Efficacy and immune modulation by BXCL701 a dipeptidyl peptidase inhibitor, NKTR-214 a CD122-biased immune agonist with PD1 blockade in murine pancreatic tumors

Luca Rastelli[1], Snigdha Gupta[2], Zeenia Jagga[2], Deborah H. Charych[3], Jonathan Zalevsky[3];

[1]Bioxcel therapeutics, Branford, CT; [2]Bioxcel corporation, Gurugram, India | [3]Nektar Therapeutics, San Francisco, CA

BACKGROUND

- BXCL701 is the outcome of an AI based proprietary Discovery platform EvolverAI, a text mining consortium of targets that may play a role in immunomodulation in the oncology setting. BXCL701 (Talabostat) is a dipeptidyl peptidase inhibitor that targets the dipeptidyle peptidases DPP8 /9 and FAP.
- DPP8/9 and FAP are the members of the dipeptidyl peptidase IV (DPPIV) enzyme family, implicated in tumorigenesis and tumor stromal remodeling. BXCL701 inhibition of these dipeptidases stimulates the migration and cytotoxicity of effector T cells as well as NK cells in addition to targeting fibroblast activator protein (FAP). This forms an immunological barrier to the tumor microenvironment, thus addressing the innate arm of the immune system.
- In the MC38 syngeneic mouse model BXCL701 has been shown to inhibit cancer growth with concomitant up-regulation of immuno-stimulatory cytokines and tumor infiltration of anti-cancer cell types including CD8+ T cells, M1 macrophages and NK cells when combined with the immune check-point inhibitor (ICI) anti-PD[1].
- NKTR-214 is a CD122-biased agonist designed to provide sustained signaling through the heterodimeric IL2receptor pathway (IL2Rβ) to preferentially activate and expand effector CD8+ T cells, which comprise the adaptive arm of the immune system. NKTR-214 has demonstrated robust anti-tumor activity when combined with anti-PD1 in multiple murine tumor models and recently with nivolumab in multiple human cancers[2,3].
- Thus, the addition of BXCL701 to the combination of NKTR-214 and anti-PD1 could further potentiate the immune mediated anti-cancer activity of this combination by removing fibrotic barriers to immune cells. Additionally, harnessing both the innate and adaptive arms of the immune system to overcome an immune-refractory tumor microenvironment.
- The work presented here establishes this rationale of synergistic therapeutic activity of the triple combination and encourages the translation of this strategy to patients who are non-responsive to ICI therapy.

1. Rastelli L., etal., In: Proceedings AACR Annual Meeting 2017; Cancer Res 2017;77(13 Suppl):Abstract nr 2629
2. Charych D etal., PLoS One. 2017 Jul 5;12(7):e0179431.
3. Adi Diab etal., Presented at SITC 2017, Maryland, Poster #O20

BXCL701 and NKTR-214 Induce Innate and Adaptive Immune Activation

Earlier data indicates BXCL701's capacity to stimulate neutrophil generation in cyclophosphamide (CYP) treated Balb/c mice mouse model of neutropenia. BXCL701 showed a dose dependent acceleration in the recovery of ANCs (absolute neutrophil counts) to normal levels indicating its capacity to harness the innate immune cells[4].

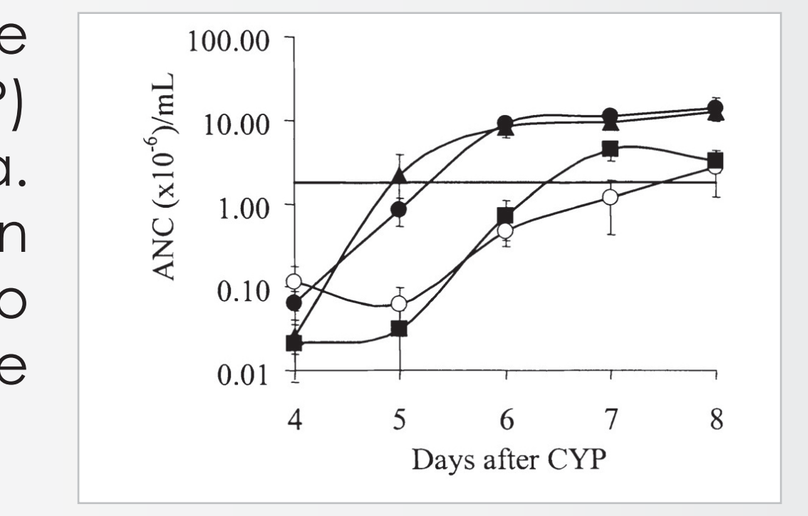
ANC (x10^6)/mL
100.00
10.00
1.00
0.10
0.01
4 5 6 7 8
Days after CYP

BALB/c mice were injected intraperitoneally with CYP (220 mg/kg) on day 0 and were administered saline or BXCL701 orally and twice daily, starting on day 3. Changes in ANCs in mice receiving 0.1 µg(▪), 2 µg(•), or 5 µg (▲) BXCL701 or saline (○) for 5 days.

NKTR-214 has demonstrated a sustained activation of pSTAT5+ CD3+ cells[2] as compared to aldesleukin and tumor-killing with a ratio of CD8+ T cells to Foxp3+regulatory T cells > 400 in a murine melanoma tumor model5, indicating stimulation of the adaptive arm of the immune system[5].

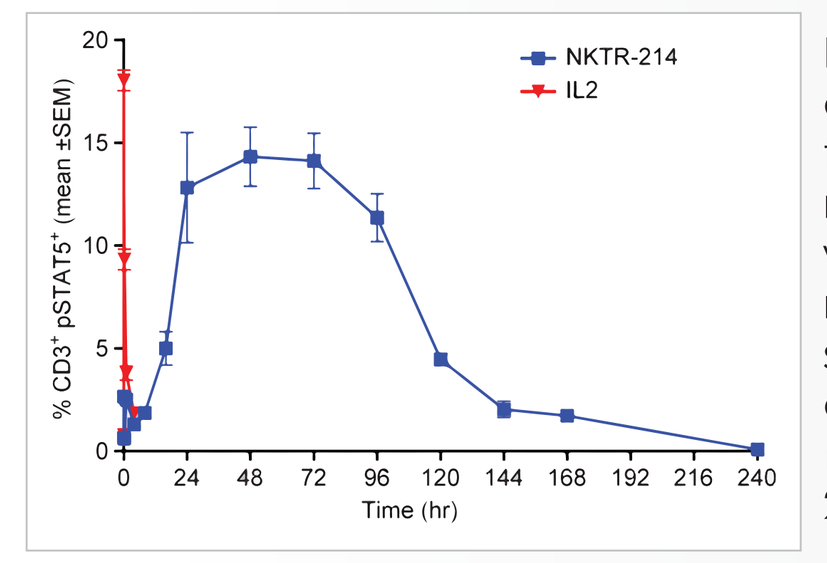
NKTR-214
IL2
% CD3+ pSTAT5+ (mean ±SEM)
0 24 48 72 96 120 144 168 192 216 240
Time (hr)

NKTR-214 delivers a controlled and sustained activation signal to the IL2 pathway in vivo measured by pSTAT5 levels in whole peripheral blood from mice (n=5) were treated with a single 0.8 mg/kg dose of either aldesleukin (red) or NKTR-214 (blue) and monitored for up to 240 hours.

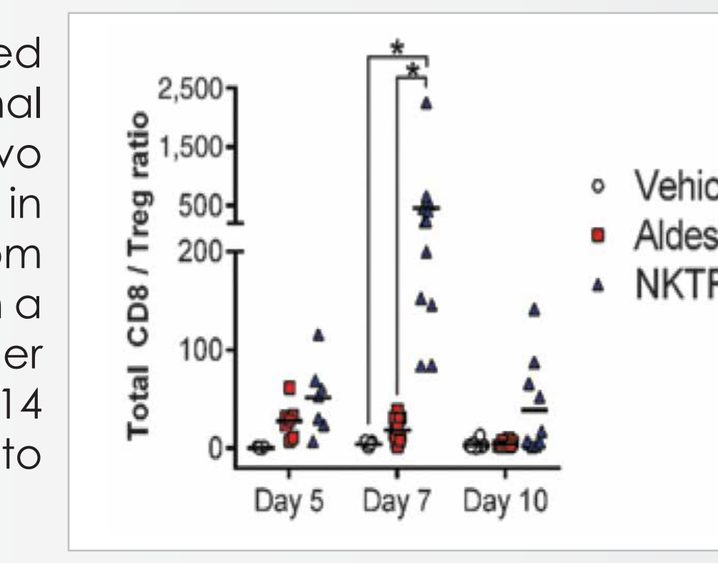
Total CD8 / Treg ratio
Vehicle
Aldesleukin
NKTR-214
Day 5 Day 7 Day 10

NKTR-214 led to significant decrease in Treg percentages along with increase in CD8+T cell infilktration, leading to higher CD8/Treg ratios of >400 compared with aldesleukin (ratio of 18) and vehicle (ratio of 4).

