Phase 1b results of BXCL701, an IL-18 inducing Innate Immune Activator in combination with pembrolizumab (Keytruda), in patients with metastatic Castration-Resistant Prostate Cancer (mCRPC) including Small Cell/Neuroendocrine Cancer

V O'Neill, D Costin, J Zhang, L Karsh, M Linch, P Monk, J de Bono, R Aggarwal

METASTATIC CASTRATION-RESISTANT PROSTATE CANCER (MCRPC)- UNMET MEDICAL NEED

- American Cancer Society estimates ~191,930 new cases of prostate cancer in U.S. for 2020, 10–20% develop CRPC within approximately 5 years of follow-up, most of them having metastases at time of diagnosis
- Treatment of mCRPC has evolved rapidly over the past few years: first line treatment with androgen deprivation therapy (ADT) or one of the newer androgen signaling inhibitors (ASI), abiraterone or enzalutamide followed by chemotherapy with docetaxel is now standard of care. Docetaxel is associated with a median overall survival of less than 2 years
- Treatment Emergent Neuroendocrine Prostate Cancer (tENPC)is an aggressive tumor that appears in about 20% of patients progressing after treatment with ADT or ASI. This phenotype is typically treated with chemotherapy and associated with a shortened survival compared to the more common adenocarcinoma phenotype
- Despite demonstrated benefit of immunotherapies targeting PD-1, such as pembrolizumab on clinical outcomes in many solid tumors, mCRPC remains largely resistant to such therapies with single agent response rates around 6% (Keynote 199 ASCO-GU 2020). Further exploration has been focused on combination therapies.
- The BXCL701 immunomodulatory mechanism may manipulate the tumor micro-environment in such a way turning a "cold" tumor environment into an inflamed "hot" environment so that prostate cancer can overcome resistance to immunotherapy

BXCL701 MECHANISM OF ACTION

- A critical role of the innate immune system is to alert the adaptive immune system of an invading pathogen
 - Induction of Inflammation mediated through cytokine
 - Stimulation of antigen presentation by professional antigen presenting cells
- BXCL701 (Talabostat/Val-BoroPro) is a first-in-class oral small molecule inhibitor of dipeptidyl peptidases (DPP) DPP8 and DPP9
- BXCL701 is designed to trigger inflammasome mediated pyroptosis to alert and prime adaptive immune cells, leading to induction of IL-18 and IL-1ß, bridging between innate and adaptive immunity
- Activation of innate immunity has been shown to compliment T-cells targeting immunotherapy in syngeneic animal models, significant tumor responses were observed with BXCL701 plus checkpoint inhibition

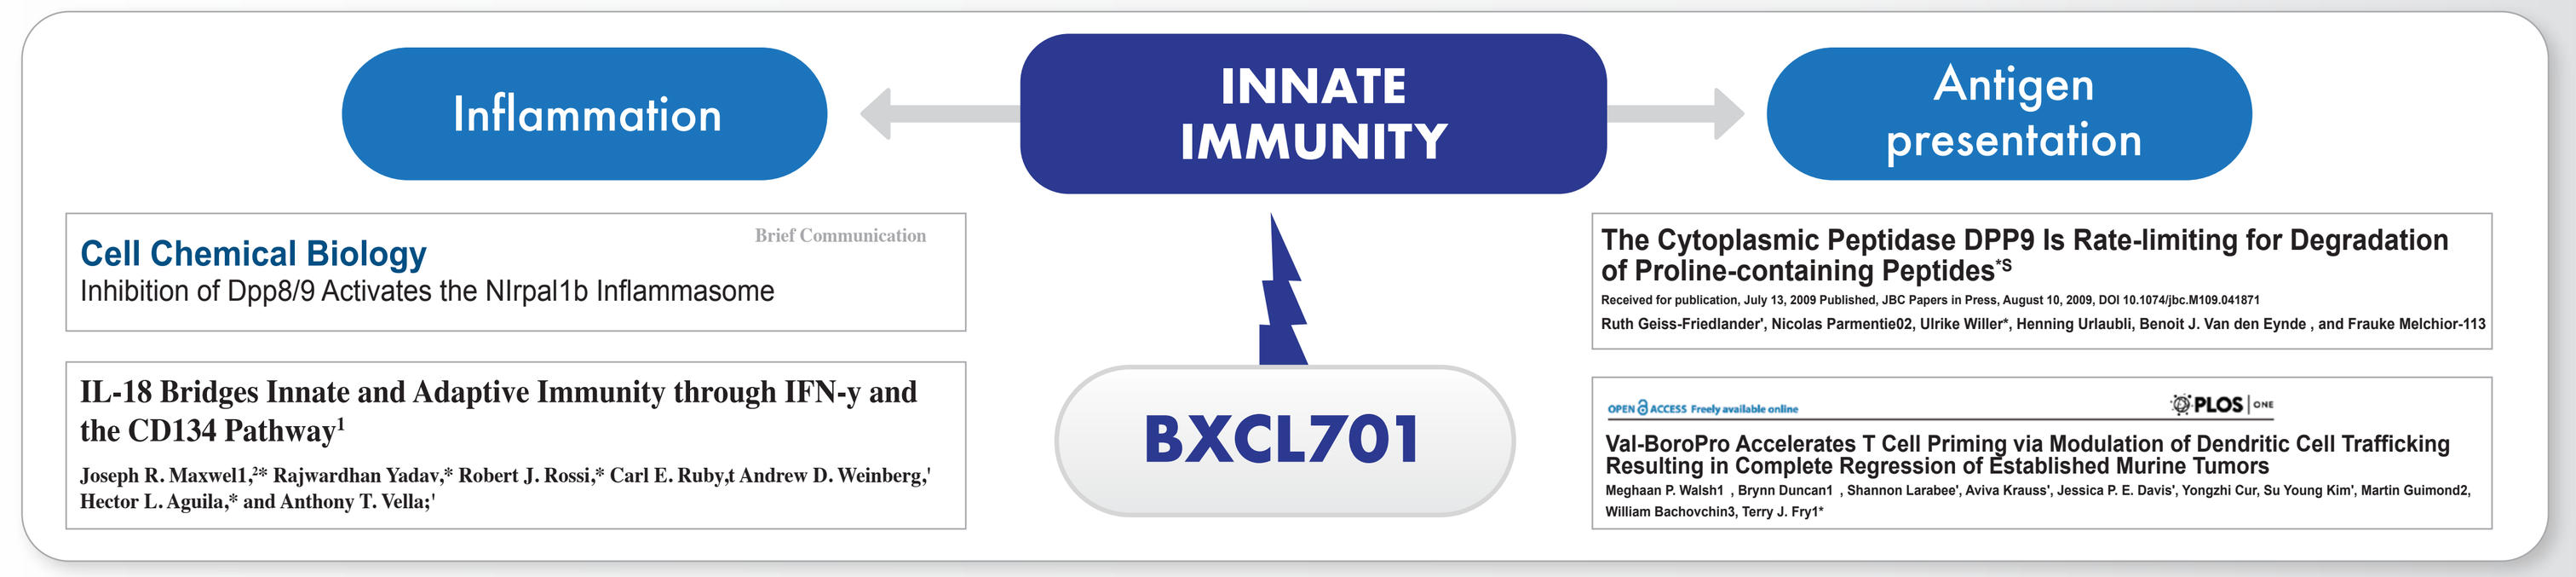

Inflammation
INNATE IMMUNITY
Antigen presentation
BXCL701
Cell Chemical Biology
Inhibition of Dpp8/9 Activates the Nlrpal1b Inflammasome
IL-18 Bridges Innate and Adaptive Immunity through IFN-y and the CD134 Pathway
The Cytoplasmic Peptidase DPP9 Is Rate-limiting for Degradation of Proline-containing Peptides
Val-BoroPro Accelerates T Cell Priming via Modulation of Dendritic Cell Trafficking Resulting in Complete Regression of Established Murine Tumors

