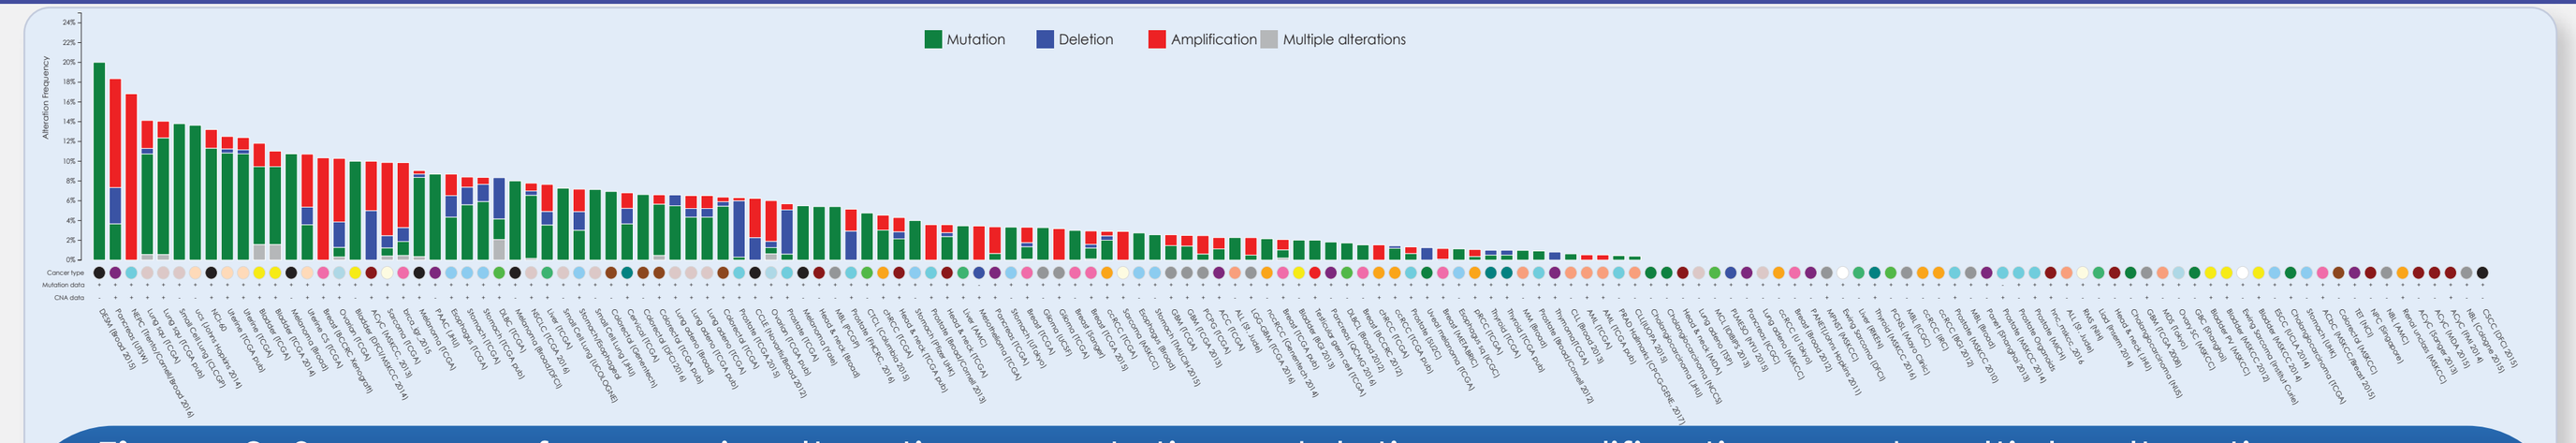

Mutation
Deletion
Amplification
Multiple alterations

Figure 9: Summary of genomic alterations - mutations, deletions, amplifications and multiple alteration - across cancers for BXL701 targets
(Accessed from cBioPortal Version 1.4.3 as on 9th March, 2017)