4. Jones B. etal., Blood 2003 102:1641-1648
5. Charych, D.H.et al., Clin Cancer Res; 22(3); 680–90. ©2016 AACR.

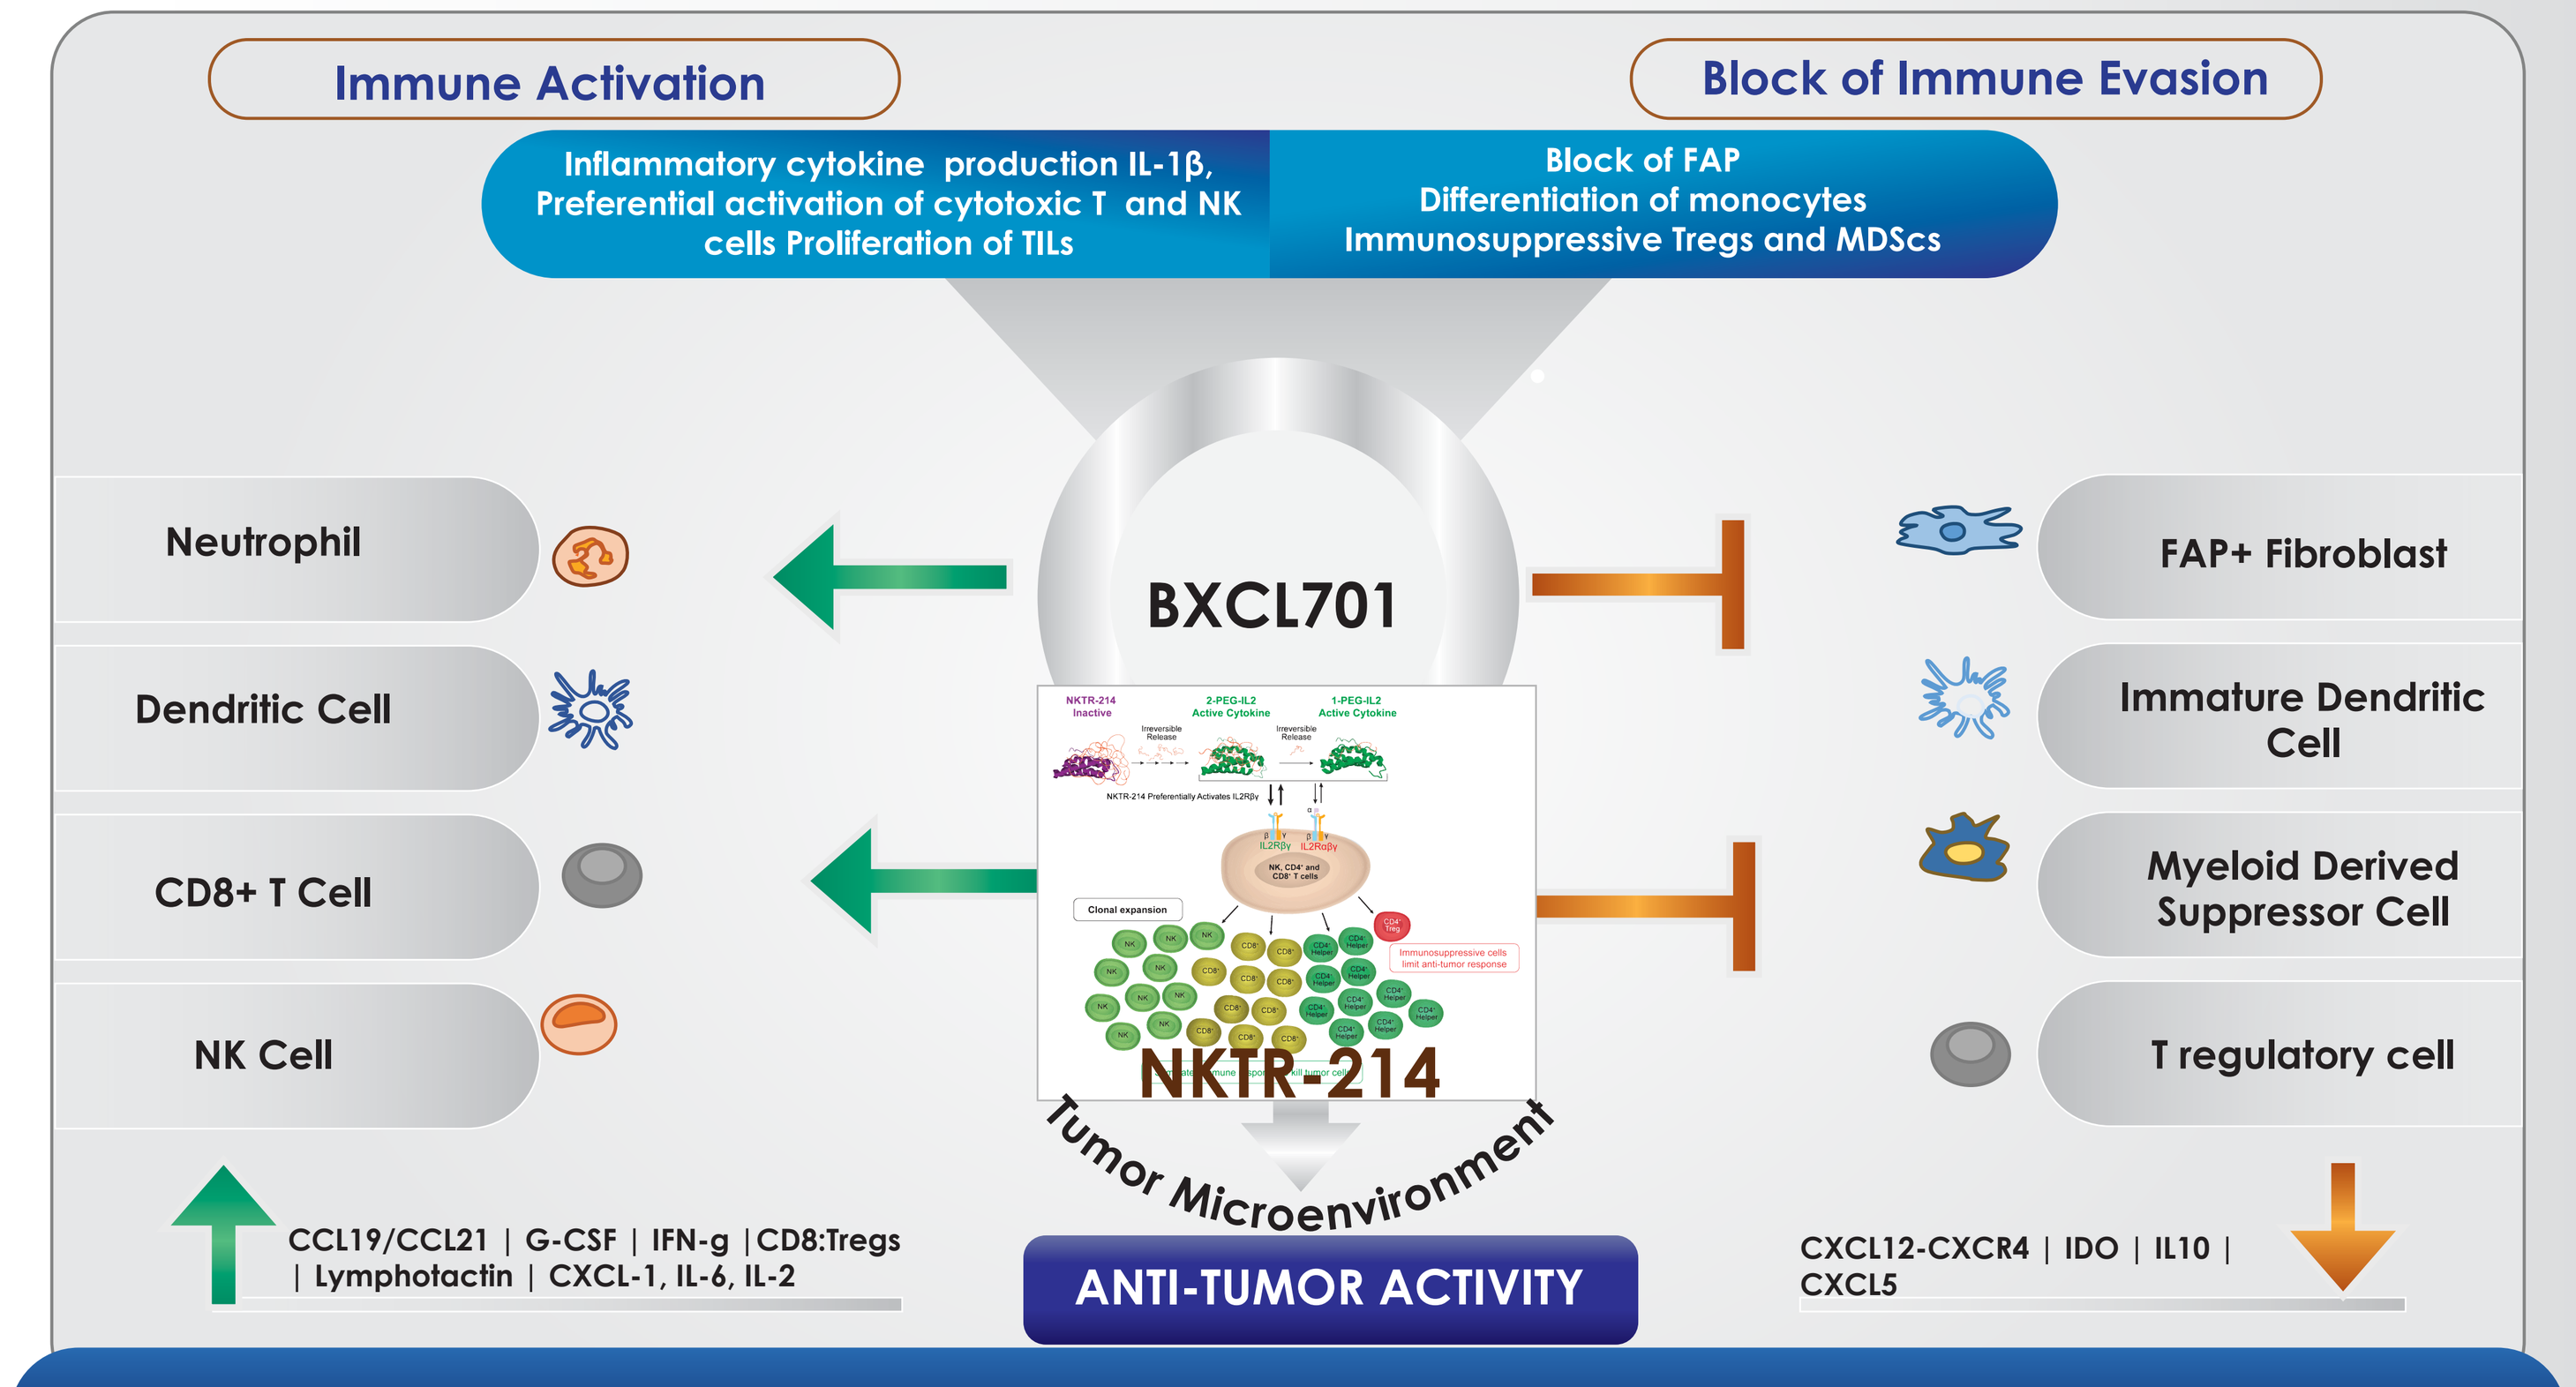
Immune Activation
Block of Immune Evasion
Inflammatory cytokine production IL-1β, Preferential activation of cytotoxic T and NK cells Proliferation of TILs
Block of FAP Differentiation of monocytes Immunosuppressive Tregs and MDScs
Neutrophil
Dendritic Cell
CD8+ T Cell
NK Cell
BXCL701
NKTR-214
Tumor Microenvironment
FAP+ Fibroblast
Immature Dendritic Cell
Myeloid Derived Suppressor Cell
T regulatory cell
CCL19/CCL21 | G-CSF | IFN-g | CD8:Tregs | Lymphotactin | CXCL-1, IL-6, IL-2
ANTI-TUMOR ACTIVITY
CXCL12-CXCR4 | IDO | IL10 | CXCL5

BXCL701 Induces Immune Activation and Blocks the Immuno-evasion

HYPOTHESIS

BXCL701 combination with NKTR-214 and anti-PD1 potentiates anti-tumor T cell responses

FAP inhibition permits T-cell infiltration into the tumor by blocking immune suppressive cytokines