MACROPHAGE PYROPTOSIS: THE CENTRAL COMPONENT OF BXCL701 ACTIVITY THROUGH DPP8/9 INHIBITION

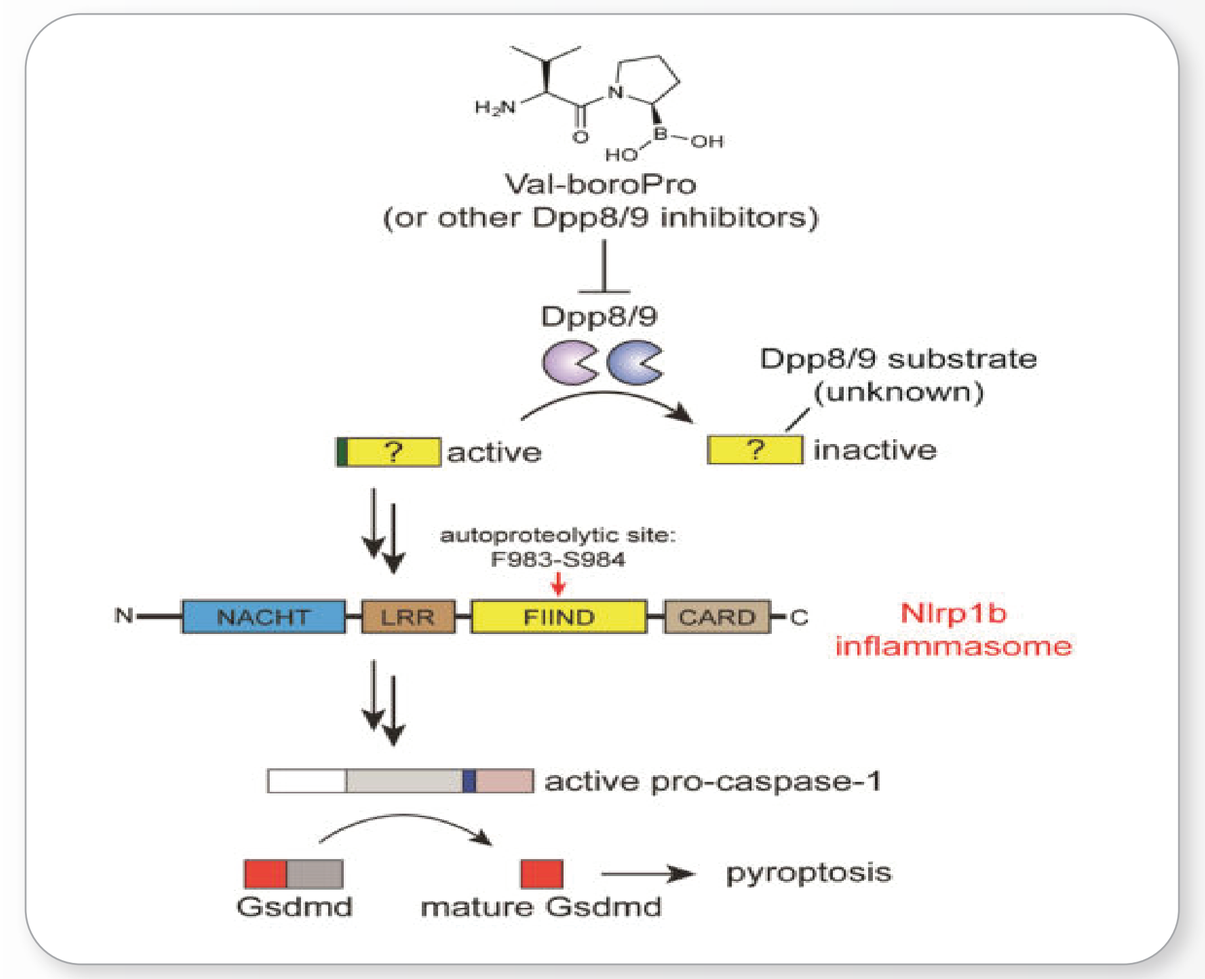

Val-boroPro
(or other Dpp8/9 inhibitors)
Dpp8/9
Dpp8/9 substrate (unknown)
active
inactive
autoproteolytic site: F983-S984
NACHT
LRR
FIIND
CARD
Nlrp1b inflammasome
active pro-caspase-1
Gsdmd
mature Gsdmd
pyroptosis

- Inhibition of DPP8/9 has been shown to activate the pro-inflammatory process of pyroptosis in macrophages
- Macrophage pyroptosis through the Nlrp1b pathway has been shown to drive the activation of caspase-1 and subsequent activation of pro-IL-18 and pro-IL-1β leading to the production of a host of cytokines and chemokines including IL-18 and IL-1β
- Mouse genetics further supports this process as the driver of BXCL701 activity via inhibition of DPP8/9

Okondo MC et al. Cell Chem Biol. 2018

PRECLINICAL DATA VALIDATES BXCL701 AND ANTI-PD-1 COMBINATION POTENTIAL

BXCL701/ANTI-PD-1 COMBINATION SHOWED:

- Synergistic antitumor effect on tumor volume in MC38 mouse model of colon adenocarcinoma
- Synergistic upregulation in the immunomodulatory parameters for pro-inflammatory cytokines*, chemokines+ and associated memory T-cells# by dose as single agent and in combination
- Synergistic increase in the cytotoxic NK cells and macrophages in the tumor with a decrease in the immunosuppressive T-regs