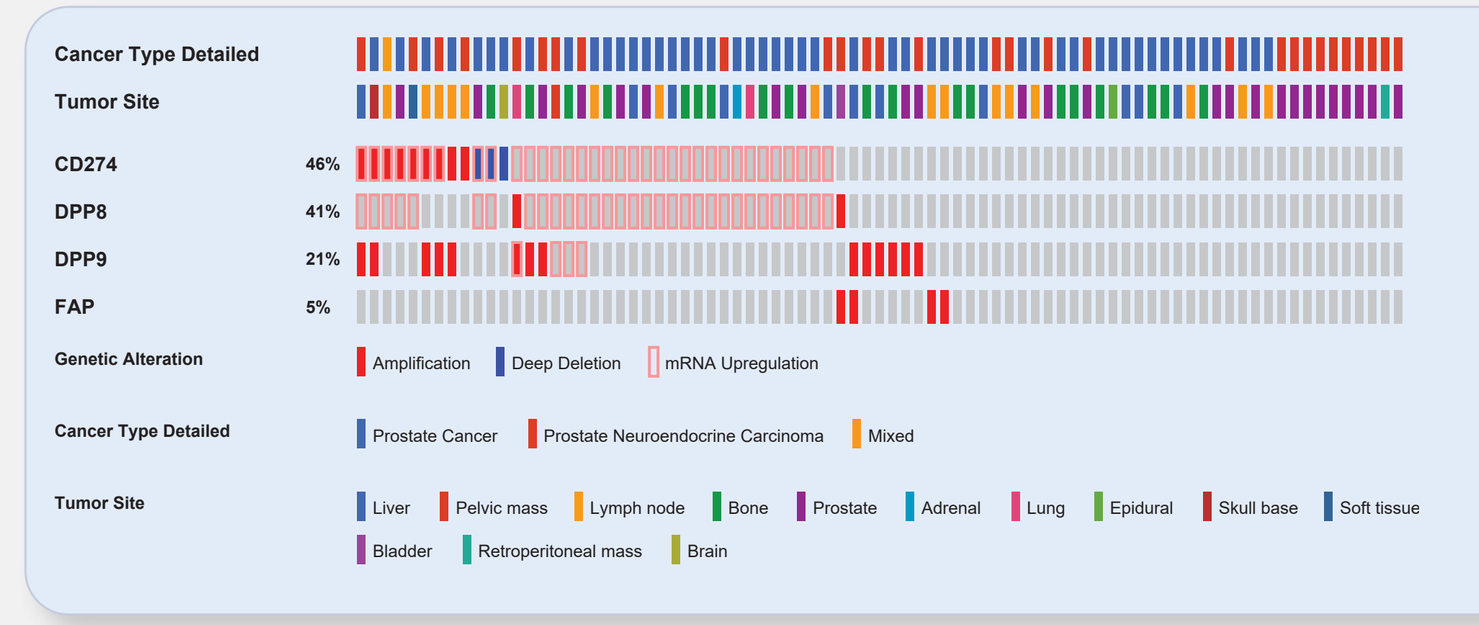

Cancer Type Detailed
Tumor Site
CD274 46%
DPP8 41%
DPP9 21%
FAP 5%
Genetic Alteration
Amplification
Deep Deletion
mRNA Upregulation
Prostate Cancer
Prostate Neuroendocrine Carcinoma
Mixed
Liver
Pelvic mass
Lymph node
Bone
Prostate
Adrenal
Lung
Epidural
Skull base
Soft tissue
Bladder
Retroperitoneal mass
Brain

Figure 10: Oncoprint representation of Prostate cancer dataset of Beltran et al, 2016 highlights co-amplification and over expression of BXCL701 targets DPP8 and DPP9 with PDL1 (CD274). Each row represents a gene and columns represent each sample.
(Accessed from cBioPortal Version 1.4.3 as on 9th March, 2017)