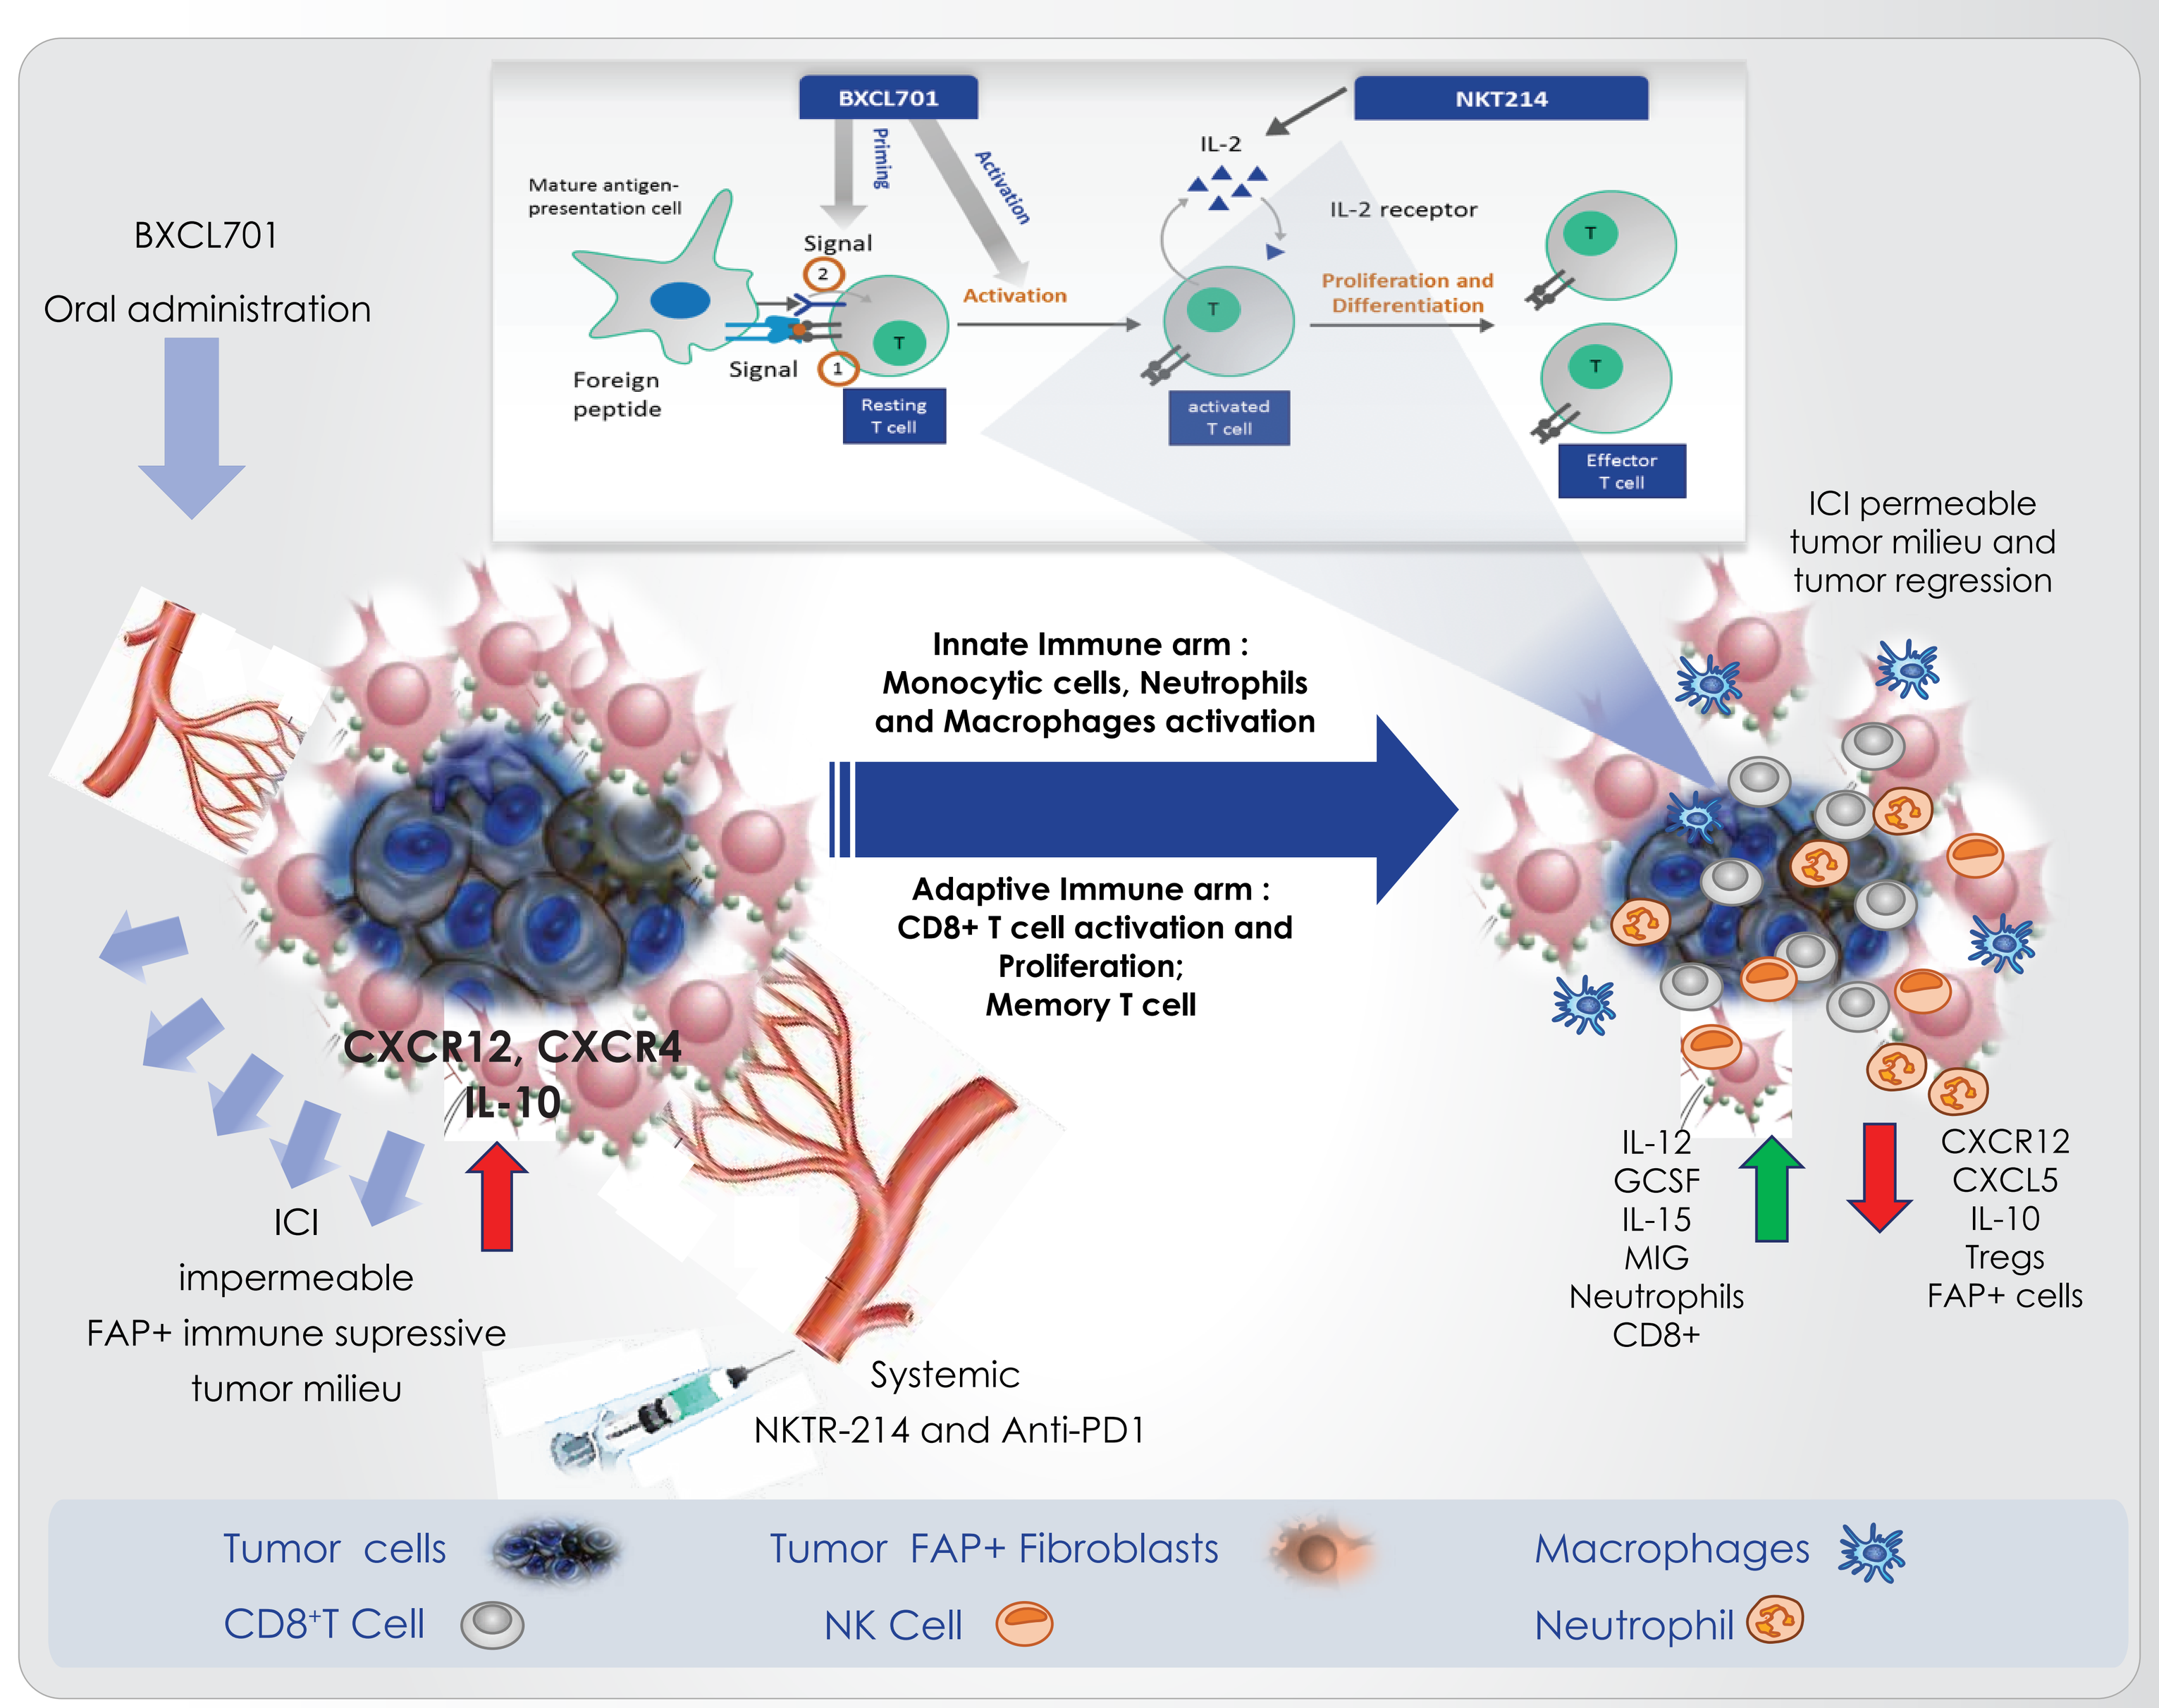
BXCL701
Oral administration
BXCL701
Priming
Activation
NKT214
Mature antigen-presentation cell
Signal
Foreign peptide
Resting T cell
IL-2
IL-2 receptor
Proliferation and Differentiation
activated T cell
Effector T cell
ICI permeable tumor milieu and tumor regression
Innate Immune arm : Monocytic cells, Neutrophils and Macrophages activation
Adaptive Immune arm : CD8+ T cell activation and Proliferation; Memory T cell
CXCR12, CXCR4
IL-10
ICI impermeable FAP+ immune supressive tumor milieu
Systemic NKTR-214 and Anti-PD1
IL-12
GCSF
IL-15
MIG
Neutrophils
CD8+
CXCR12
CXCL5
IL-10
Tregs
FAP+ cells
Tumor cells
Tumor FAP+ Fibroblasts
Macrophages
CD8+T Cell
NK Cell
Neutrophil

METHODOLOGY

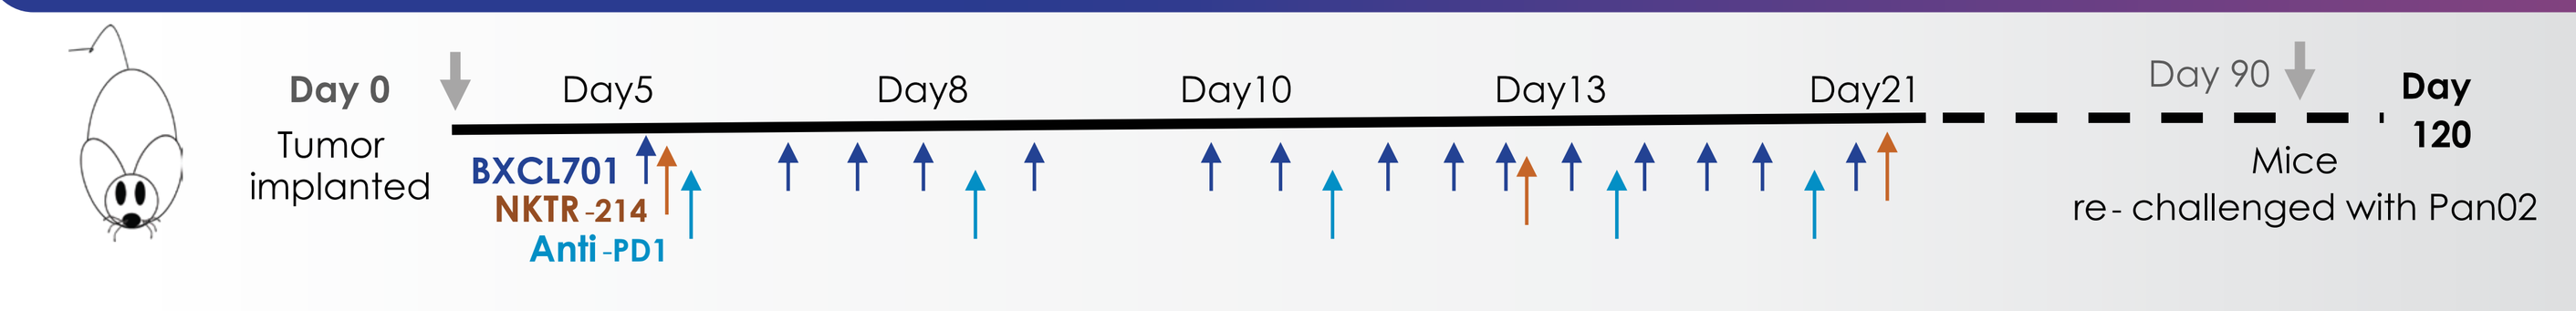
Day 0
Tumor implanted
Day5
Day8
Day10
Day13
Day21
Day 90
Day 120
Mice re- challenged with Pan02
BXCL701
NKTR-214
Anti-PD1

Tumor model: Pancreatic adenocarcinoma , Pan02
Tumor implant: Each mouse was inoculated subcutaneously at the front right flank region with Pan02 cells (3 x 10^6) in 0.1 ml of PBS. The date of tumor cell inoculation was denoted as day 0. Five days following tumor implant, mice were randomized into groups of 12 mice/group with a mean tumor volume of ~ 140 mm^3. The test articles were administered according to the doses and schedules described in Table 1.
Treatments: Control (Vehicle), NKTR-214 (0.8 mg/kg Q9D), BXCL701 (20 µg QD) and anti-PD1 (10 mg/kg BIW) were dosed as single agents, doublets and the triplet combination in mice bearing established Pan02 tumors.

Group	N	Treatment	Dose (mg/kg)	Dosing Route	Schedule
1	12	NKTR-214 vehicle	0	*i.v.*	Q9D
		BXCL701 vehicle		*p.o.*	QD
		Anti-PD1 vehicle		*i.p.*	Twice weekly x 3 weeks
2	12	BXCL701	20 µg/dose	*p.o.*	QD
3	12	Anti-PD1	0.8	*i.v.*	Q9D
4	12	Anti-PD1	10	*i.p.*	Twice weekly x 3 weeks
5	12	BXCL701	20 µg/dose	*p.o.*	QD
		NKTR-214	0.8	*i.v.*	Q9D
6	12	BXCL701	20 µg/dose	*p.o.*	QD
		Anti-PD1	10	*i.p.*	Twice weekly x 3 weeks
7	12	NKTR-214	0.8	*i.v.*	Q9D
		Anti-PD1	10	*i.p.*	Twice weekly x 3 weeks
8	12	NKTR-214	0.8	*i.v.*	Q9D
		BXCL701	20 µg/dose	*p.o.*	QD
		Anti-PD1	10	*i.p.*	Twice weekly x 3 weeks

N : number of mice, Q9D: once in 9 days, BIW: twice per week , QD: once daily. Of the 12 mice in each group, three from each were sacrificed three days after the first dose of treatment was provided

METHODOLOGY

Protocol: Tumor size and body weights were measured twice weekly. Tumors were profiled using IHC from satellite animals (3 out of 12 mice) sacrificed 3 days after the first dosing. Tissue sections were stained with anti-mouse FAP Ab (ab53066, Abcam), anti-mouse CD8 Ab (14-0808-80, eBiosciences), anti-mouse Ly6G Ab (BE0075-1, Bioxcell) and H&E staining (6765009, ThermoFisher) according to the manufacturer's protocol. Multiplex serum cytokine/chemokine analysis (using MILLIPLEX® MAP, Merck Millipore) was performed on plasma collected on day 5 after tumor inoculation (pre-treatment) and day 7 after the dosing (post-treatment) from tumor bearing animals treated with the triple combination. For the re-challenge experiment the tumor-free mice were re-challenged with of 3x10^6 Pan02 cells and tumor size was measured in the mentioned time period.

KEY FINDINGS ...

1. Tumor regression for BXCL701 and NKTR-214 with anti-PD1 in Pan02 mouse model
The mice treated with BXCL701 20 µg QD, in combination with NKTR-214 (0.8 mg/kg; Q9D) and anti-PD-1 (10 mg/kg; BIW) exhibited significant tumor reduction as noted from Day 10 onwards as compared to anti-PD-1 as well as vehicle control (table insert and Fig 1) in the syngeneic mouse model of pancreatic adenocarcinoma Pan02. While BXCL701 alone showed a significant effect, the triple combination resulted in complete regression of the tumor.
Of the mice treated with the triplet combination, 100% (9 of 9) became tumor-free by day 26. These animals remained tumor free for more than 60 days.