*IL-2, IL-12p40, IL-6; +GM-CSF, G-CSF; #IL-15, IL-7

TREATMENT EFFECT OF BXCL701 WITH ANTI-PD-1 ON MEAN TUMOR VOLUME IN MC-38 MOUSE MODEL

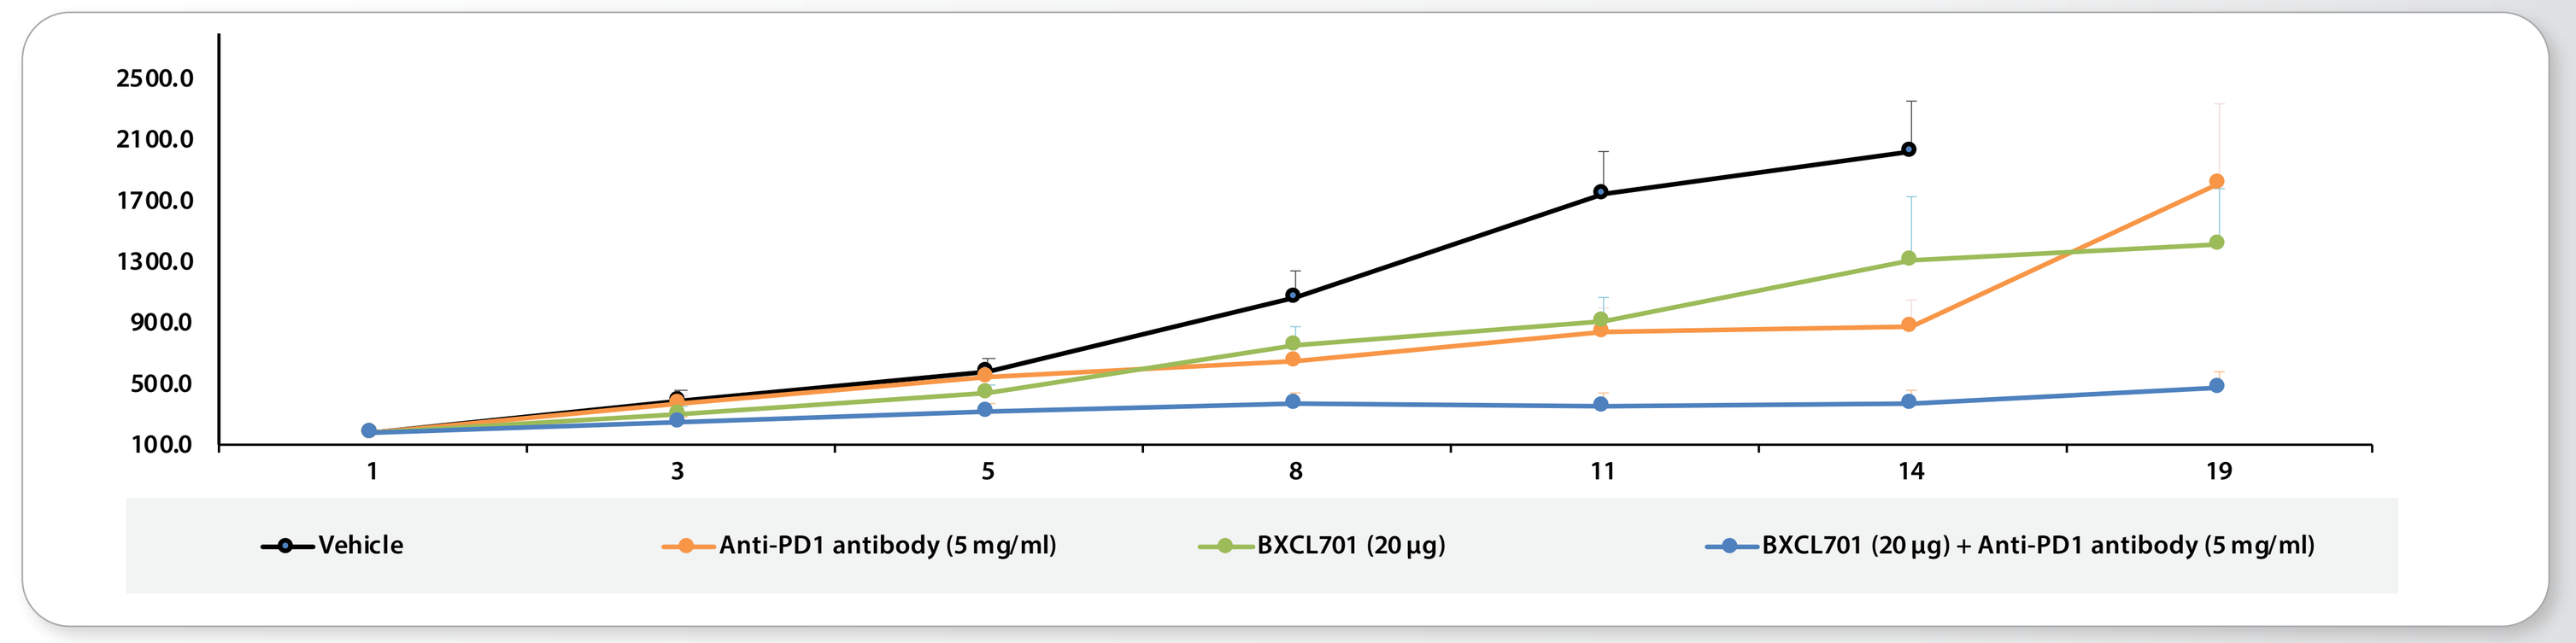

2500.0
2100.0
1700.0
1300.0
900.0
500.0
100.0
1
3
5
8
11
14
19
Vehicle
Anti-PD1 antibody (5 mg/ml)
BXCL701 (20 µg)
BXCL701 (20 µg) + Anti-PD1 antibody (5 mg/ml)