CONCLUSIONS

Demonstration of the capability of Evolvere, an artificial intelligence driven big data analytics platform to generate in-silico hypotheses for synergistic combinations in ICI therapy

Validation of the synergistic impact of BXCL701 and anti-PD1 combination in a syngeneic mouse model for colon adenocarcinoma

Identification of exciting opportunity for BXCL701, alone and in combination with anti-PD1 therapy in prostate cancer patients based on landscape analysis of target expression profiling

FUTURE PERSPECTIVE

A prediction model for combination therapy of ICI+BXCL701 is being developed for to select putative responder patients based on the effect of BXCL701 mechanism on ICI response and resistance

- ICI-Response modeling based on target expression and/or amplification levels, high mutational load, low intra-tumor heterogeneity, deficiencies in DNA repair machinery and levels of immune cell infiltration.
- ICI-Resistance modeling based on alterations in signaling pathways of cell proliferation and apoptosis, stabilization of immune check points and alteration in MHC-peptide

ACKNOWLEDGEMENTS

We would like to acknowledge the contribution of Dr. Aparna Sapra, for the ground work provided by her during the conceptualization of the programme. Further, we would like to appreciate the technical support provided by Genscript for carrying out all the work associated with the animal studies. A continuous support by Ranjeeta Kanojia, IP Team for enabling us to patent the potential concepts and we hereby acknowledge their constant support. Further we would also like to express our gratitude to Vandna Sharma of the designing team for the constant support extented for the designing inputs as needed for this poster.
We are thankful to Dr. Himisha Beltran, Dr. Johan Debono and Dr. Emmanuel Stylianos Antonarakis for the discussions on future prospective of the work presented here.

REFERENCES

1. Kaw S, Upmanyu S, Sharma H, and Nandabalan K. "Targeting immune checkpoints: using a big data approach for their identification, prioritization and application." (2015): 64-64.
2. Rodriguez-Bravo V, Carceles-Cordon M, Hoshida Y, Cordon-Cardo C, Galsky M D and Domingo-Domenech J. "The role of GATA2 in lethal prostate cancer aggressiveness." Nature Reviews Urology 14 (2016): 38-48.
3. Kim MH and Field J. "Translationally regulated C/EBPβ isoform expression upregulates metastatic genes in hormone-independent prostate cancer cells." Prostate 68,12 (2008): 1362-1371.
4. Don-Doncow N. "STAT3 in Prostate Cancer." (2017).
5. Jacob S, Nayak S, Kakar R, Chaudhari UK, Joshi D, Vundinti BR, Fernandes G, Barai RS, Kholkute SD and Sachdeva G "A triad of telomerase, androgen receptor and early growth response 1 in prostate cancer cells." Cancer biology & therapy 17, 4 (2016): 439-448.
6. http://www.genecards.org/cgi-bin/carddisp.pl?gene=CTCF
7. Gao J, Aksoy BA, Dogrusoz U, Dresdner G, Gross B, Sumer SO, Sun Y, Jacobsen A, Sinha R, Larsson E, Cerami E, Sander C and Schultz N "Integrative analysis of complex cancer genomics and clinical profiles using the cBioPortal." Science signaling 6, 269 (2013): pl1.
8. Beltran H, Prandi D, Mosquera JM, Benelli M, Puca L, Cyrta J, Marotz C, Giannopoulou E, Chakravarthi BV, Varambally S, Tomlins SA, Nanus DM, Tagawa ST,, Van Allen EM, Elemento O, Sboner A, Garraway LA, Rubin MA and Demichelis F "Divergent clonal evolution of castration-resistant neuroendocrine prostate cancer." Nature medicine 3 (2016): 298-305.
9. Braun DA, Burke KP, and Van Allen EM. "Genomic Approaches to Understanding Response and Resistance to Immunotherapy." Clinical Cancer Research 22, 23 (2016): 5642-5650.