Figure1 : Anti-tumor efficacy of the BXCL701+NKTR-214+Anti-PD1 in comparison to the respective combinations and as single agents as tested till day 29 in the Pan02 syngeneic mouse model of pancreatic adenocarcinoma

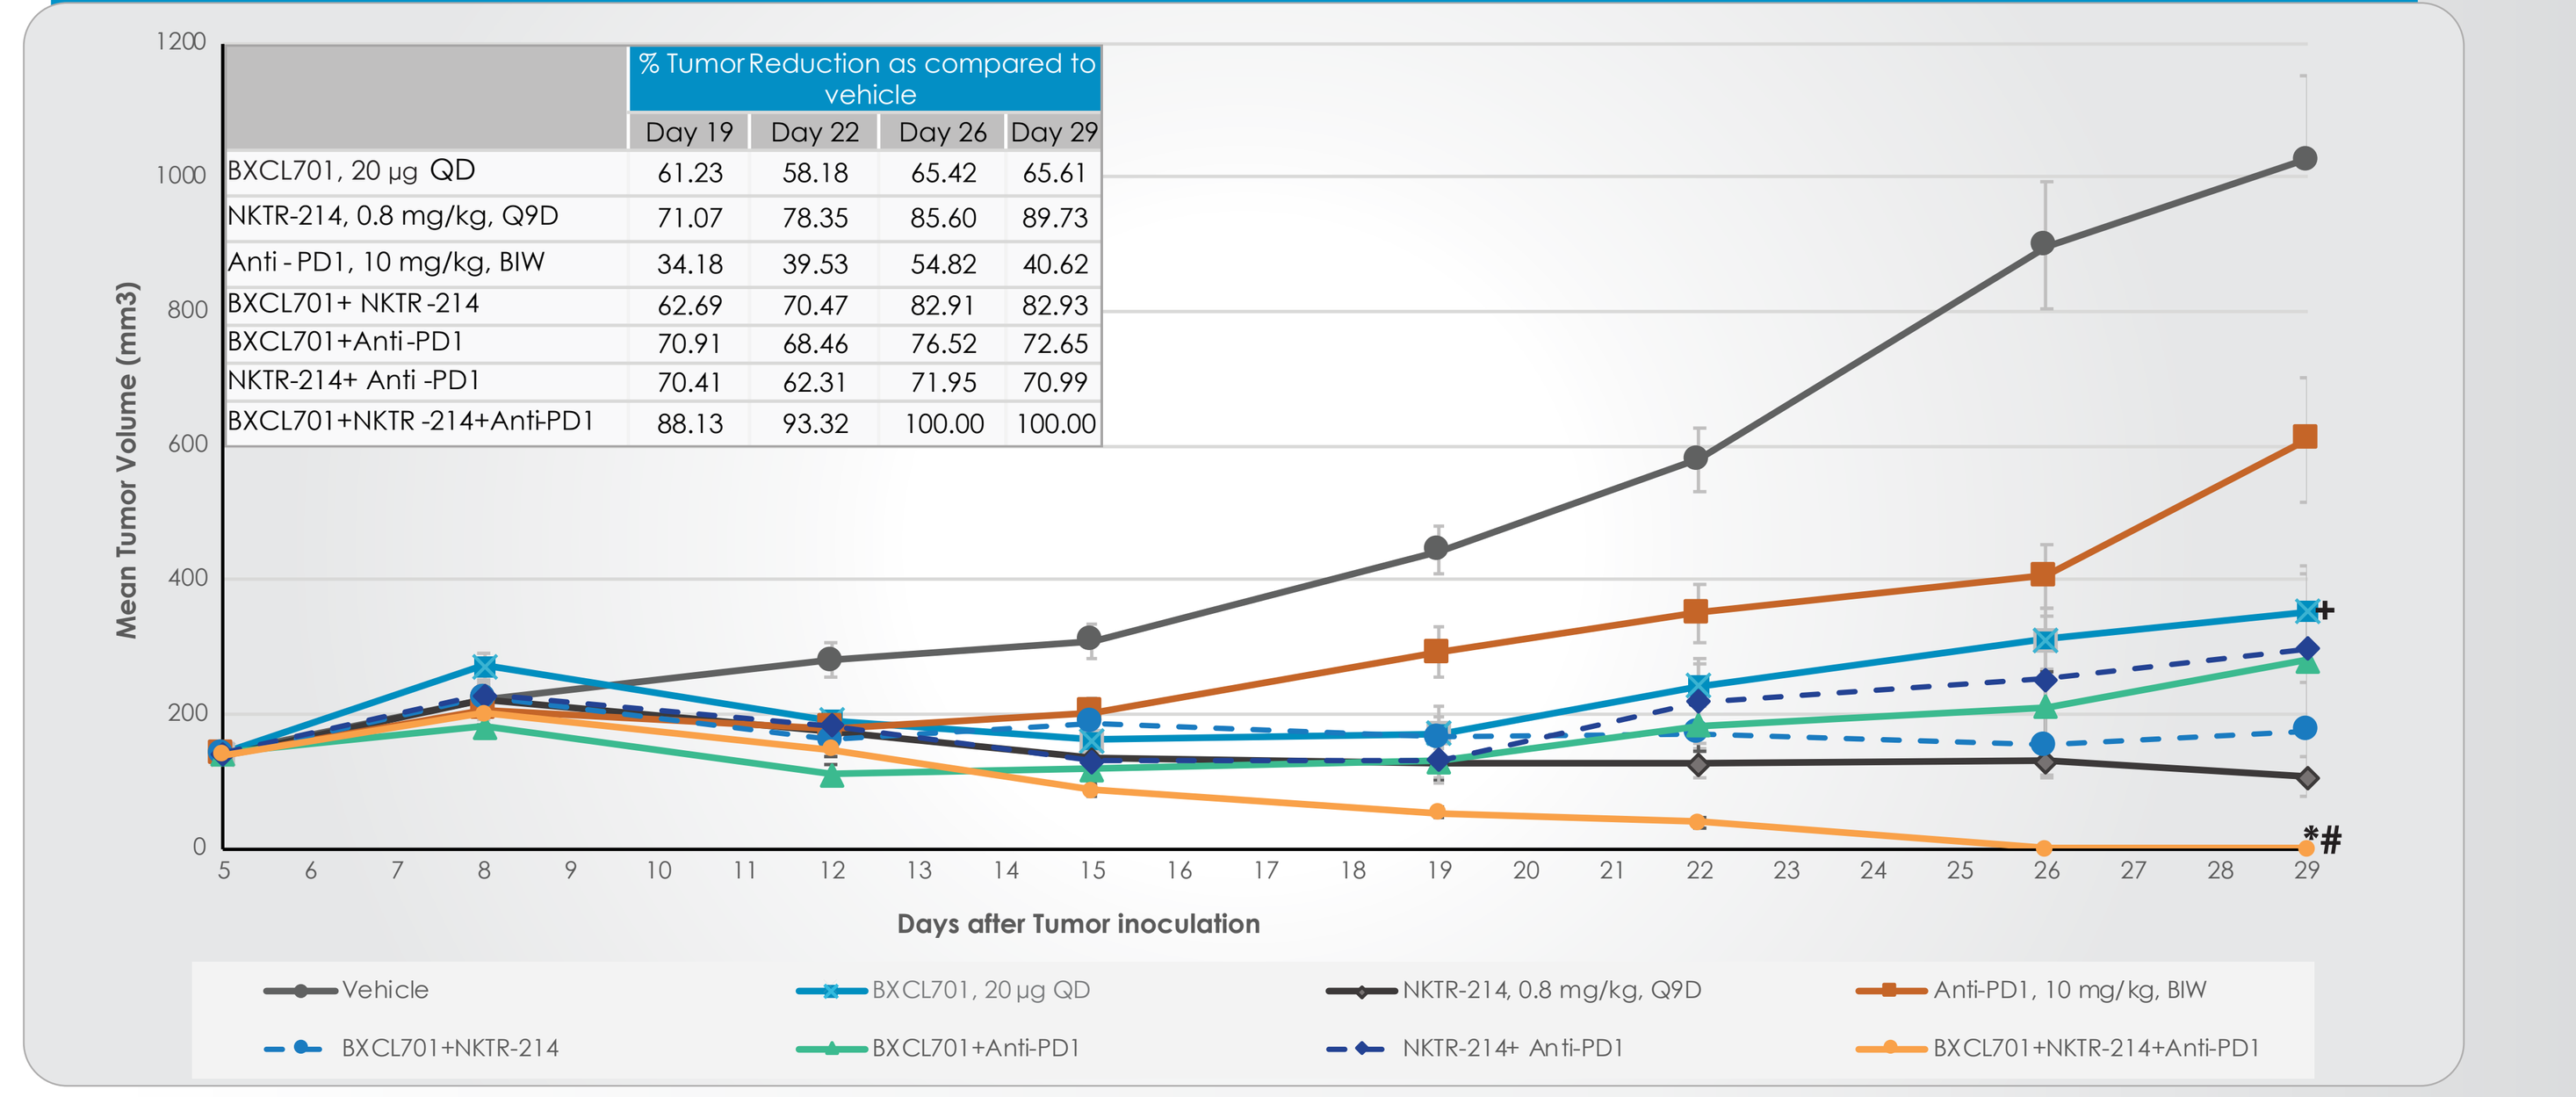

	% Tumor Reduction as compared to vehicle			
	Day 19	Day 22	Day 26	Day 29
BXCL701, 20 µg QD	61.23	58.18	65.42	65.61
NKTR-214, 0.8 mg/kg, Q9D	71.07	78.35	85.60	89.73
Anti - PD1, 10 mg/kg, BIW	34.18	39.53	54.82	40.62
BXCL701+ NKTR -214	62.69	70.47	82.91	82.93
BXCL701+Anti-PD1	70.91	68.46	76.52	72.65
NKTR-214+ Anti -PD1	70.41	62.31	71.95	70.99
BXCL701+NKTR -214+Anti-PD1	88.13	93.32	100.00	100.00

Mean Tumor Volume (mm3)
Days after Tumor inoculation
Vehicle
BXCL701, 20 µg QD
NKTR-214, 0.8 mg/kg, Q9D
Anti-PD1, 10 mg/kg, BIW
BXCL701+NKTR-214
BXCL701+Anti-PD1
NKTR-214+ Anti-PD1
BXCL701+NKTR-214+Anti-PD1

*The triple combination shows a p<0.001 as compared to BXCL701 and Anti-PD-1 as single agents
#p<0.05, when compared to the combinations and single agent
+p<0.05, for BXCL701+ Anti-PD-1 vs PD-1 antagonist

2.BXCL701, NKTR-214 and anti-PD1 induces anti- immunity
Sixty days after dosing initiation the tumor-free animals exhibiting complete response to combined immunotherapy were enrolled to be re-challenged with 3x106 Pan02 cells. Upon subcutaneous inoculation of a subset of mice with Pan02 pancreatic adenocarcinoma cells 5 of 6 mice rejected tumor growth demonstrating durable immunity and generation of anti-tumor memory after the triplet therapy (Figure 2 insert)