AACR2017 Abstract 2629

TRIAL SCHEMATIC AND KEY OBJECTIVES

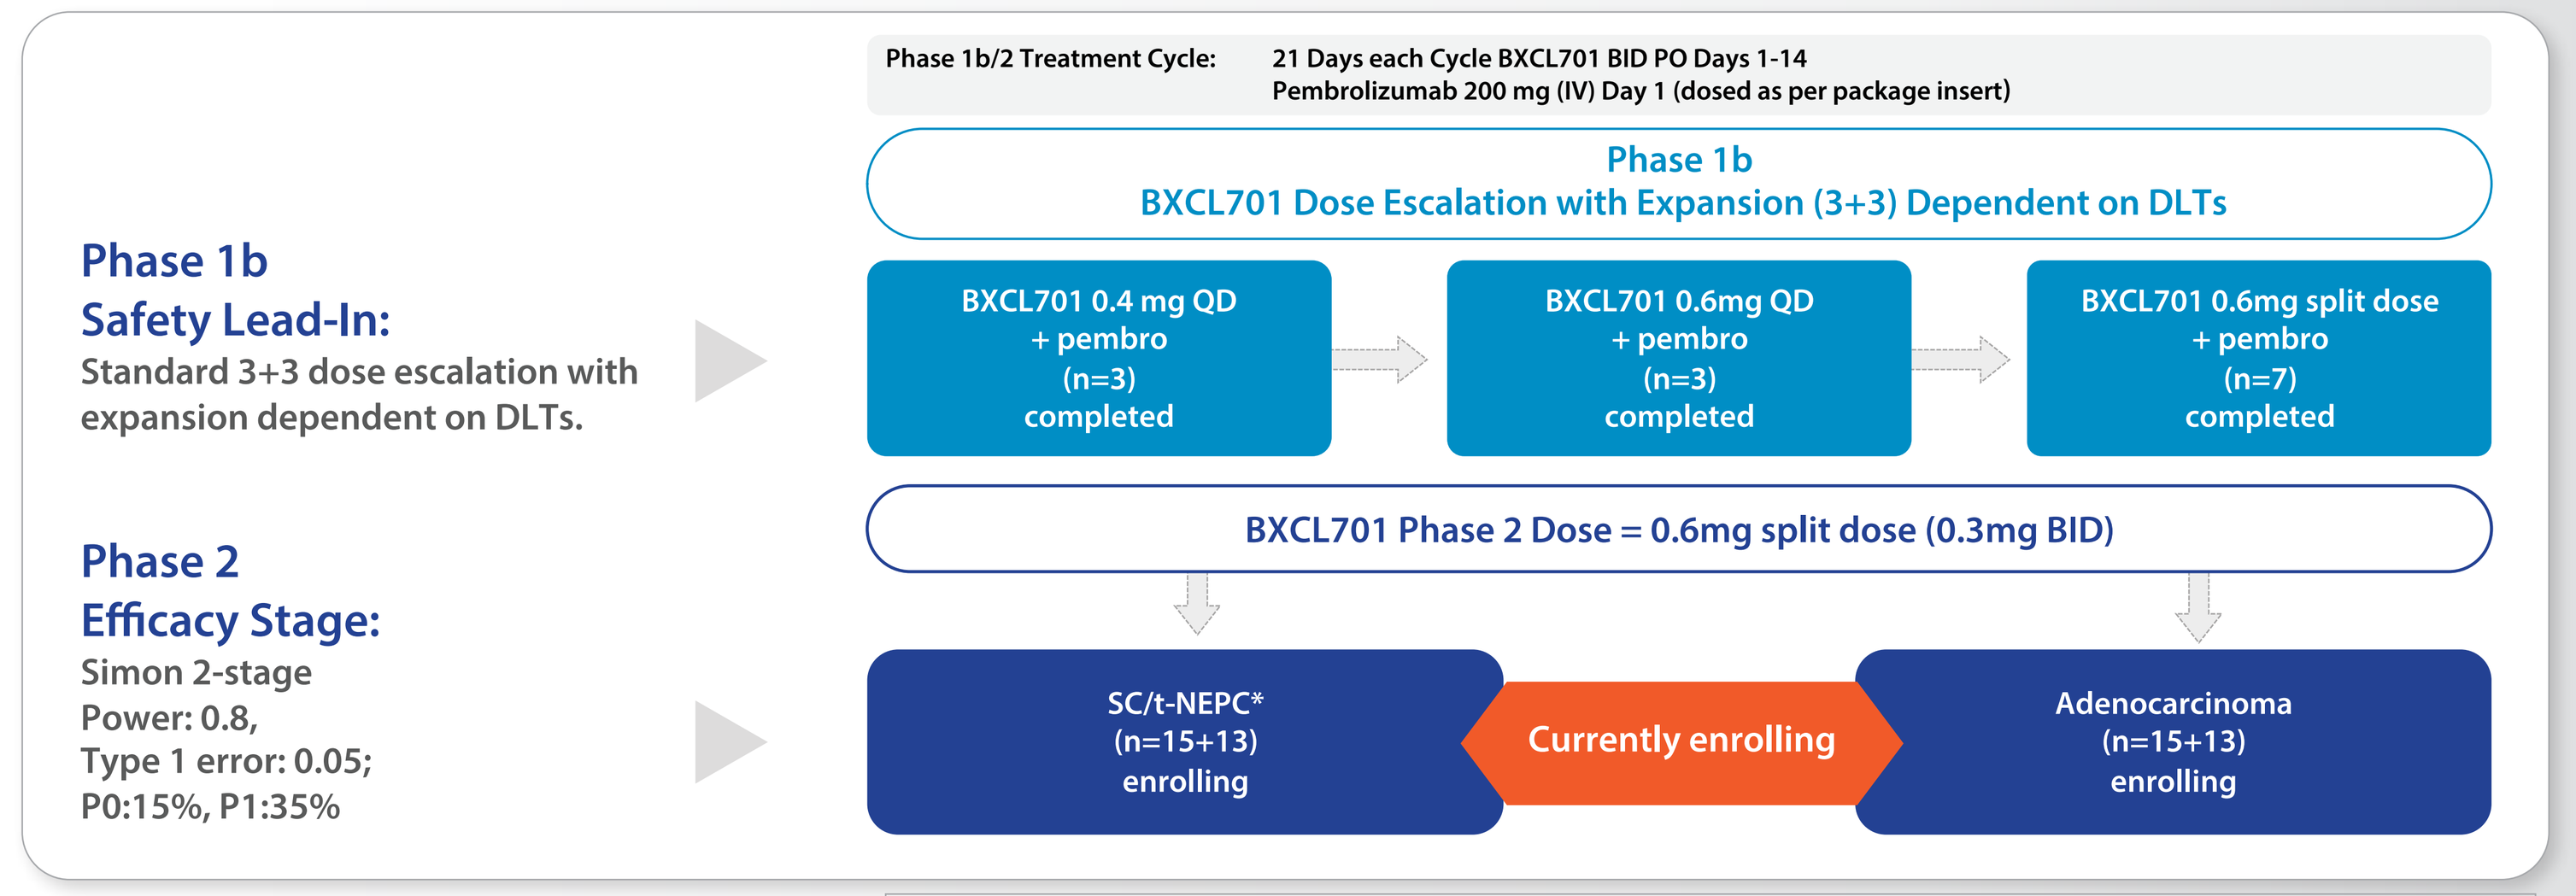

Phase 1b/2 Treatment Cycle: 21 Days each Cycle BXCL701 BID PO Days 1-14
Pembrolizumab 200 mg (IV) Day 1 (dosed as per package insert)
Phase 1b Safety Lead-In:
Standard 3+3 dose escalation with expansion dependent on DLTs.
Phase 1b
BXCL701 Dose Escalation with Expansion (3+3) Dependent on DLTs
BXCL701 0.4 mg QD + pembro (n=3) completed
BXCL701 0.6mg QD + pembro (n=3) completed
BXCL701 0.6mg split dose + pembro (n=7) completed
Phase 2 Efficacy Stage:
Simon 2-stage
Power: 0.8,
Type 1 error: 0.05;
P0:15%, P1:35%
BXCL701 Phase 2 Dose = 0.6mg split dose (0.3mg BID)
SC/t-NEPC* (n=15+13) enrolling
Currently enrolling
Adenocarcinoma (n=15+13) enrolling

Small-Cell/therapy-induced neuroendocrine prostate cancer

Primary Objective for each group:
Composite Response Rate: Target >15%
Need >2 composite responses to proceed to Stage 2

Additional Objectives:
DoR, PFS, changes in circulating cytokines and correlation of outcome with baseline tumor characteristics

KEY INCLUSION AND EXCLUSION CRITERIA

KEY INCLUSION CRITERIA

- Progression as defined by PCWG3 criteria
- At least 1 prior line of systemic therapy for locally advanced or metastatic prostate cancer
- Serum testosterone <50 ng/dL during Screening except for those with de novo small cell prostate cancer
- Eastern Cooperative Oncology Group (ECOG) performance status of 0-2
- Phase 2 Efficacy Stage only:
 - **For Cohort A (SCNC):**
 At least 1 prior line of chemotherapy | Measurable disease by RECIST 1.1
 - **For Cohort B (Adenocarcinoma):**
 - At least 1 but no more than 2, Androgen signaling inhibitors (ASI) and at least 1 prior line of taxane containing chemotherapy
 - Measurable disease by RECIST 1.1 or bone metastases

KEY EXCLUSION CRITERIA

- More than 2 cytotoxic chemotherapy regimens for CRPC
- Prior treatment with an anti-PD-1, anti-PD-L1, anti-programmed death-ligand 2 (PD-L2) agent or with an agent directed to another co-inhibitory T-cell receptor
- Additional active malignancy that may confound the assessment of the study endpoints
- Brain metastases that are symptomatic and progressive on imaging
- Significant cardiovascular or pulmonary disease

See ClinicalTrials.gov Identifier: NCT03910660 for more details

PHASE 1b RESULTS

Baseline Characteristics		BXCL701 0.4mg qd + Pembro (N=3)	BXCL701 0.6mg qd + Pembro (N=3)	BXCL701 0.6mg split dose + Pembro (N=7)	All (N=13)
		No. of Subjects			
Age	Mean (range), years	70 (61-75)	71 (62-86)	73 (62-84)	72 (61-86)
ECOG PS 0/1/2 (n)	PS 0/1/2 (n)	2/1/0	2/1/0	1/6/0	5/8/0
Most Recent Histopathology	Adenocarcinoma	3	2	2	7
	SC/t-NEPC	-	1	5	6
Prior Systemic Therapies**	Any ADT therapy	2	3	5	10
	1st generation ADT therapy	1	2	3	6
	2nd generation ASI	2	2	5	9
	Chemotherapy	3	1	7	11
	Radiotherapy	2	3	6	11

*ADT= Androgen deprivation therapy; ASI= Androgen signaling inhibitor | * For Patients with SC/t-NEPC mean age is 72 (range 62-84) | 30-Sep-2020*