Figure 2: In vivo anti-tumor immunity established for Triple Combination

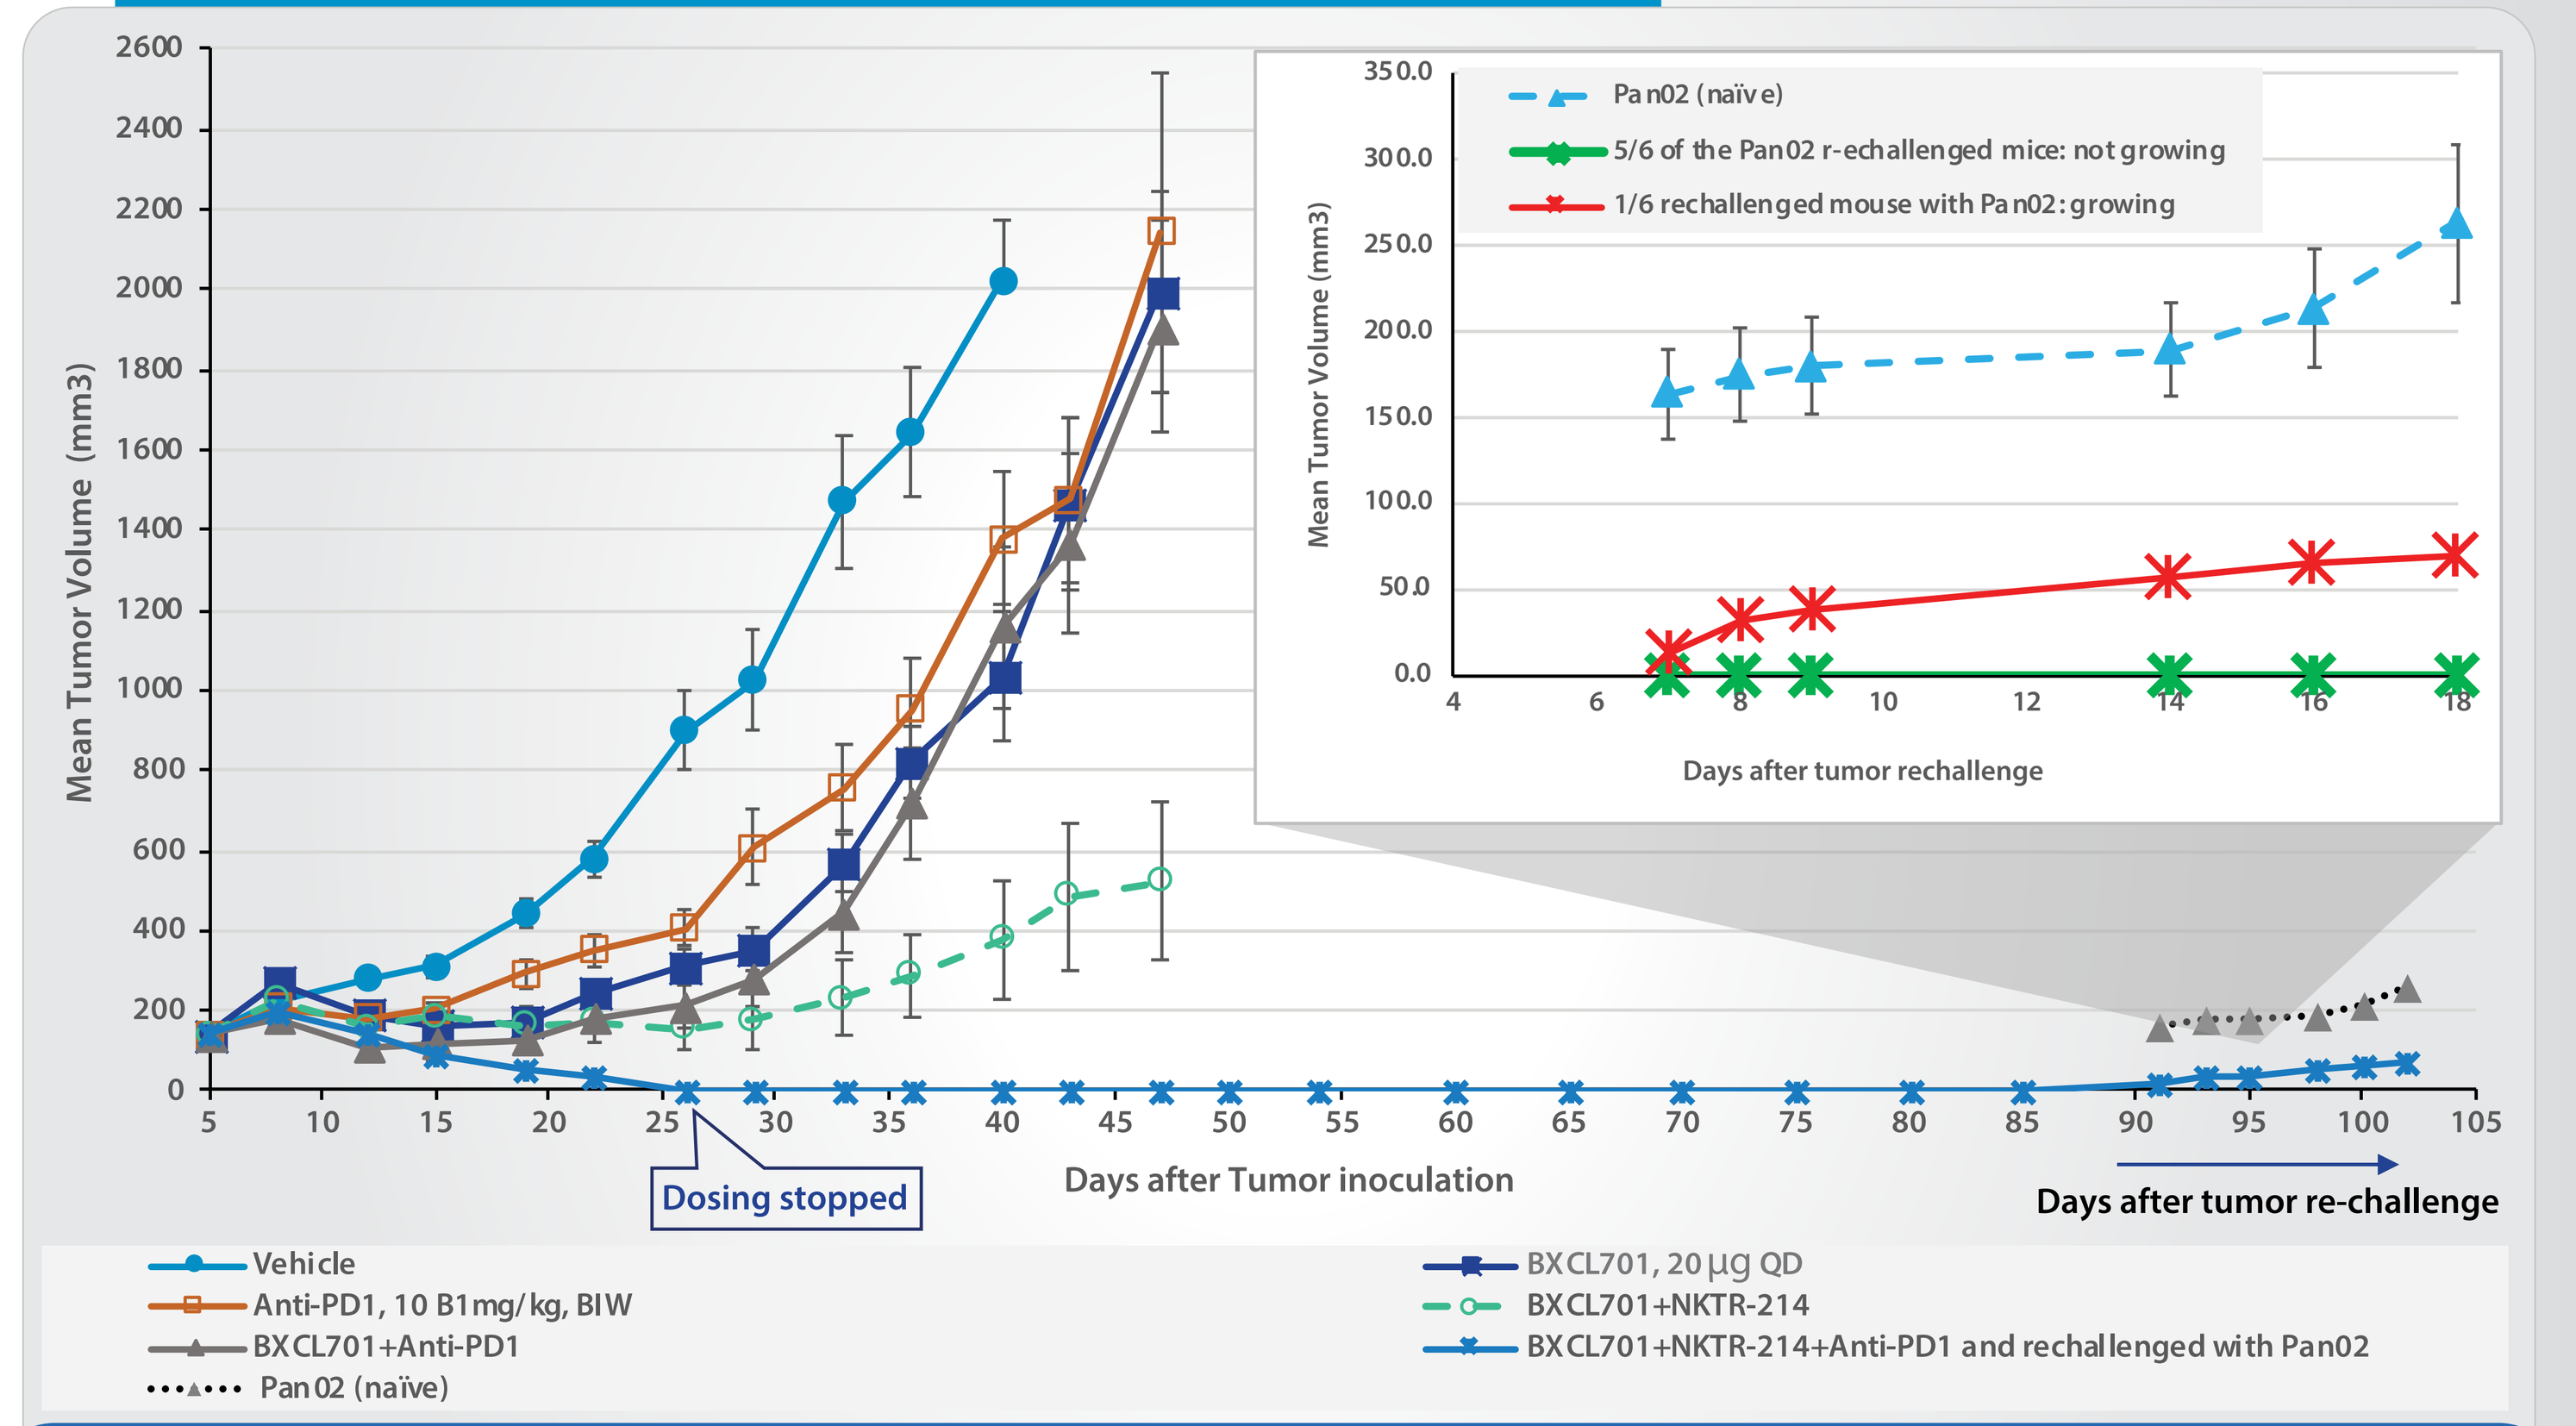
Mean Tumor Volume (mm3)
Days after Tumor inoculation
Dosing stopped
Days after tumor re-challenge
Pan02 (naïve)
5/6 of the Pan02 r-echallenged mice: not growing
1/6 rechallenged mouse with Pan02: growing
Days after tumor rechallenge
Vehicle
Anti-PD1, 10 B1mg/kg, BIW
BXCL701+Anti-PD1
Pan02 (naïve)
BXCL701, 20 µg QD
BXCL701+NKTR-214
BXCL701+NKTR-214+Anti-PD1 and rechallenged with Pan02

Combo with anti-PD1 and NKTR-214 stimulates immune system "curing" mice and making them resistant to new tumors

KEY FINDINGS ...

3. Stimulation of innate immunity, T-cell response and check-point inhibition by the triple combination
A. IHC of the tumors from satellite animals sacrificed on day 3 with anti-mouse FAP Ab, anti-mouse CD8 Ab and anti-mouse Ly6G Ab (procedure as per the manufacturer's protocol revealed that the triplet significantly:

(i) reduced FAP expression,

(ii) increased the number of immune cell infiltrates including Ly6G + neutrophils in the tumor

(iii) increased the number of CD8+ T cells and

(iv) induction of tumor reduction, as also reflected in the H&E staining in the respective groups:

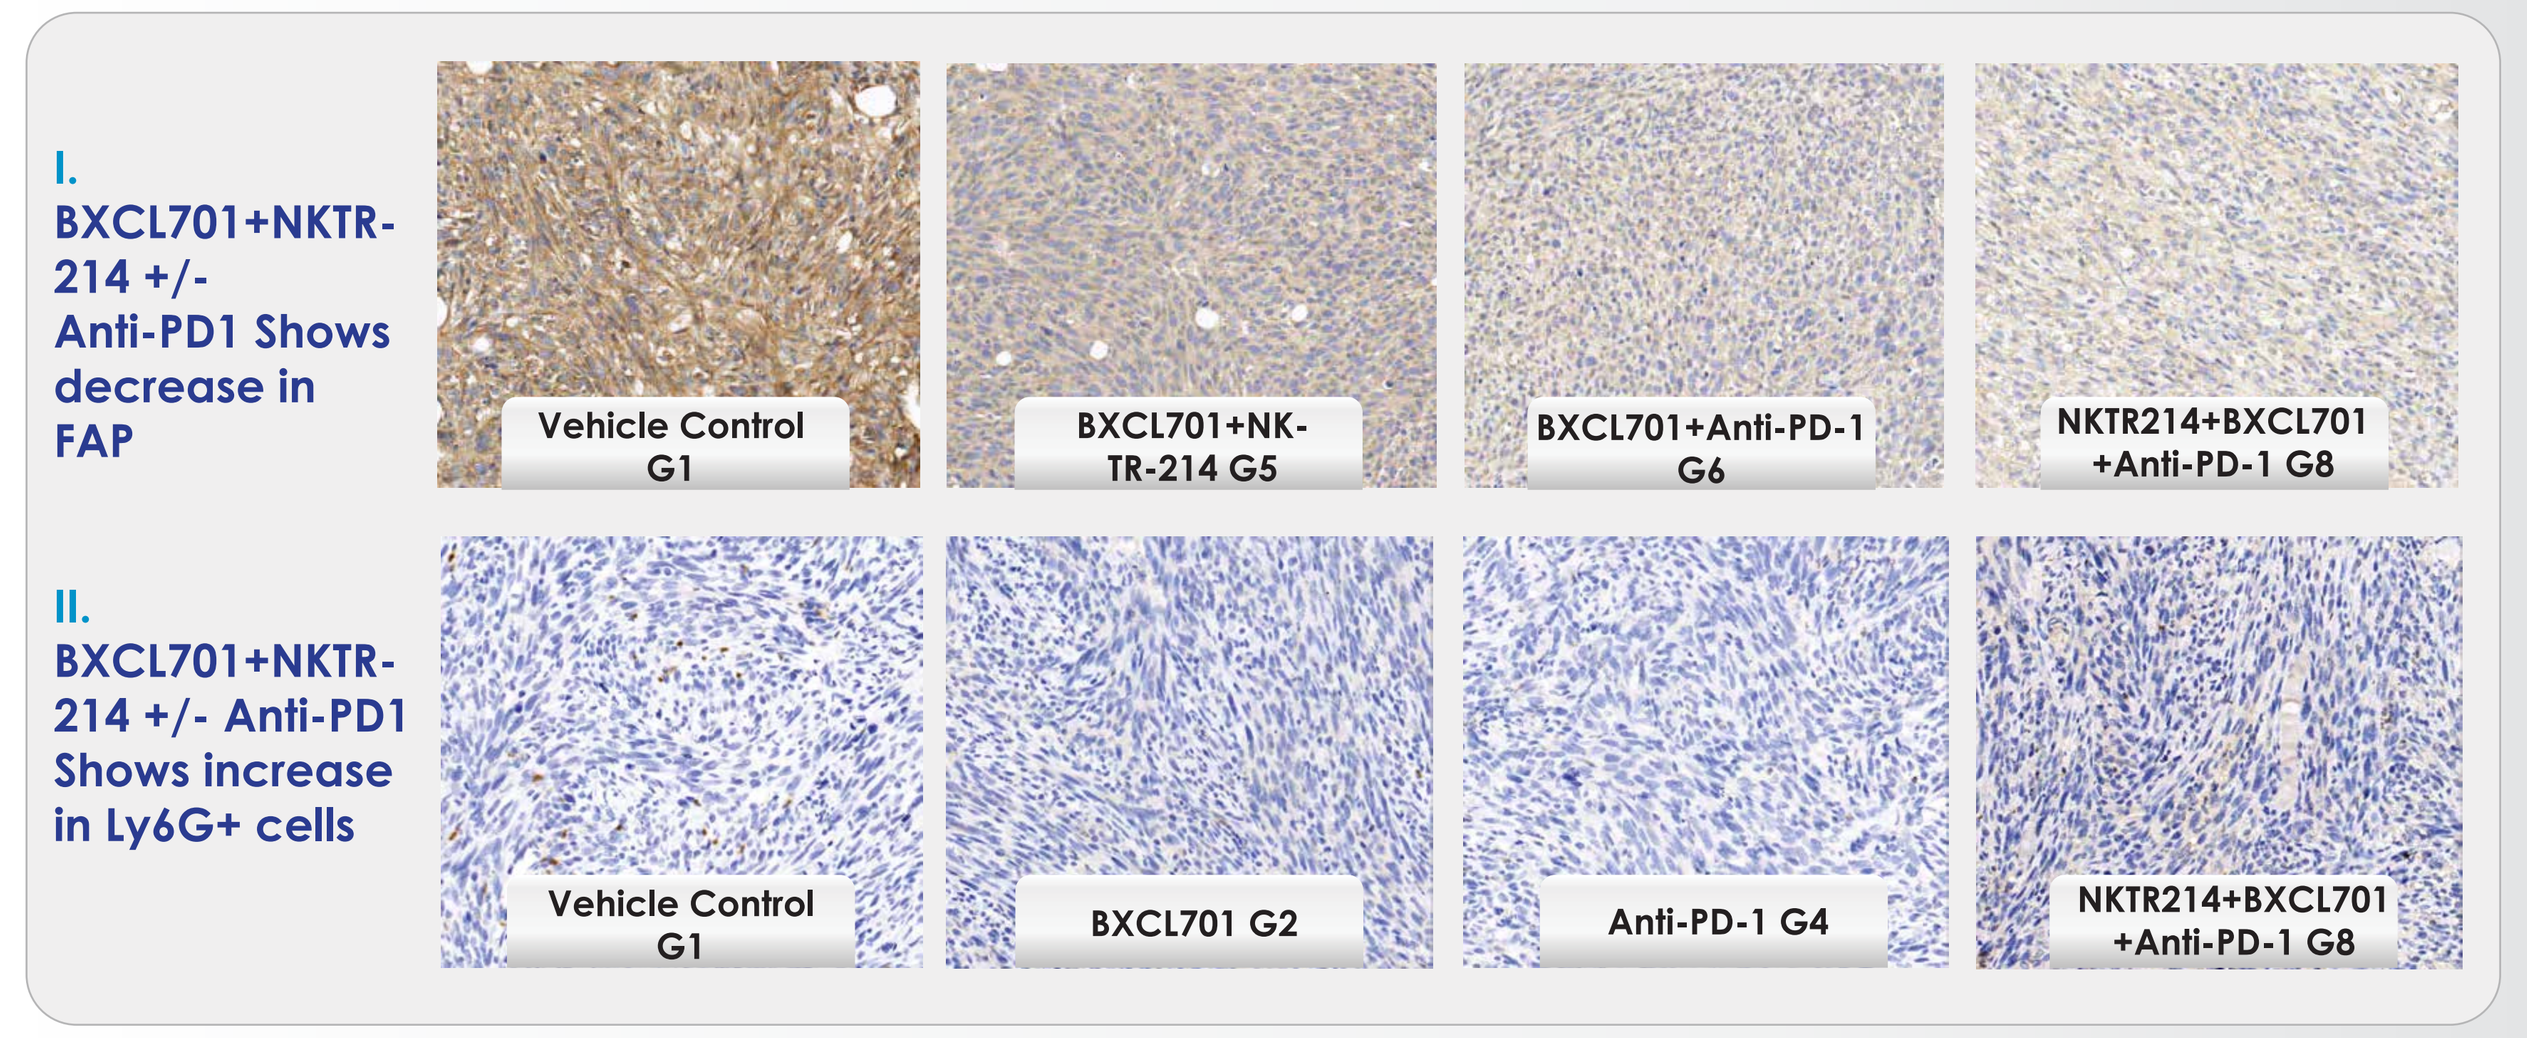
I. BXCL701+NKTR-214 +/- Anti-PD1 Shows decrease in FAP
Vehicle Control G1
BXCL701+NKTR-214 G5
BXCL701+Anti-PD-1 G6
NKTR214+BXCL701 +Anti-PD-1 G8
II. BXCL701+NKTR-214 +/- Anti-PD1 Shows increase in Ly6G+ cells
Vehicle Control G1
BXCL701 G2
Anti-PD-1 G4
NKTR214+BXCL701 +Anti-PD-1 G8

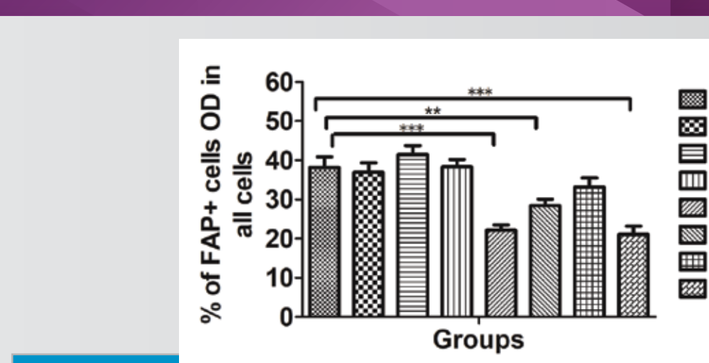
% of FAP+ cells OD in all cells
Groups
G1 G2 G3 G4 G5 G6 G7 G8

BXCL701 combination with NKTR-214 +/- Anti-PD1 shows FAP Reduction

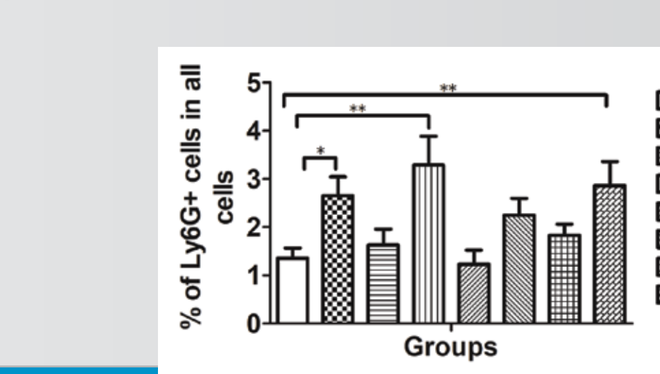
% of Ly6G+ cells in all cells
Groups
G1 G2 G3 G4 G5 G6 G7 G8

BXCL701 alone and when combined with Anti-PD1 as well as with NKTR-214 shows increase in Ly6G+ neutrophils

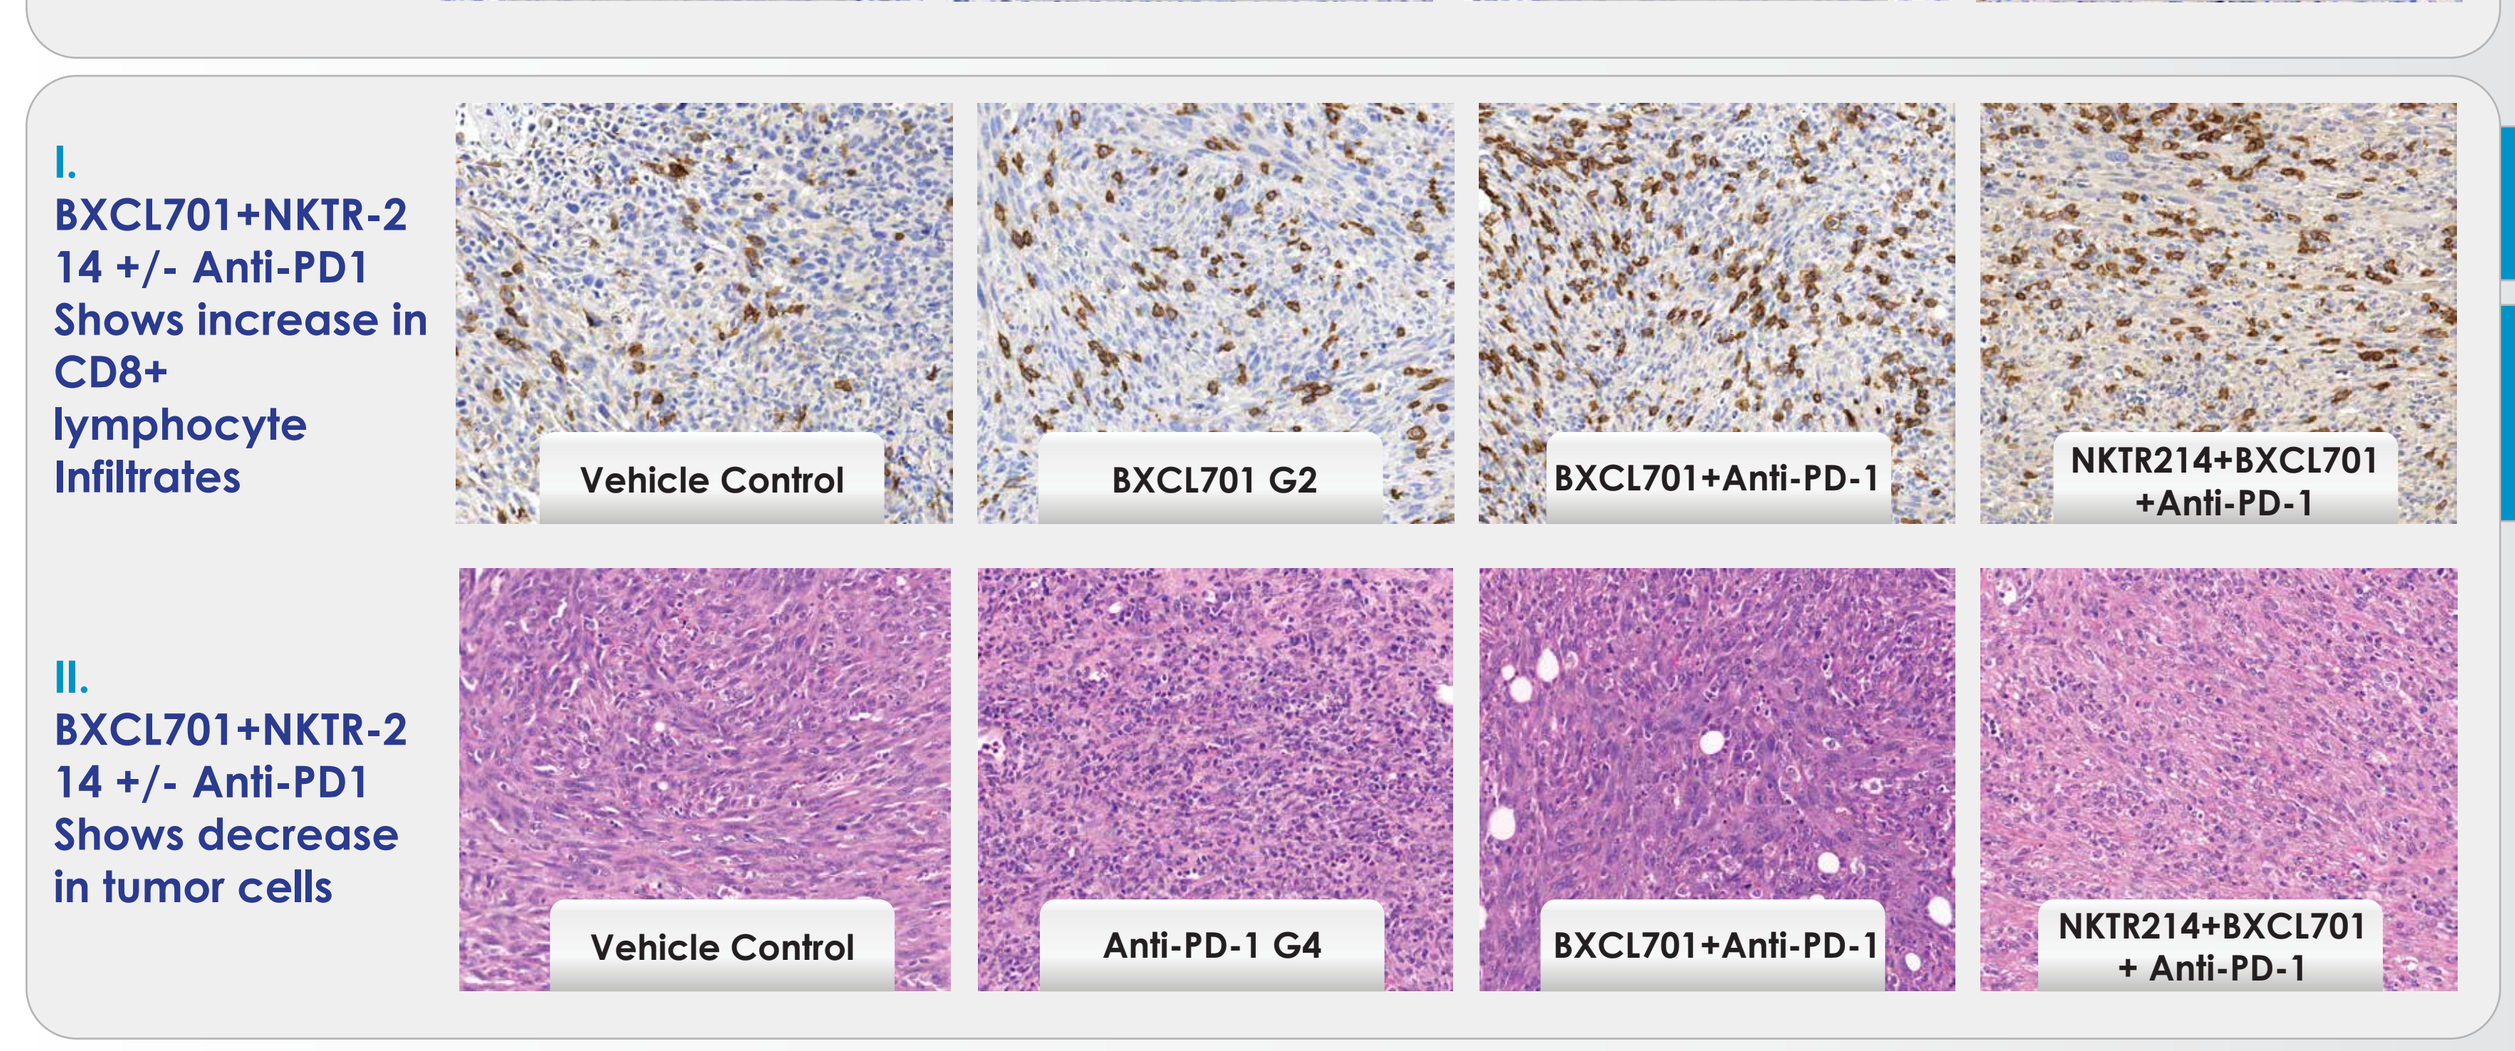
I. BXCL701+NKTR-214 +/- Anti-PD1 Shows increase in CD8+ lymphocyte Infiltrates
Vehicle Control
BXCL701 G2
BXCL701+Anti-PD-1
NKTR214+BXCL701 +Anti-PD-1
II. BXCL701+NKTR-214 +/- Anti-PD1 Shows decrease in tumor cells
Vehicle Control
Anti-PD-1 G4
BXCL701+Anti-PD-1
NKTR214+BXCL701 + Anti-PD-1

The increase in CD8+ T cell infiltration correlates with BXCL701 and NKTR-214 anti-tumor effect

The presence of BXCL701 leads to reduction in tumor cells as reflected in the H&E staining across the groups; prominent in the triple combination group

Group	Group-1	Group-2	Group-3	Group-4	Group-5	Group-6	Group-7	Group-8
Treatment	Vehicle	BXCL701	NKTR-214	Anti-PD-1	BXCL701 +NKTR-214	BXCL701 +Anti-PD1	NKTR-214+Anti-PD1	BXCL701 +NKTR-214+Anti-PD1

B. Luminex analysis of the plasma sample from pre- and post-treatment (day 0 vs day 7) of the mice receiving the triple combination were observed in terms of increase in:

(I) pro-inflammatory cytokines (IL-6, IL-12p40, TNF alpha)
(II) immunostimulatory chemokines (GM-CSF)
(III) cytokines inducing T cell migration (MIG, MIP1-beta)
(IV) cytokines predicting memory T-cell response (IL-15 and IL-7)
(V) Cytokine involved in cell proliferation, invasion and migration (CXCL5)

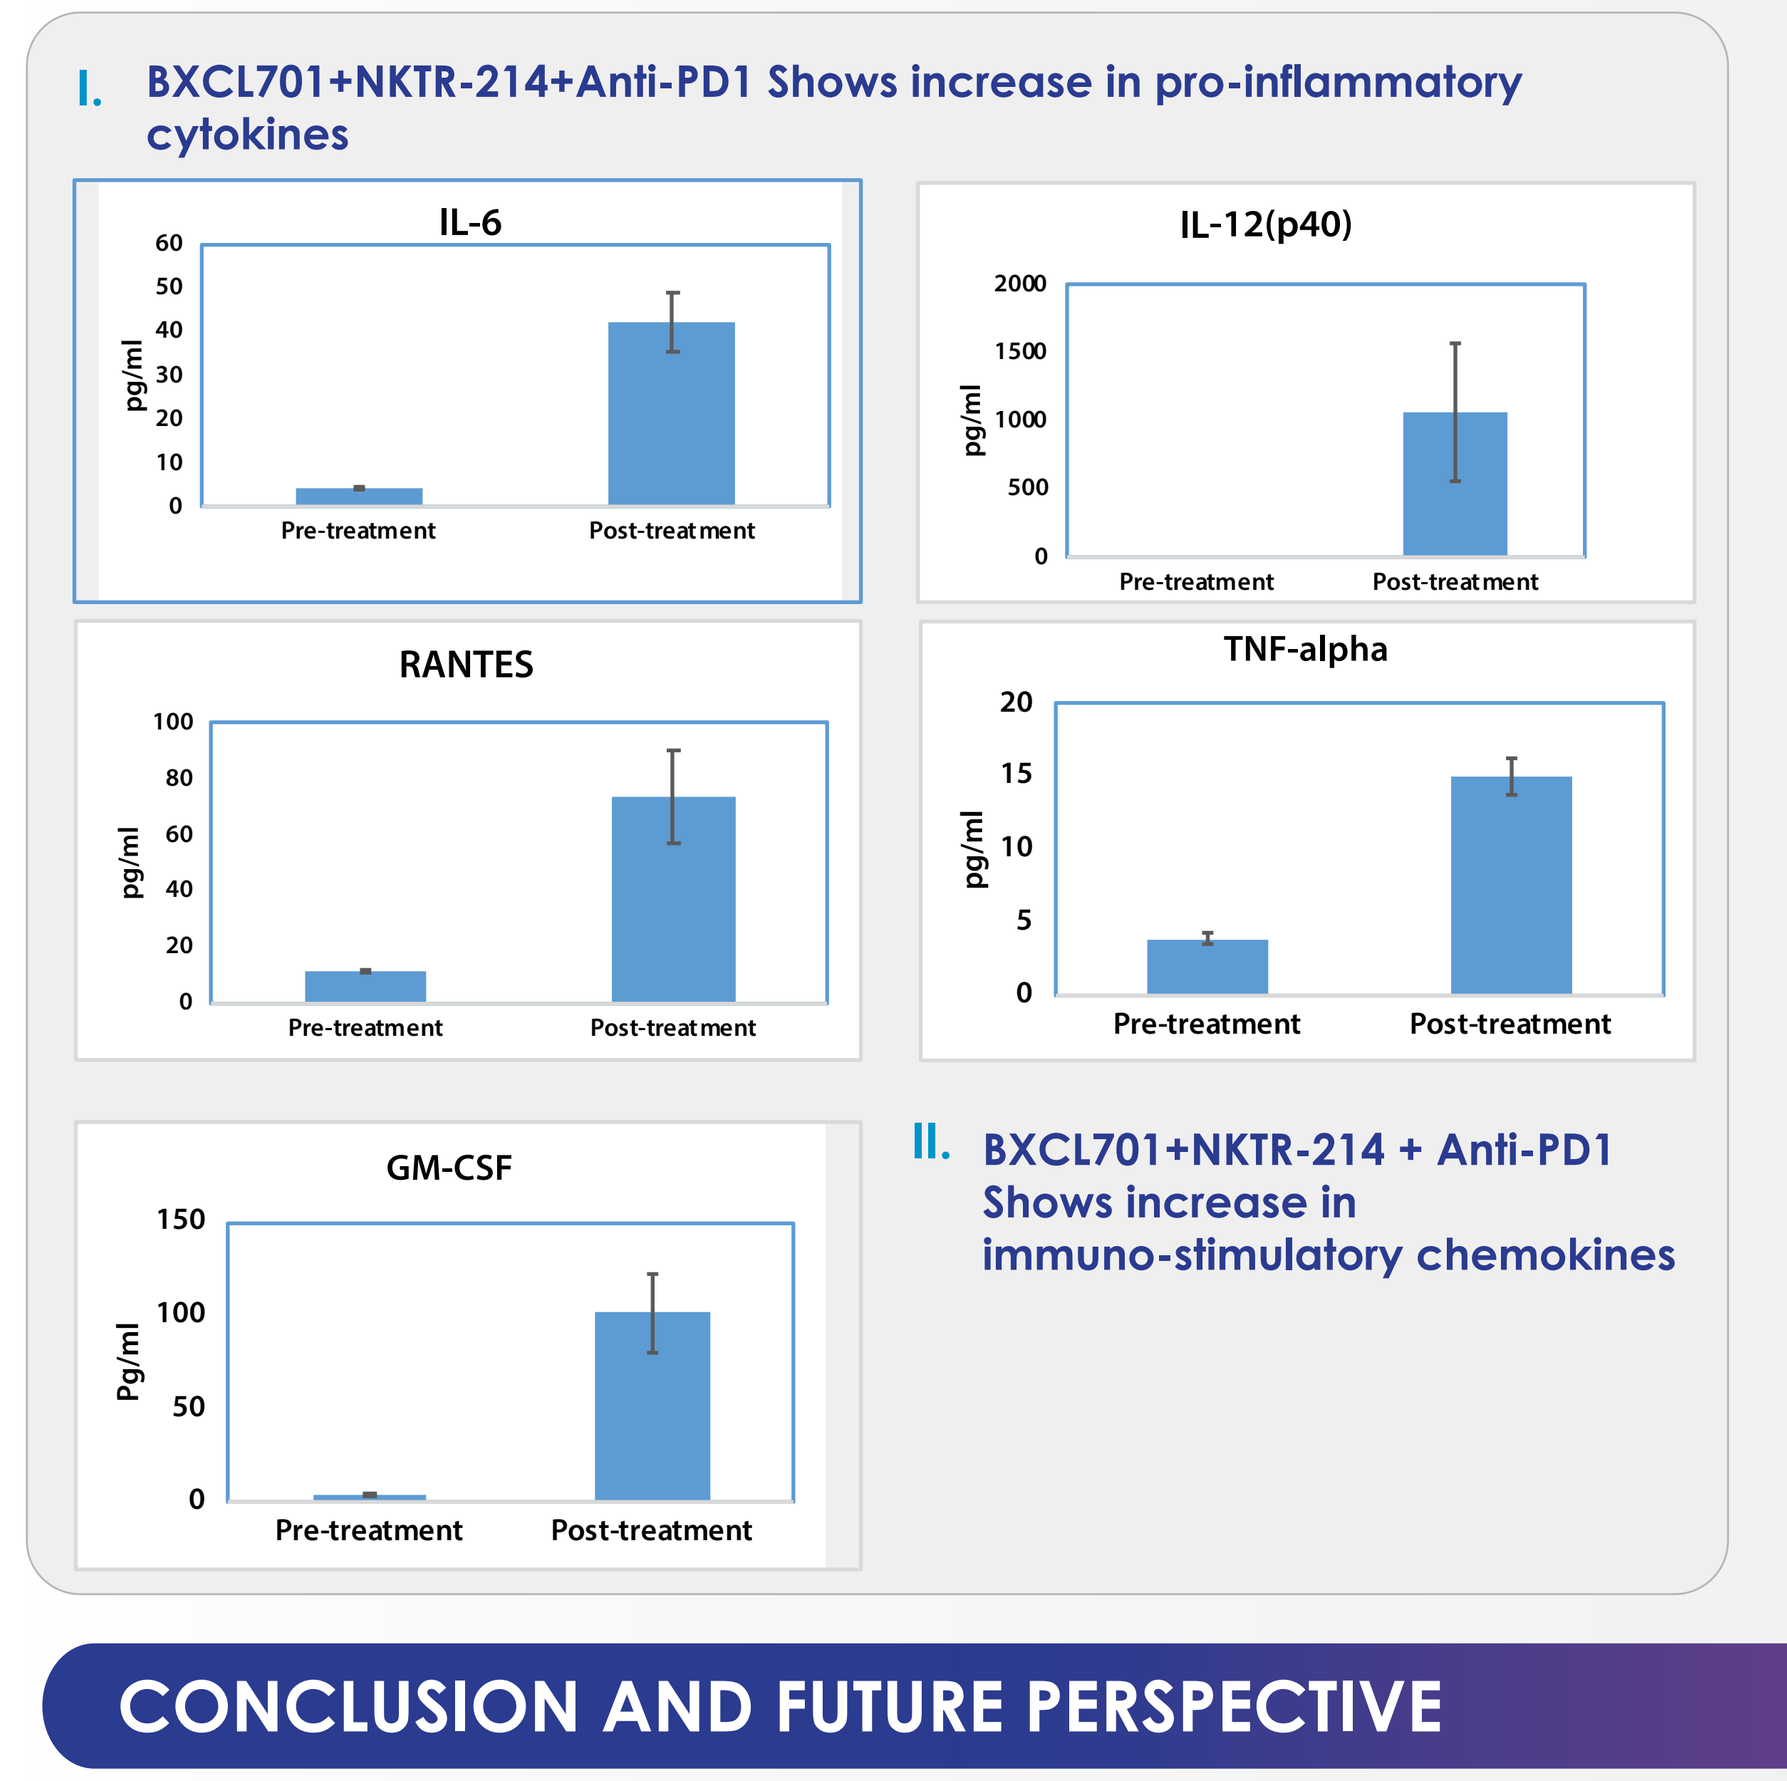
I. BXCL701+NKTR-214+Anti-PD1 Shows increase in pro-inflammatory cytokines
IL-6
IL-12(p40)
RANTES
TNF-alpha
GM-CSF
pg/ml
Pre-treatment
Post-treatment
II. BXCL701+NKTR-214 + Anti-PD1 Shows increase in immuno-stimulatory chemokines