EXPOSURE DURATION AND SUBJECT DISPOSITION

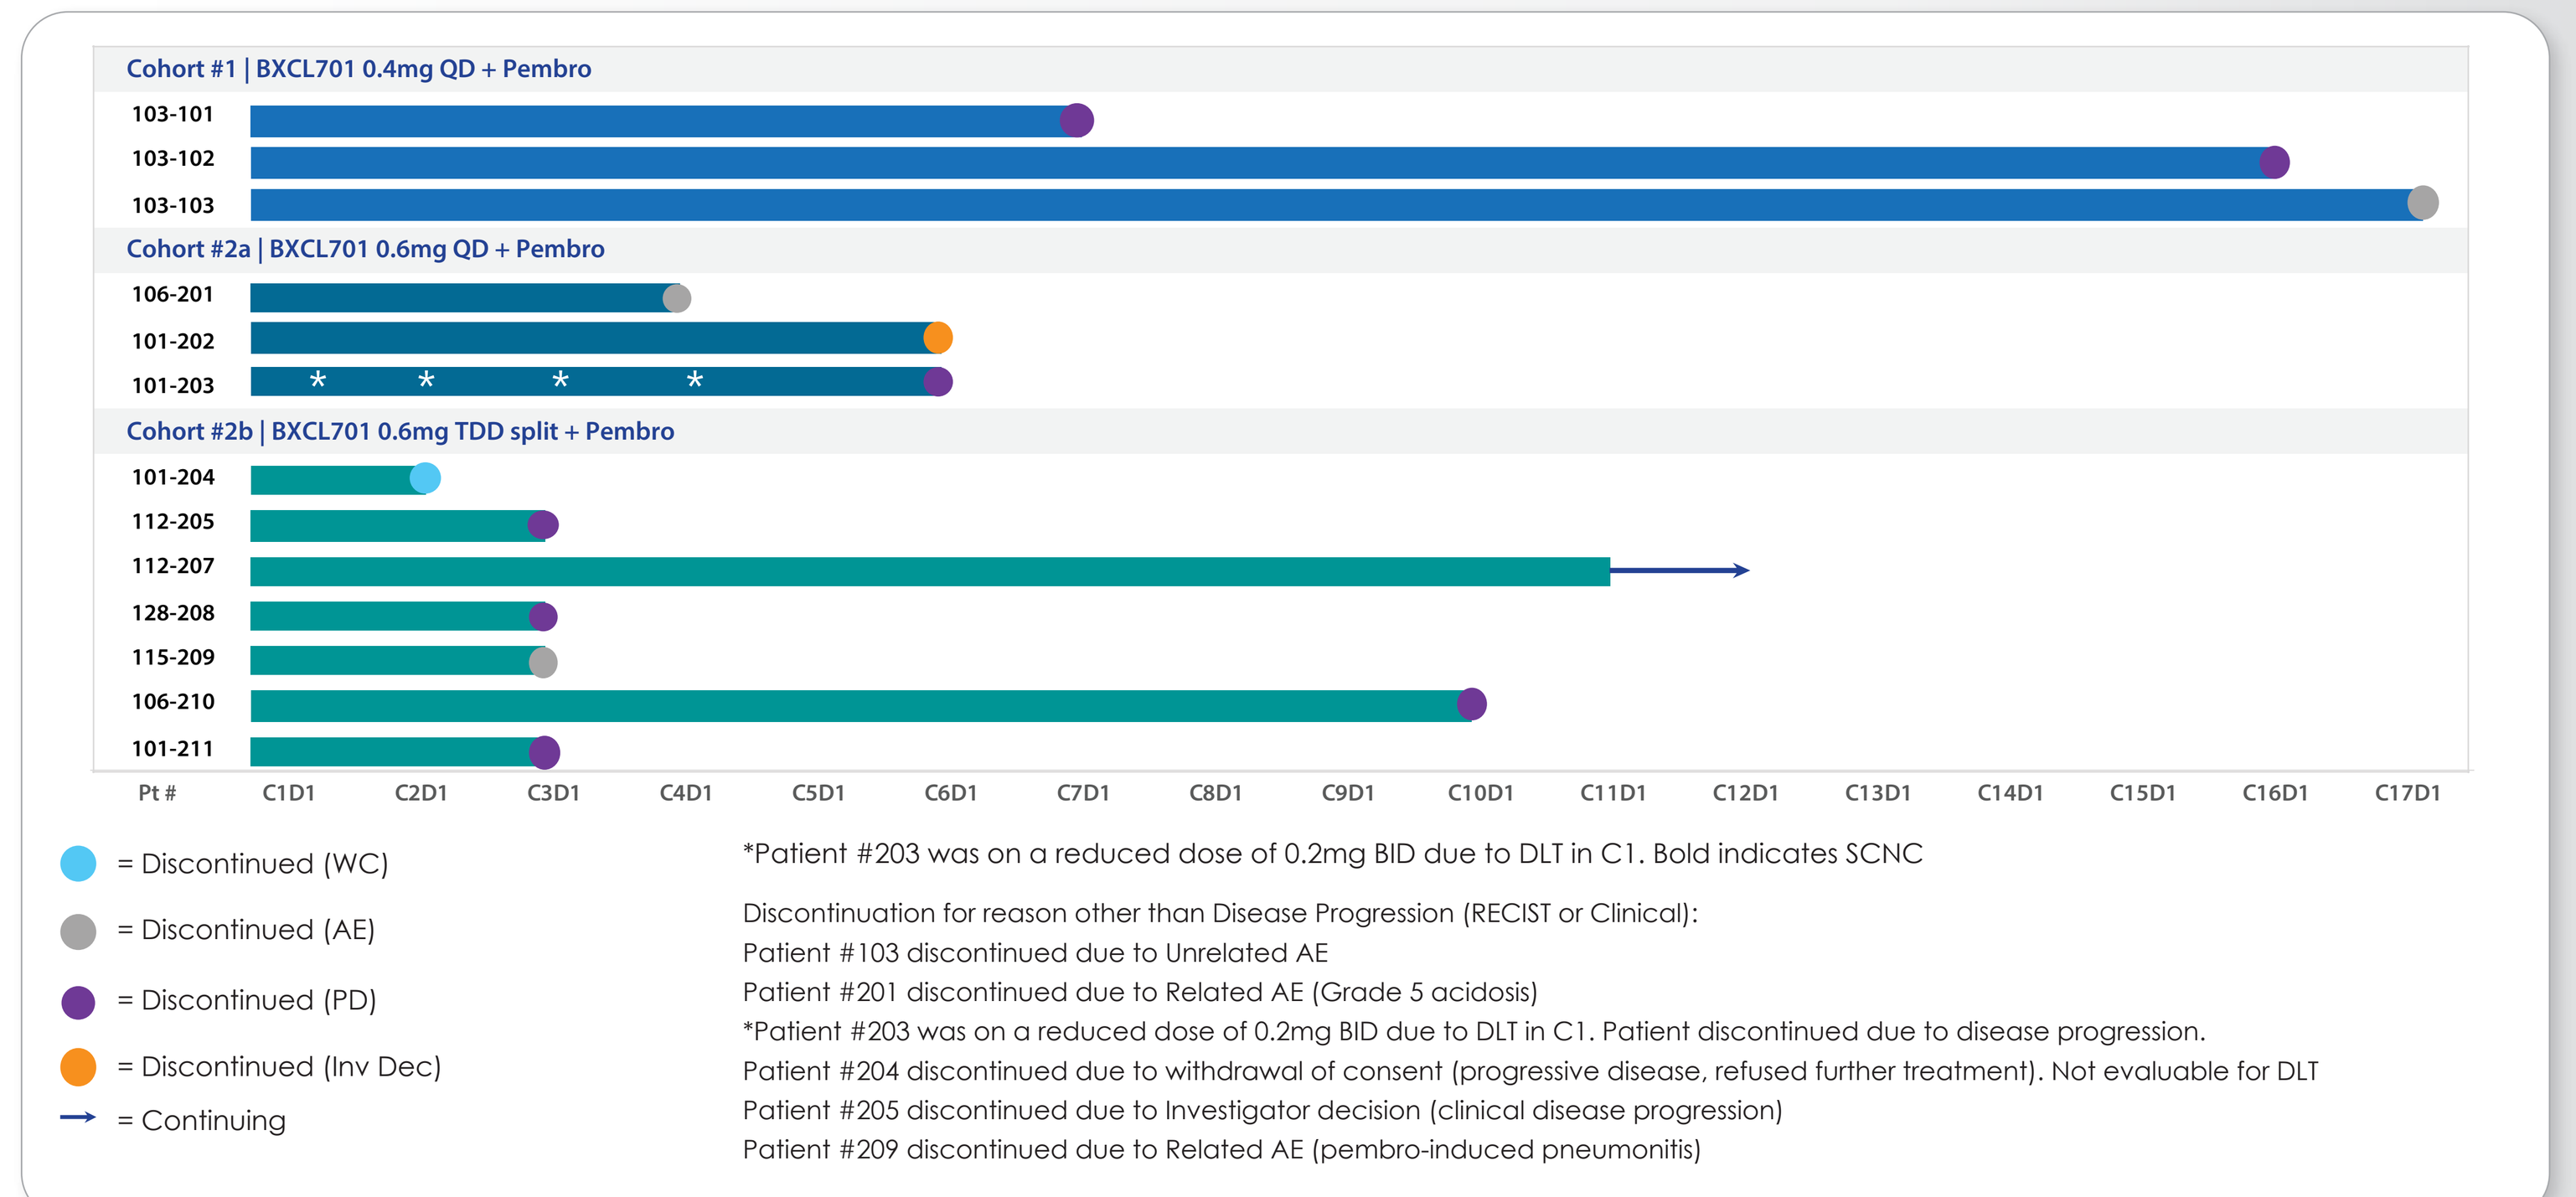

Cohort #1 | BXCL701 0.4mg QD + Pembro
103-101
103-102
103-103
Cohort #2a | BXCL701 0.6mg QD + Pembro
106-201
101-202
101-203
Cohort #2b | BXCL701 0.6mg TDD split + Pembro
101-204
112-205
112-207
128-208
115-209
106-210
101-211
Pt #
C1D1
C2D1
C3D1
C4D1
C5D1
C6D1
C7D1
C8D1
C9D1
C10D1
C11D1
C12D1
C13D1
C14D1
C15D1
C16D1
C17D1
= Discontinued (WC)
= Discontinued (AE)
= Discontinued (PD)
= Discontinued (Inv Dec)
= Continuing

*Patient #203 was on a reduced dose of 0.2mg BID due to DLT in C1. Bold indicates SCNC

Discontinuation for reason other than Disease Progression (RECIST or Clinical):
Patient #103 discontinued due to Unrelated AE
Patient #201 discontinued due to Related AE (Grade 5 acidosis)
*Patient #203 was on a reduced dose of 0.2mg BID due to DLT in C1. Patient discontinued due to disease progression.
Patient #204 discontinued due to withdrawal of consent (progressive disease, refused further treatment). Not evaluable for DLT
Patient #205 discontinued due to Investigator decision (clinical disease progression)
Patient #209 discontinued due to Related AE (pembro-induced pneumonitis)

SAFETY RESULTS SUMMARY

AE Category	BXCL701 0.4mg qd + Pembro N=3	BXCL701 0.6mg qd + Pembro N=3	BXCL701 0.6mg split dose + Pembro (N=7)
	No. of Patients		
Any DLT in Cycle 1	0	1	0
Any SAE	2	1	4
Any treatment-related SAE*	0	1	1
Treatment-related AE leading to Discontinuation	0	1	1
Any BXCL701 treatment-related AE*	3	3	5
Any pembro treatment-related AE*	2	2	5
Any BXCL701 or pembro treatment-related ≥Grade 3 AE*			
Non-hematologic	-	2#	3^
Hematologic	3**	1##	1^^

* Includes related and possibly-related events:
** 2 Patients: Grade 3 anemia, 1 pt Grade 4 thrombocytopenia
\# 1Patients each: Grade 3 syncope (DLT- C1D6) and Grade 5 acidosis (C3)
\## 1Patients with Grade 3 Hgb and WBC increase
^ 1Patients: Grade 3 Fatigue, fatigue present at baseline, 1pt Grade 3 pneumonitis, SOB, fatigue, hyponatremia, 1pt Grade 3 lactic acid increase
^^ 1Patients: Grade 3 Anemia

BXCL701 ON-TARGET ADVERSE EVENTS CONSISTENT WITH CYTOKINE ACTIVATION WERE SEEN AT THE HIGHEST DOSES

Preferred Term	BXCL701 0.4mg qd+ Pembro N=3				BXCL701 0.6mg qd + Pembro N=3				BXCL701 0.6mg Split dose+ Pembro N=7			
	(Grade) N											
BXCL701 Related Events#	(Any)	(1)	(2)	(3)	(Any)	(1)	(2)	(3)	(Any)	(1)	(2)	(3)
Hypotension*	-	-	-	-	3	2	1	-	1	1	-	-
Dizziness	-	-	-	-	2	2	-	-	-	-	-	-
Headache	-	-	-	-	1	1	-	-	-	-	-	-
Syncope	-	-	-	-	1	-	-	1	-	-	-	-
Dyspnea	-	-	-	-	1	1	-	-	1	-	1	-
Chills	-	-	-	-	-	-	-	-	1	-	1	-
Edema	-	-	-	-	2	1	1	-	-	-	-	-
Fatigue	-	-	-	-	1	-	1	-	3	2	-	1†
Myalgia	-	-	-	-	1	1	-	-	1	1	-	-

\# Relatedness as assessed by investigator, *Includes orthostatic hypotension, † Fatigue present at baseline

BXCL701-201: IL-18 CYTOKINE EVALUATION IN PHASE 1B COHORTS

Mechanistic relevance of IL-18 : IL-18, produced mainly by macrophages, induces NK cell activation, and induces IFN-γ in T cells has been shown to act as a bridge between innate and adaptive immunity, favouring anti-tumor immunity.

Results :

- IL-18 levels generally increased post dose in all the dose cohorts (0.4 mg qd, 0.6 mg qd and 0.6mg split dose in combination with pembro)
- 106-201 (0.6 mg QD + pembro) showed maximum IL-18 levels (45 fold), clinical findings of URI may have contributed
- 0.6 mg split dose + pembro showed consistent increases in IL-18 on day 14 post dose (8-24 fold)
- Consistent dose and time dependent increases in IL-18 levels observed in 0.6 mg split dose, maximal changes noted on day 14.