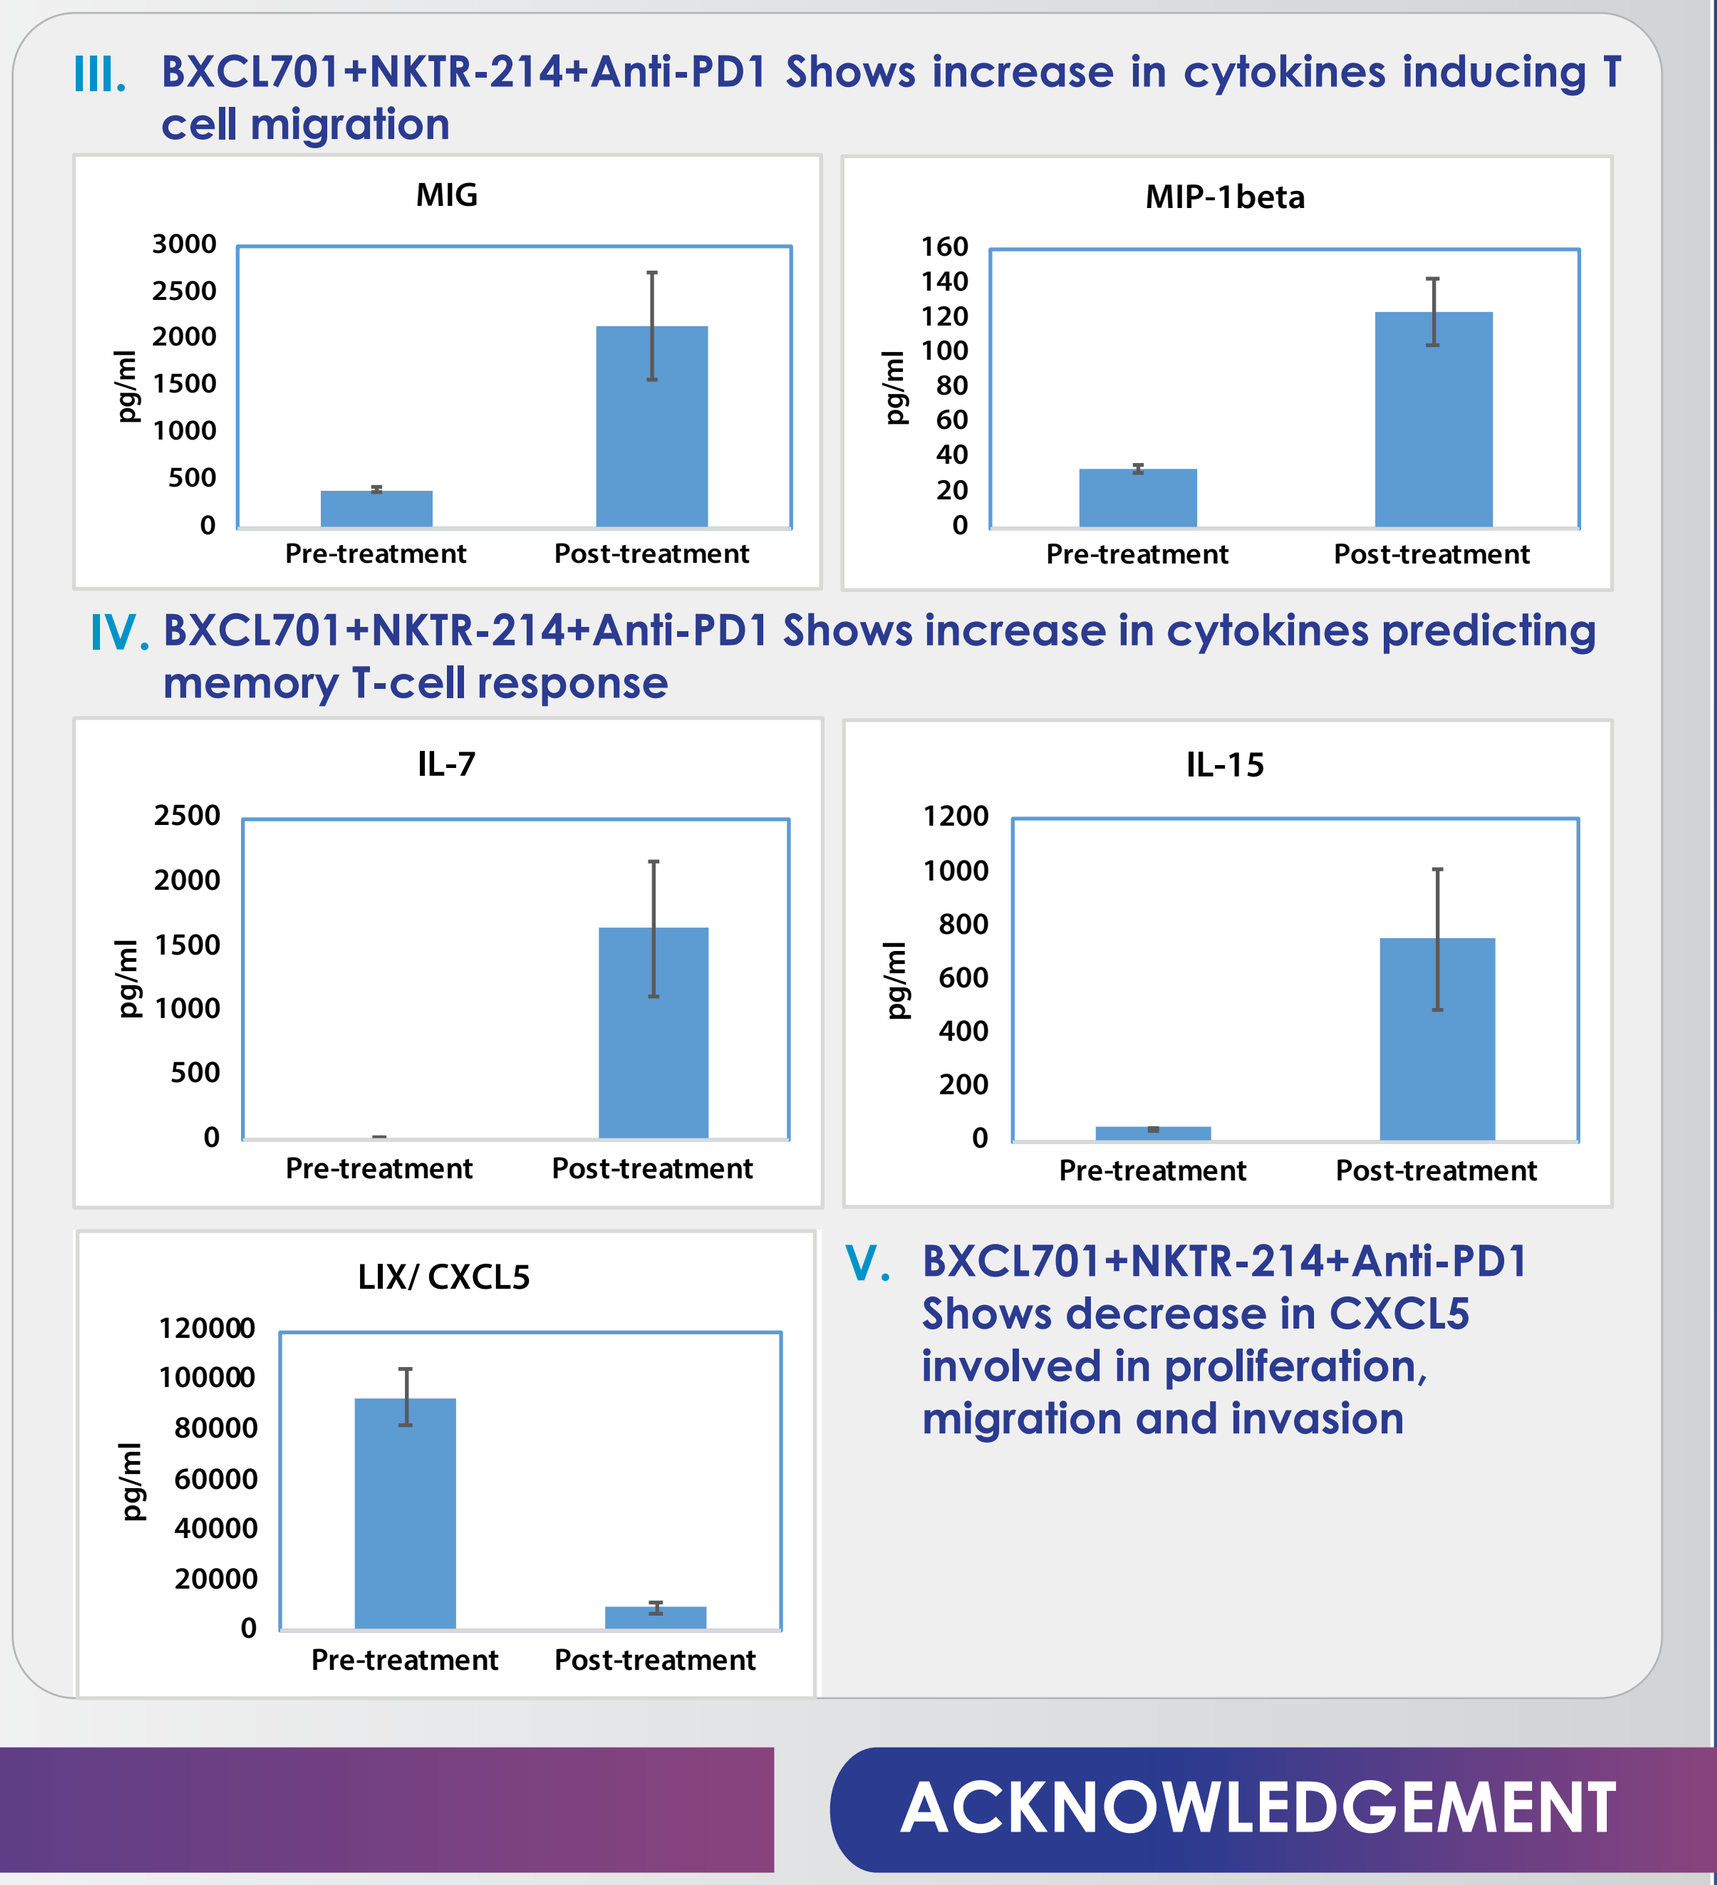
III. BXCL701+NKTR-214+Anti-PD1 Shows increase in cytokines inducing T cell migration
MIG
MIP-1beta
IV. BXCL701+NKTR-214+Anti-PD1 Shows increase in cytokines predicting memory T-cell response
IL-7
IL-15
LIX/ CXCL5
pg/ml
Pre-treatment
Post-treatment
V. BXCL701+NKTR-214+Anti-PD1 Shows decrease in CXCL5 involved in proliferation, migration and invasion

CONCLUSION AND FUTURE PERSPECTIVE

- The triple combination shows the induction of anti-tumor efficacy by non-overlapping complementary mechanisms in terms of 100% complete tumor regression.
- The triple combination demonstrates the development of long lasting anti-tumor immunity that persists after the cessation of the dosing, as observed upon tumor re-challenge. This further corroborated with the increase in cytokines involved in generation of memory T cell responses and pro-inflammatory cytokines as has been seen clinically for BXCL701.
- The combination also demonstrates involvement of both innate and adaptive immune responses in terms of increase in Ly6G+neutrophils, CD8+ T cells infiltrates within the tumor along with increase in immune stimulatory cytokines. This is accompanied by reduction in fibrotic barrier (FAP+ cells) as well as cytokines involved in invasion and migration.
- The study provides a validation of the anti-tumor impact of BXCL701, NKTR-214 and anti-PD1 combination in a syngeneic mouse model for pancreatic adenocarcinoma and indicates an exciting opportunity for the triple combination therapy in pancreatic cancer patients with maximum likelihood for a therapeutic benefit of this combination in ICI-resistant tumor cancer.

ACKNOWLEDGEMENT

We are immensely grateful to Dr. John McDougall for providing his valuable inputs during the editorial process of the poster content. We would like to appreciate the technical support provided by CrownBiosciences team for carrying out all the work associated with the animal studies. Further, it is the continuous support by our IP team that has enabled us to patent the potential concepts and we hereby acknowledge their constant support. We also thank the marketing and designing team for their constant support in making this poster attractive and presentable.

Abstract #3085/Poster #299, ASCO Annual Meeting, Monday June 4, 2018, Chicago