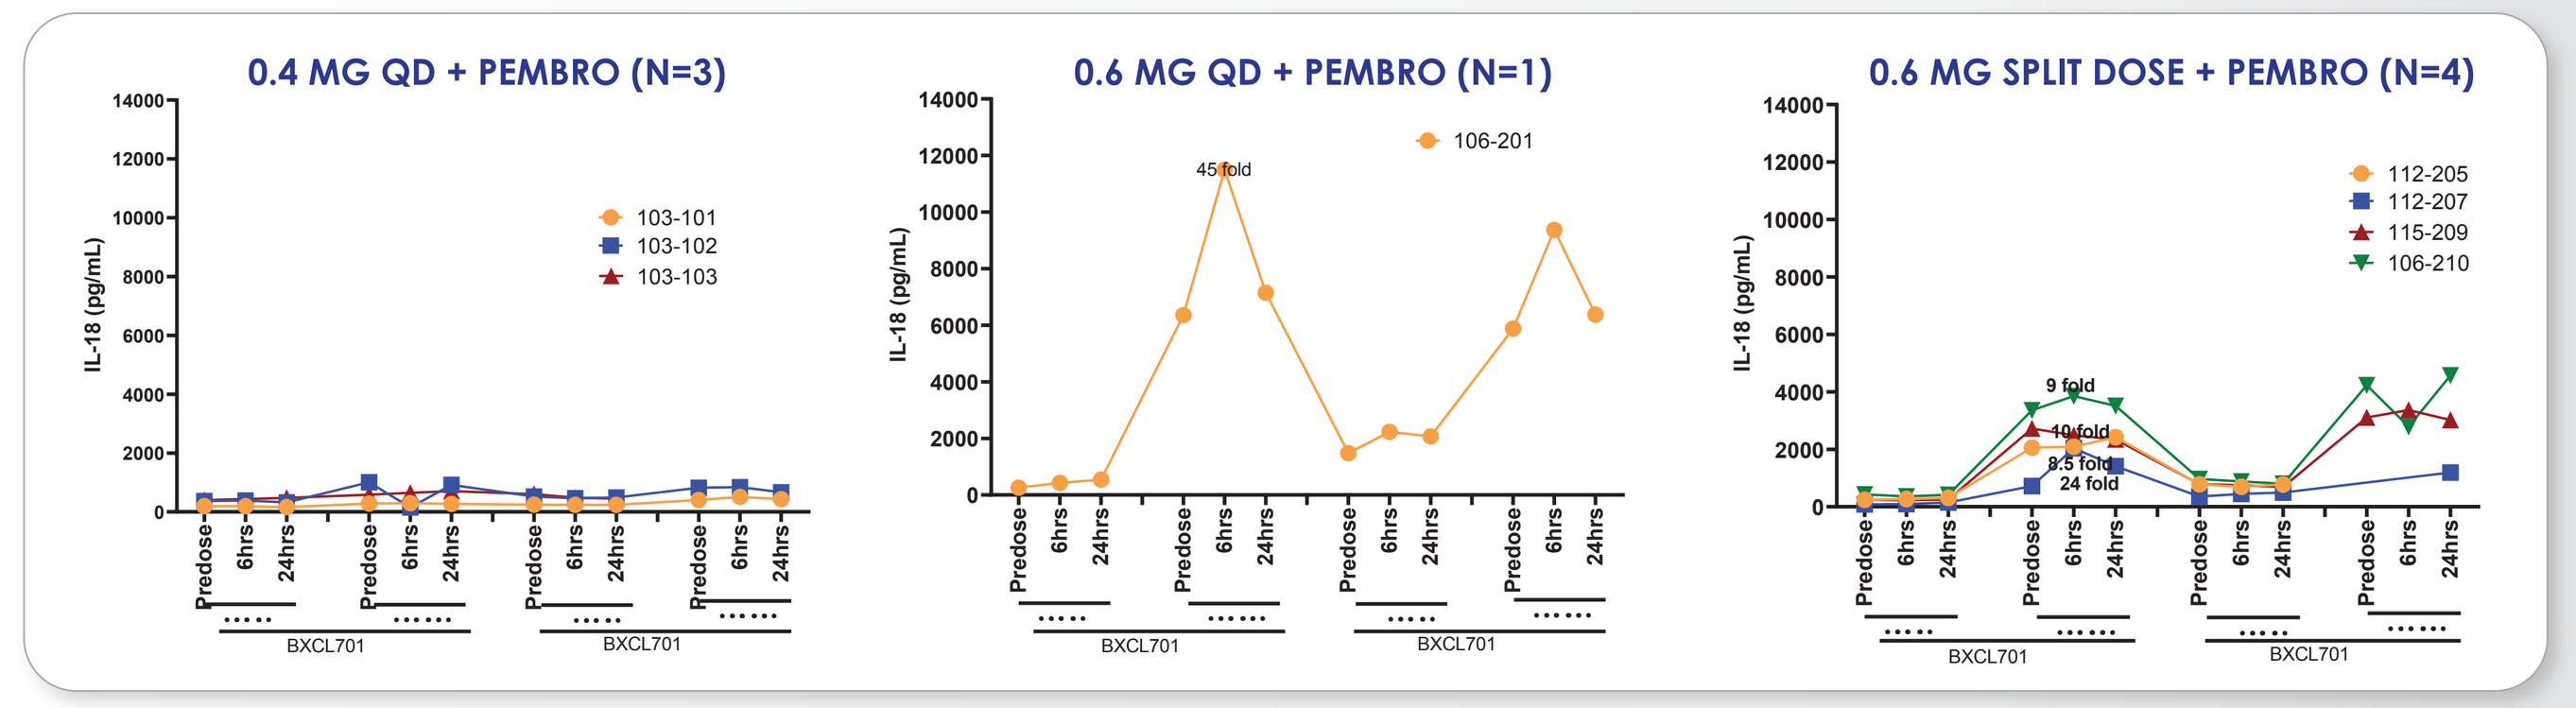

0.4 MG QD + PEMBRO (N=3)
0.6 MG QD + PEMBRO (N=1)
0.6 MG SPLIT DOSE + PEMBRO (N=4)
IL-18 (pg/mL)
103-101
103-102
103-103
106-201
45 fold
112-205
112-207
115-209
106-210
9 fold
10 fold
8.5 fold
24 fold
Predose
6hrs
24hrs
BXCL701

INTERIM EFFICACY RESULTS

Responses	BXCL701 0.4mg qd + Pembro N=3	BXCL701 0.6mg qd + Pembro N=3	BXCL701 0.6mg split dose+Pembro N=7
RECIST 1.1			
SD	2	1	1
Non-CR/Non-PD	1	-	1
PD	-	1	2
NE	-	1*	3*
PSA Evaluable	3	2	5
PSA Response†	0	0	1

† ≥50% decline by week 12
* Early death or early DC without follow-up disease assessment

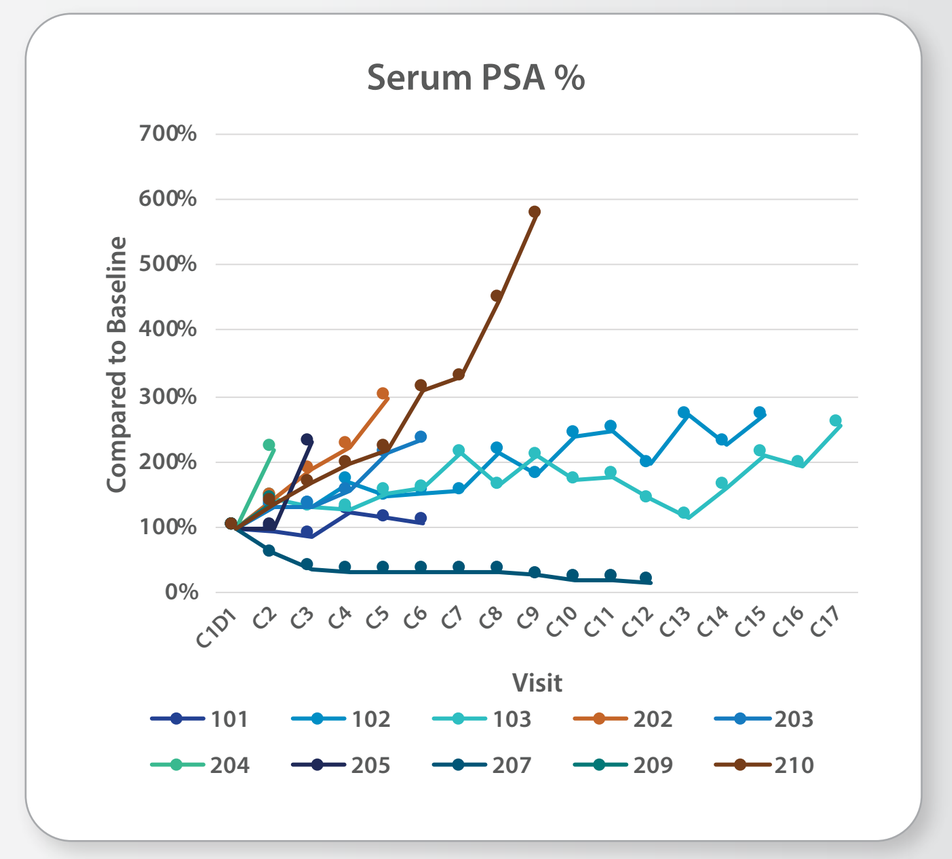

Serum PSA %
Compared to Baseline
700%
600%
500%
400%
300%
200%
100%
0%
Visit
101
102
103
202
203
204
205
207
209
210

CONFIRMED PSA RESPONDER

PRE-STUDY DETAILS

- Patient Demographics and History:
 - 73 y.o.
 - Diagnosed 2006 with Adenocarcinoma
 - Other medical history: coronary artery disease, hypertension, MI (2003), limb edema
 - Sites of disease at study entry: multiple lymph nodes and bone lesions
 - Baseline PSA 163.1ng/mL
- Prior Systemic Therapy included:
 - Bicalutamide
 - Abiraterone + Prednisone
 - Enzalutamide
 - Sipuleucel
 - Docetaxel/Cabazitaxel

ON-TREATMENT DISEASE STATUS

- Duration of BXCL701+Pembro: 11+ cycles
- Confirmed PSA Response:
 - Time to PSA response: 6weeks (60.4ng/mL - 63% reduction) Ongoing at 30 weeks (25.9ng/mL - 84% reduction)
- RECIST 1.1 Status: Minor Response
 - Target Lesions: Minor Response ongoing (22% reduction 51mm>40mm)
 - Non-Target Lesions: Disappearance of 2/4 non-target lymph nodes and non-PD/non-CR bone disease.

CONCLUSION

- Results from the Phase 1b safety assessment of BXCL701 support the use of a split dose totaling 0.6mg per day as the recommended dose when used in combination with pembrolizumab.
- In this study:
 - On-target adverse events consistent with cytokine activation were seen at the highest daily dose tested.
 - Splitting the daily dose was associated with improved tolerability as evidenced by no reported DLTs and lower rates of other adverse events of interest such as hypotension and peripheral edema
 - Long-term safety was evaluated at the lower dose level, while data accumulates from patients continuing with the split dose regimen at 0.6mg daily.
 - Consistent dose and time dependent increases in IL-18 levels observed in 0.6 mg split dose, maximal changes noted after 14 days of continuous dosing.
 - Preliminary evaluation of anti-tumor activity showed 1 patient with a PSA response and four patients with stable disease per RECIST 1.1 including 1 patient still on treatment with minor response
- The Phase 2 efficacy portion of this study is currently enrolling patients with SC/t-NEPC or adenocarcinoma and will assess the anti-tumor activity of the combination in a setting where checkpoint inhibitor monotherapies have demonstrated limited clinical benefit

THANK YOU

BioXcel Therapeutics would like to thank all patients, their families, and caregivers who make this study possible. BioXcel would also like to thank the participating investigators and their staff for their support on this study and their dedication to their patients, despite the additional challenges as a circumstance of the COVID-19 pandemic.